Poster W118
FOCIS 2025 Annual Meeting
June 25, 2025

HiFiBiO THERAPEUTICS

HFB202501, a Novel Anti-PD-1 Antibody with Enhanced Effector Functions for Precision Depletion of Pathogenic T Cells in Autoimmune Diseases

Roshan Kumar, Qian Zhang, Chun Gao, Wenhua Xu, Pin Cai, Tengfei Wang, Qiu Jin, Song Cheng, Jia Wu, Jinxu Liao, Ling Dong, Hillary Shah, Juying Li, Yun-Yueh Lu, Jinping Gan, Francisco Adrian, Liang Schweizer

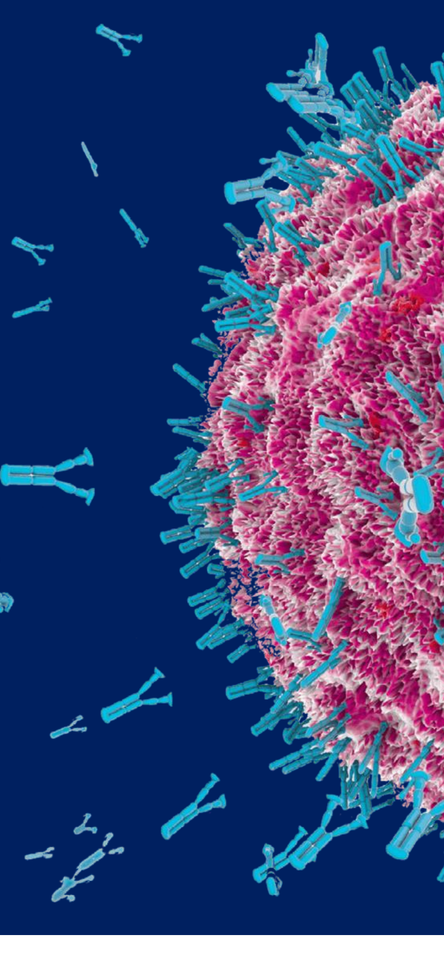

BACKGROUND

Precision depletion of pathogenic immune cells is an attractive therapeutic strategy with the potential to reset immune responses and restore tolerance while retaining aspects of protective immunity[1]. The immune checkpoint PD-1 has drawn interest as a therapeutic target in autoimmune and inflammatory diseases where it marks multiple pathogenic T cell subtypes[2–4]. These include T peripheral and follicular helper (Tph and Tfh) cells, critical for the maturation and development of autoreactive B cells, which express very high levels of PD-1, and activated effector cells which contribute to inflammation and tissue damage and express lower levels of PD-1. Antibodies that agonize PD-1-expressing T cells and deplete PD-1(high) T cells have recently demonstrated clinical proof-of-concept in rheumatoid arthritis[5,6].

We present here HFB202501, an anti-PD-1 agonist antibody engineered for enhanced depletion of PD-1+ cells. HFB202501 shows more potent and deeper depletion of PD-1+ T cells in vitro compared to clinical-stage benchmark antibodies. HFB202501 is able to deplete multiple PD-1+ cell populations from human tonsils in vitro and is highly efficacious in a mouse model of lupus. Deep depletion of PD-1+ cells has the potential for broad application in autoantibody-mediated autoimmune diseases and other indications where activated PD-1+ effector cells play a pathogenic role. HFB202501 is progressing towards preclinical development with a biomarker strategy for identifying patients with expanded PD-1+ cell populations.

__Depletion of T peripheral (Tph) and T follicular (Tfh) helper cells__ expressing high levels of PD-1 along with __pathogenic PD-1+ T effector cells__ is expected to disrupt auto-antibody production, reset the immune response and restore tolerance

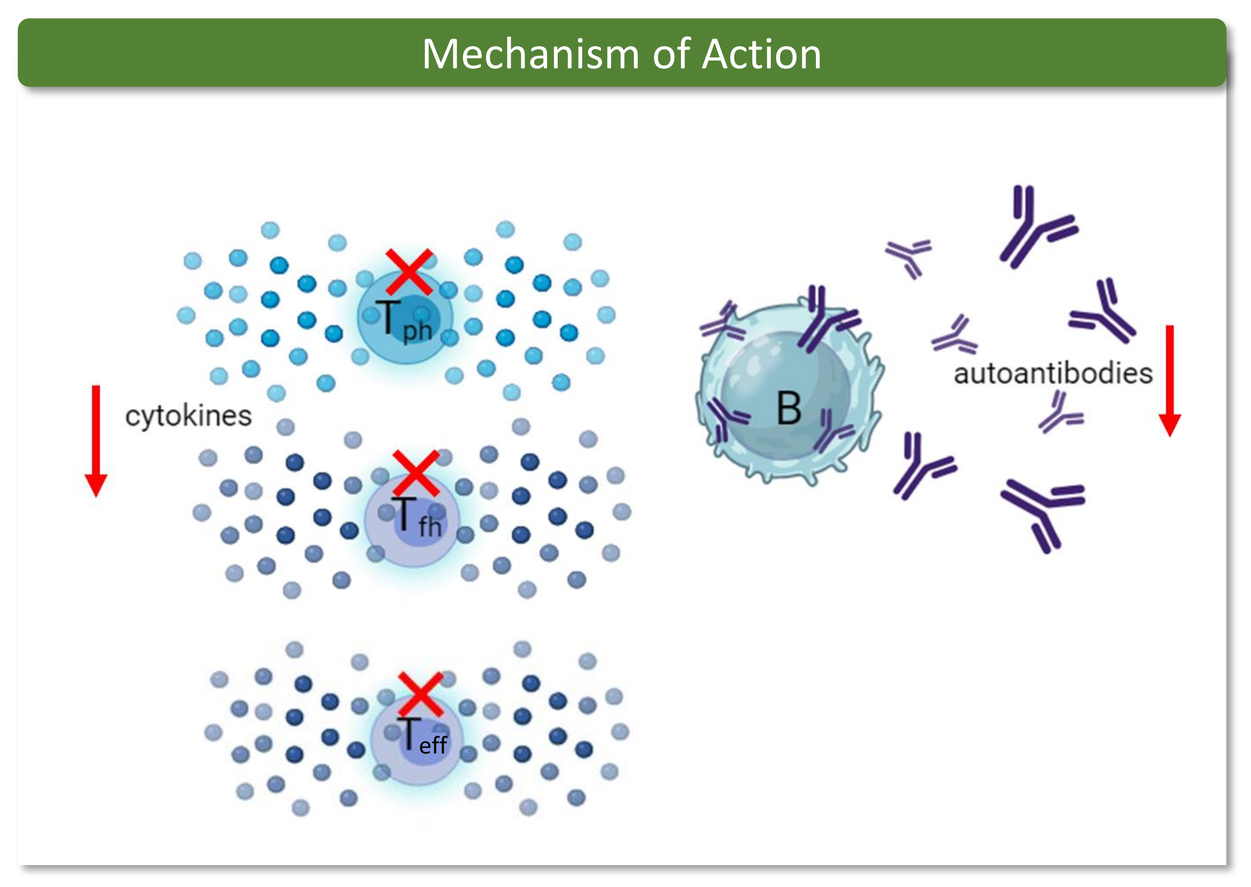
Mechanism of Action
cytokines
Tph
Tfh
Teff
B
autoantibodies

RESULTS | Target analysis

Single cell analysis of PD-1 Expression in HiFiBiO Disease Cell Atlas

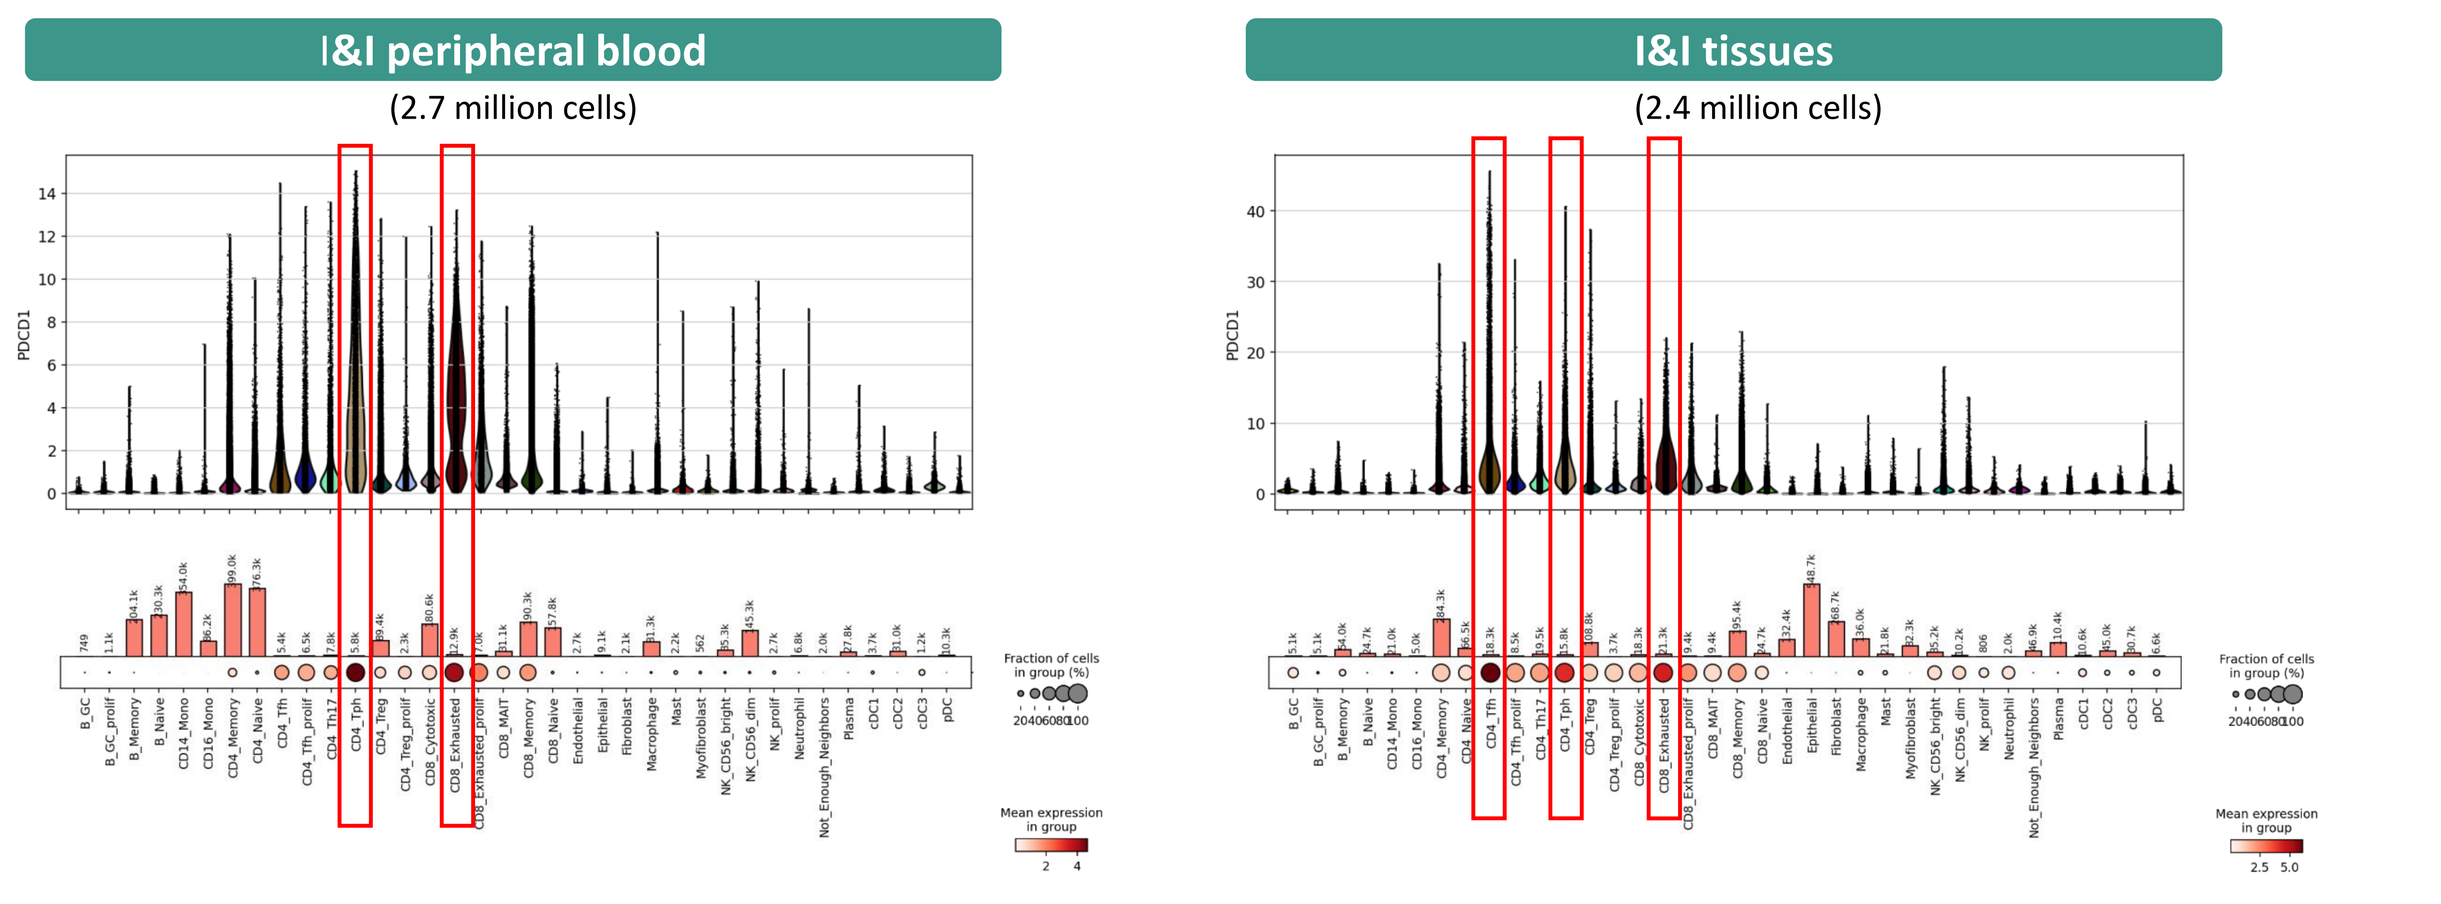
I&I peripheral blood
(2.7 million cells)
I&I tissues
(2.4 million cells)
PDCD1

Figure 1. Expression of PD-1 by immune cells in HiFiBiO's single-cell database from patients with autoimmune and inflammatory diseases. PD-1 is most highly expressed by T peripheral and follicular helper cells (Tph and Tfh), followed by exhausted/activated CD8 T cells.

Integrated autoimmune disease patient scRNA-Seq database

High PD-1 expression is associated with expression of the critical B cell helper cytokine IL-21

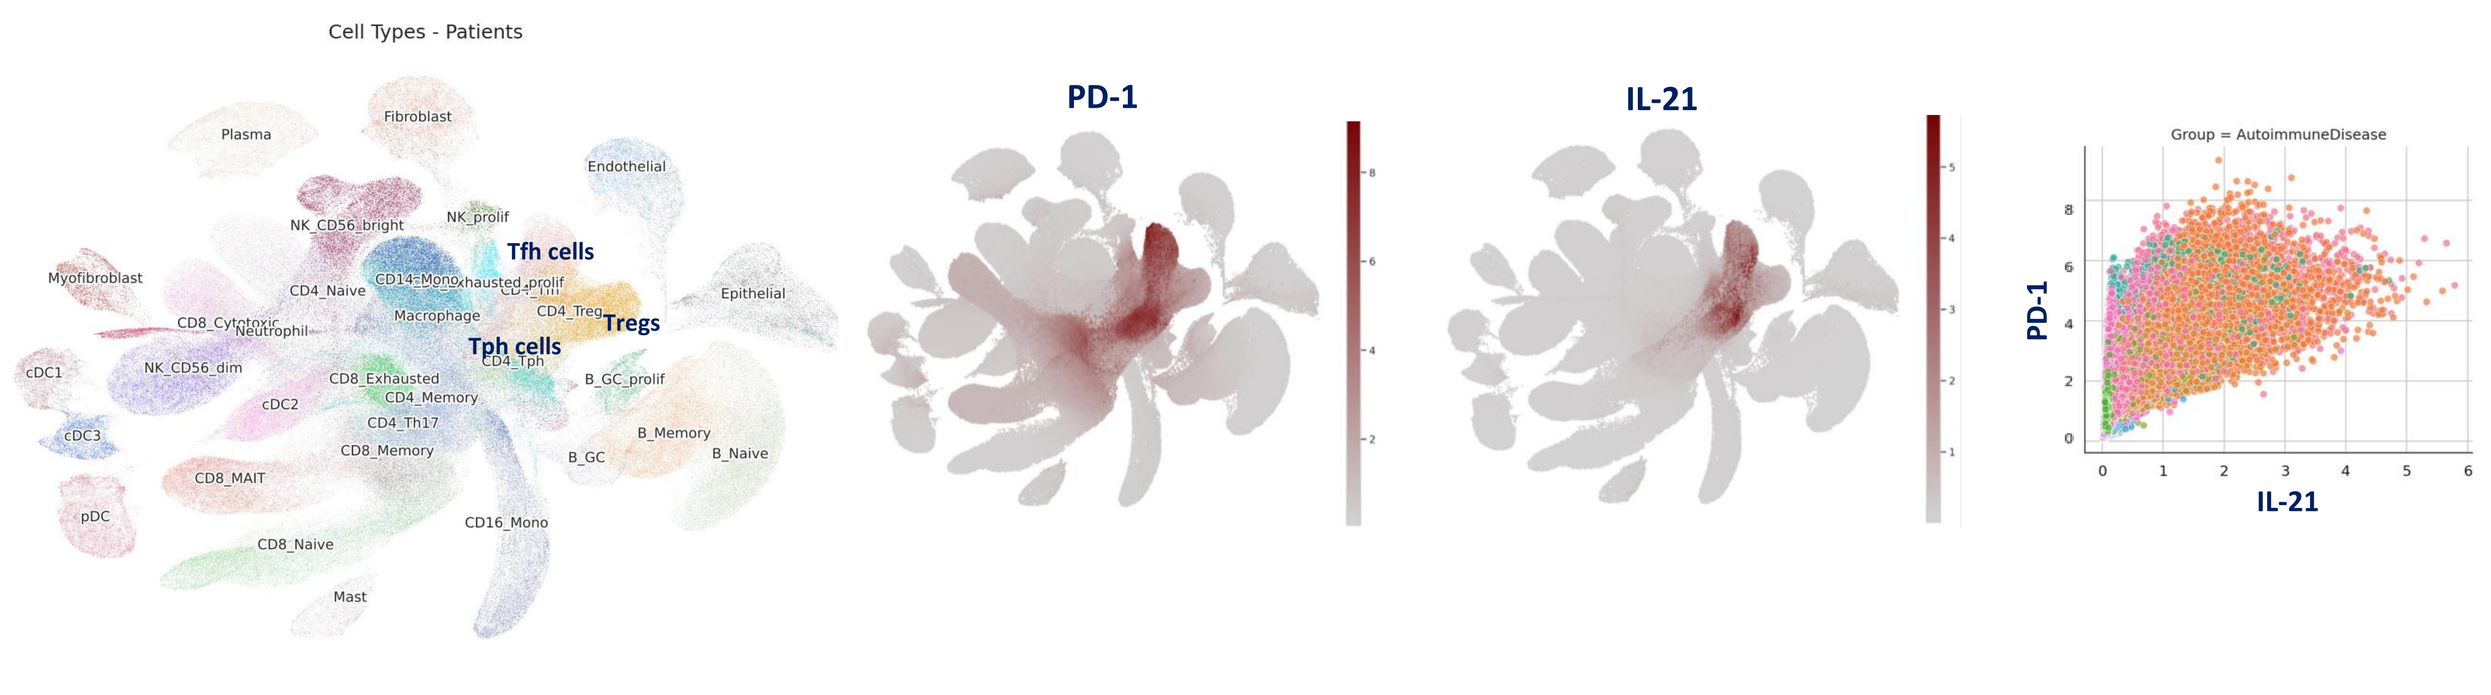
Cell Types - Patients
Tfh cells
Tph cells
Tregs
PD-1
IL-21
PD-1
IL-21

Figure 2. Association of PD-1 and IL-21 expression. Cells with the highest PD-1 expression also tend to express IL-21, which is critical for the maturation and development of B cells, in keeping with the role of Tph and Tfh cells in supporting autoreactive B cells in autoimmune diseases.

RESULTS | In Vitro Antibody Characterization

Binding and Blocking

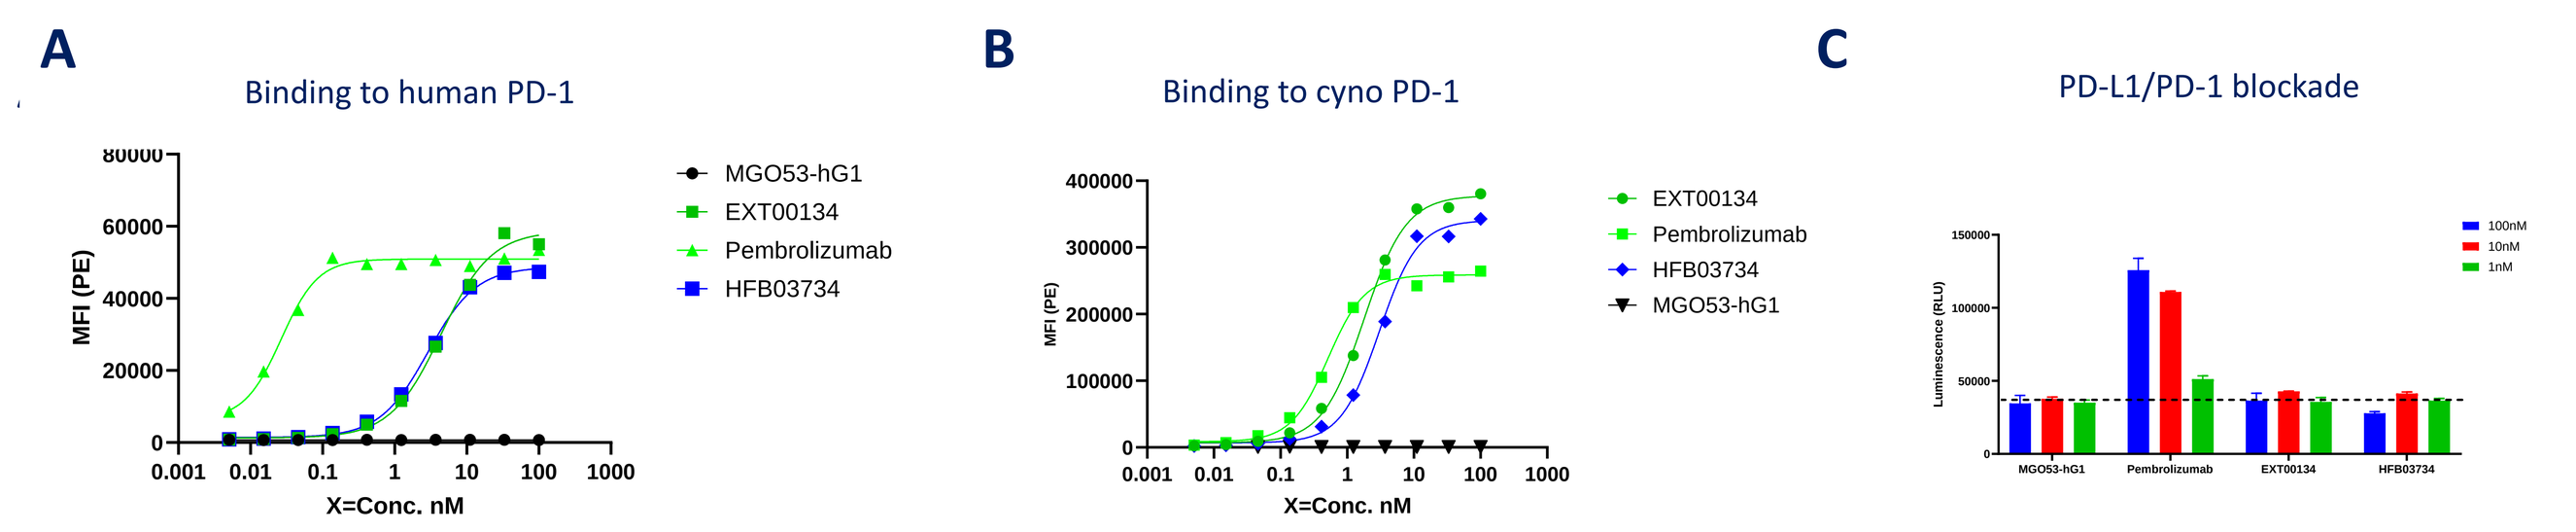
A
Binding to human PD-1
MGO53-hG1
EXT00134
Pembrolizumab
HFB03734
B
Binding to cyno PD-1
C
PD-L1/PD-1 blockade
X=Conc. nM

Figure 3. HFB03734 (HFB202501 parental antibody) binds to human and cyno PD-1 and does not block the PD-1/PD-L1 interaction. Binding of HFB03734 and selected comparators to **(A)** human and **(B)** cynomolgus monkey PD-1. **C.** Activity of HFB03734 and selected comparators in a PD-1 reporter assay. Unlike pembrolizumab, HFB03734 does not block the PD-1/PD-L1 interaction.

Agonist Activity

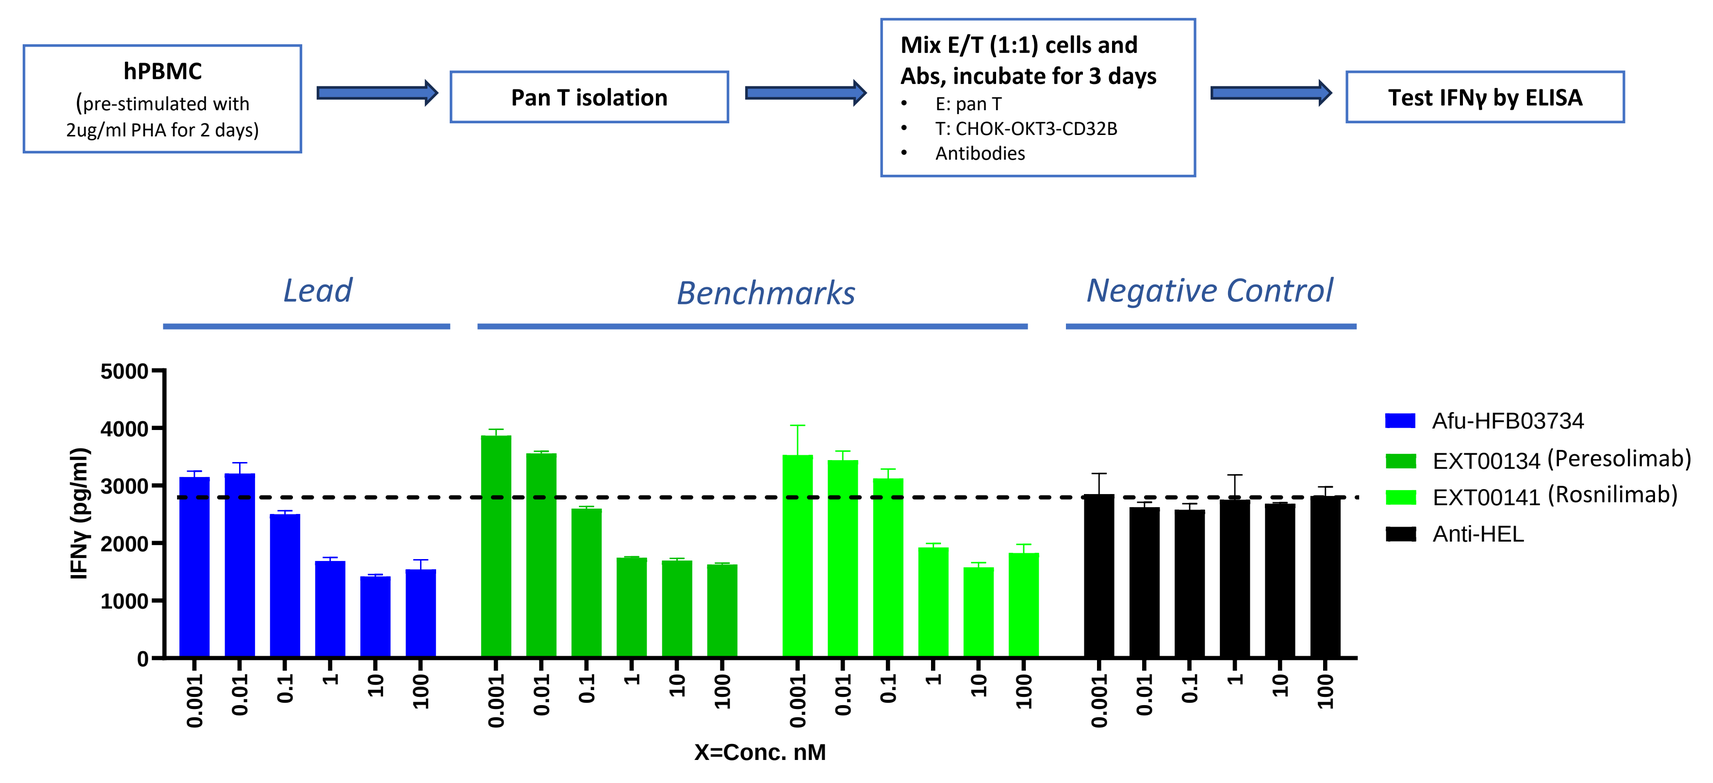
hPBMC (pre-stimulated with 2ug/ml PHA for 2 days)
Pan T isolation
Mix E/T (1:1) cells and Abs, incubate for 3 days
E: pan T
T: CHOK-OKT3-CD32B
Antibodies
Test IFNγ by ELISA
Lead
Benchmarks
Negative Control
Afu-HFB03734
EXT00134 (Peresolimab)
EXT00141 (Rosnilimab)
Anti-HEL
IFNγ (pg/ml)
X=Conc. nM

Figure 4. HFB03734 shows PD-1 agonist activity. T cells from healthy donor PBMCs were cultured with CHO cells expressing the anti-CD3 activating antibody OKT3 and the CD32B receptor to provide Fc receptor-mediated crosslinking and treated with HFB03734 (the HFB202501 parental antibody) or various comparator antibodies, and the ability of the antibodies to suppress IFNγ production through PD-1 agonism was measured. HFB03734 shows comparable PD-1 agonist activity to clinical-stage comparator antibodies.

ADCC Enhancement

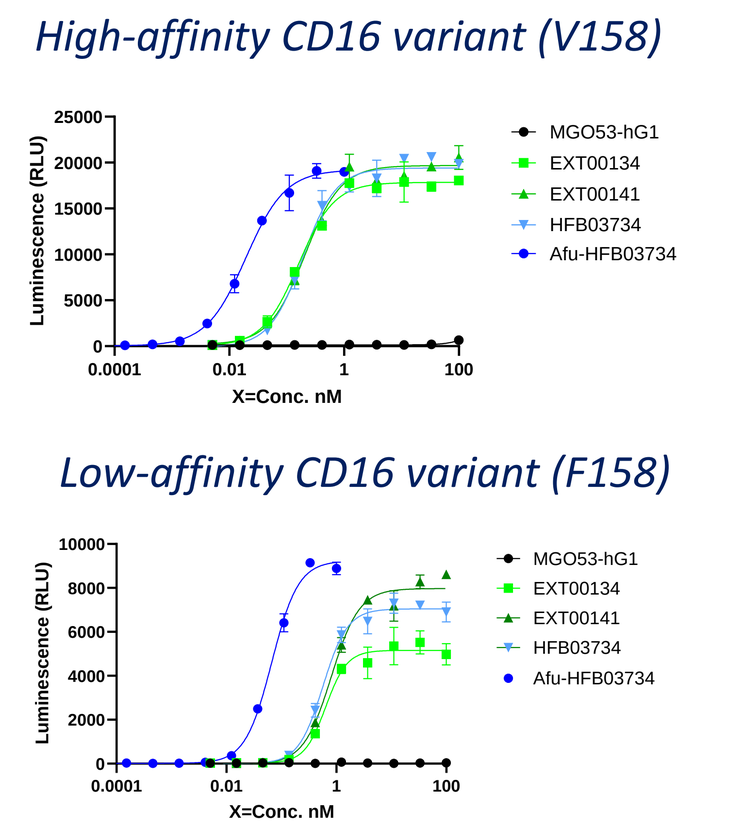
High-affinity CD16 variant (V158)
Low-affinity CD16 variant (F158)
MGO53-hG1
EXT00134
EXT00141
HFB03734
Afu-HFB03734
Luminescence (RLU)
X=Conc. nM

Figure 5. Afucosylation of the Fc region enhances the ADCC activity of HFB03734. Activity of standard and afucosylated versions of HFB03734 and selected standard comparator antibodies in ADCC reporter assays employing CD16 high-affinity *(top)* and low-affinity *(bottom)* variants are shown.

Primary Cell Killing

NK cell killing of SEA-activated human PD-1+ CD4+ T cells

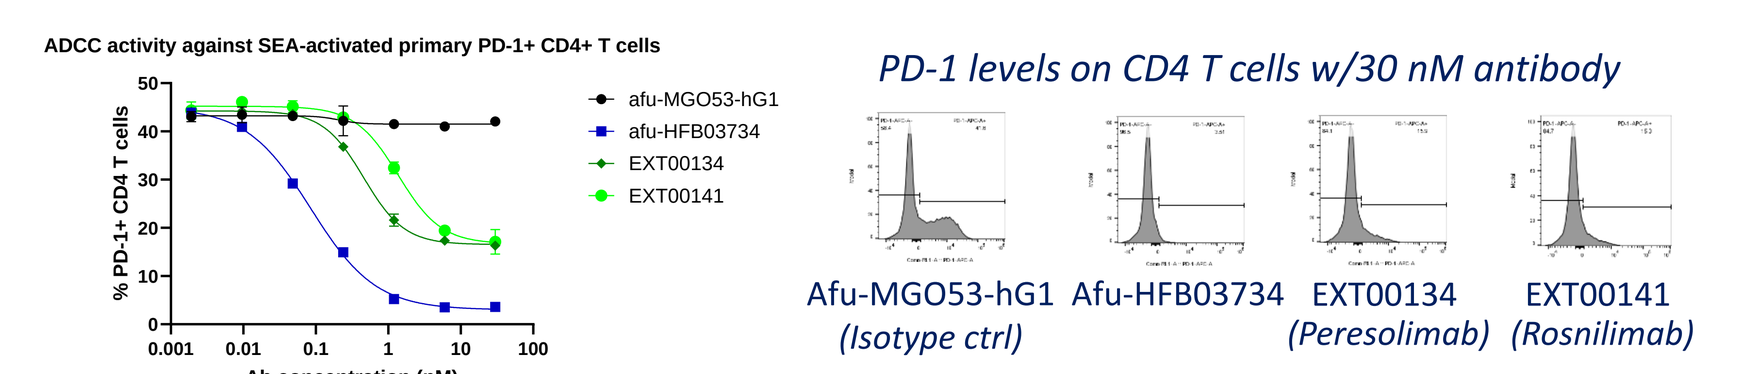
ADCC activity against SEA-activated primary PD-1+ CD4+ T cells
afu-MGO53-hG1
afu-HFB03734
EXT00134
EXT00141
Ab concentration (nM)
PD-1 levels on CD4 T cells w/30 nM antibody
Afu-MGO53-hG1 (Isotype ctrl)
Afu-HFB03734
EXT00134 (Peresolimab)
EXT00141 (Rosnilimab)

Antibody	EC50 (nM)	% PD-1+ Remaining
Afu-HFB03734	**0.09**	**3.0**
EXT00134 (Peresolimab)	0.46	16.5
EXT00141 (Rosnilimab)	1.37	16.7

Figure 6. Afucosylated HFB03734 (HFB202501 parental antibody) shows superior killing of activated PD-1+ T cells compared to clinical-stage comparators. Healthy donor PBMCs were stimulated with 100 ng/mL staphylococcal enterotoxin A (SEA) for 4 days and then incubated with activated NK cells at a ratio of 1:1 and selected antibodies for 6h. Live T cells were then quantified by flow cytometry to determine the extent of antibody-dependent cellular cytotoxicity (ADCC).

NK cell killing of T cells from primary human tonsils

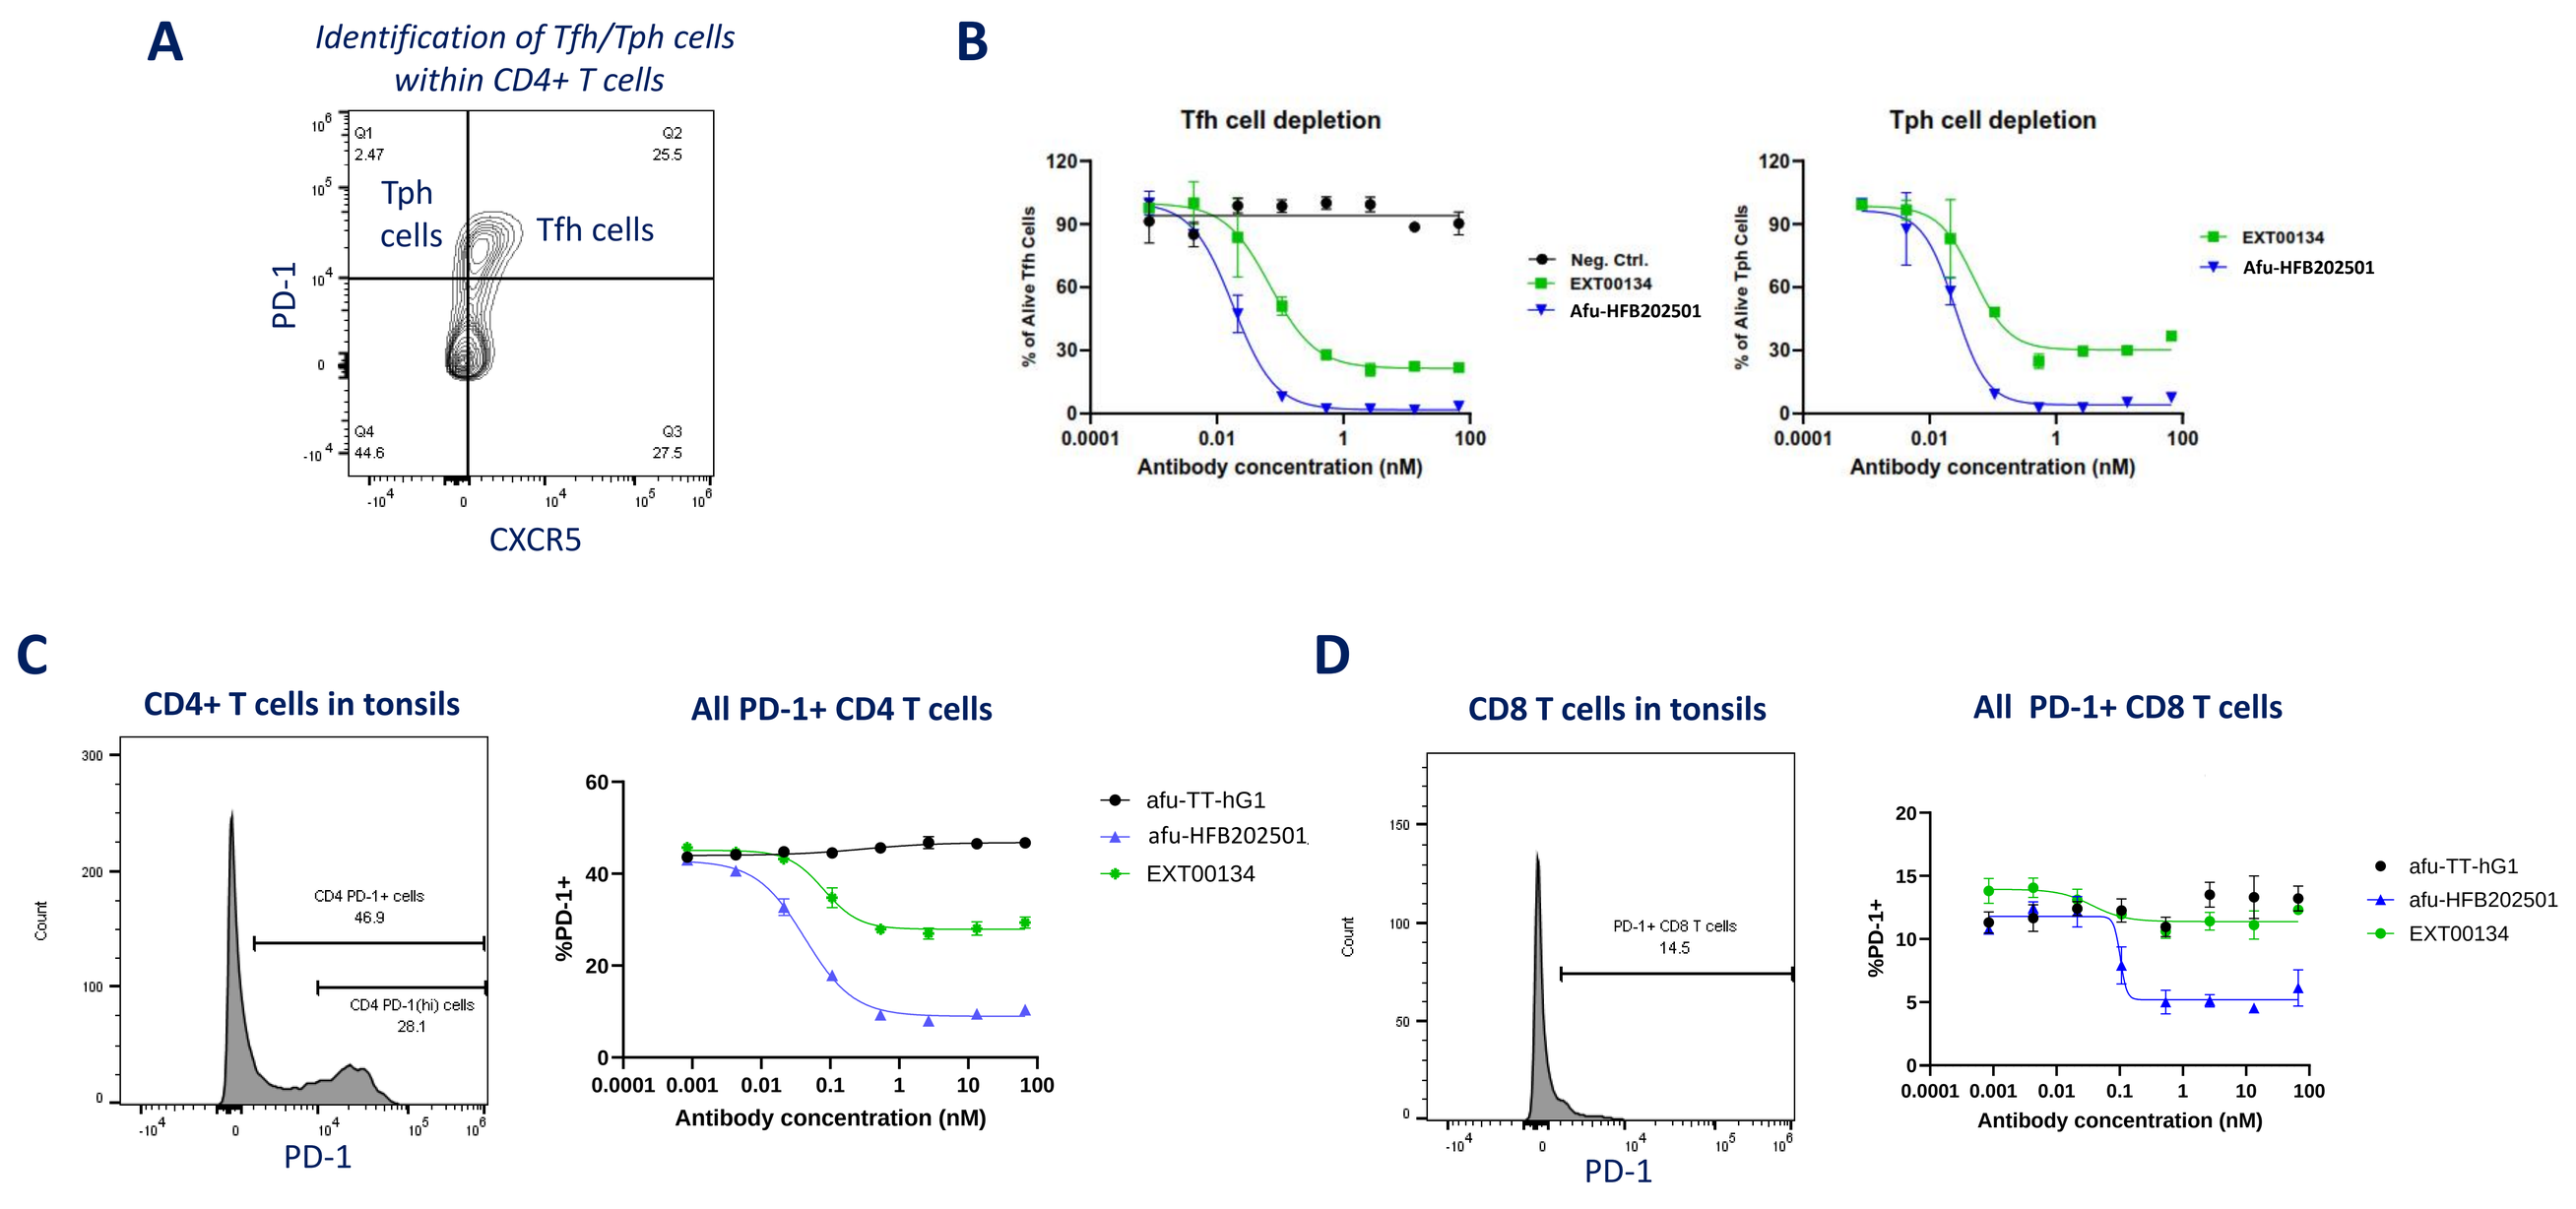
A
Identification of Tfh/Tph cells within CD4+ T cells
Tph cells
Tfh cells
PD-1
CXCR5
B
Tfh cell depletion
Neg. Ctrl.
EXT00134
Afu-HFB202501
Tph cell depletion
Antibody concentration (nM)
C
CD4+ T cells in tonsils
All PD-1+ CD4 T cells
afu-TT-hG1
afu-HFB202501
EXT00134
D
CD8 T cells in tonsils
All PD-1+ CD8 T cells
PD-1

Figure 7. Afucosylated HFB202501 shows superior killing of PD-1+ T cells from human tonsils compared to clinical-stage comparator. (A) Identification of PD-1(high) CXCR5+ CD4+ T follicular helper (Tfh) and PD-1(high) CXCR5- CD4+ T peripheral helper (Tph) cells by flow cytometry from primary human tonsils. **(B)** ADCC-mediated depletion of Tfh *(left)* or Tph *(right)* cells from human tonsils by peresolimab or afucosylated HFB202501. Dissociated primary human tonsil cells were incubated with activated NK cells and selected antibodies and Tfh and Tph cells were measured by flow cytometry. Depletion of all PD-1+ CD4+ T cells **(C)** or PD-1+ CD8+ T cells **(D)** is shown in the lower two panels. For reference, histograms of PD-1 levels on CD4+ and CD8+ cells treated with the isotype control antibody are shown.

Antigen Recall

Method

- CMV+ healthy donor PBMC (donor 7), 2e5 cells/well
- CMV peptide: 1µg/mL
- Test Ab: 10µg/ml (66.7 nM)
- 96h, collect supernatant, measure IFNγ by ELISA

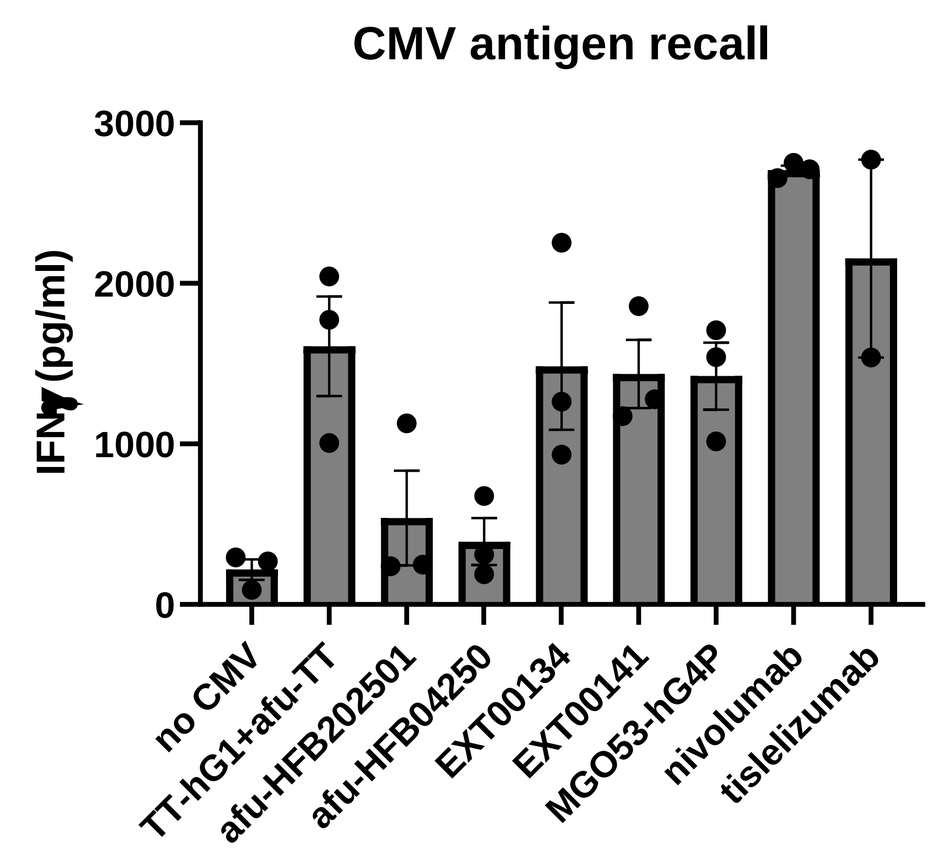
CMV antigen recall
IFNγ (pg/ml)
no CMV
TT-hG1+afu-TT
afu-HFB202501
afu-HFB04250
EXT00134
EXT00141
MGO53-hG4P
nivolumab
tislelizumab

Figure 8. Afucosylated HFB202501 shows more potent suppression of CMV antigen recall responses compared to clinical-stage comparators. CMV+ healthy donor PBMCs were stimulated with an immunogenic CMV peptide and treated with selected antibodies for 4 days, and supernatant was collected and IFNγ levels were measured. Afu-HFB202501 and afu-HFB04250 (another anti-PD-1 antibody in development) were able to potently suppress the CMV antigen recall response, while the PD-1/PD-L1 blocking antibodies nivolumab and tislelizumab enhanced the response.

Combined Suppressive and Depleting Effect on Activated T Cells

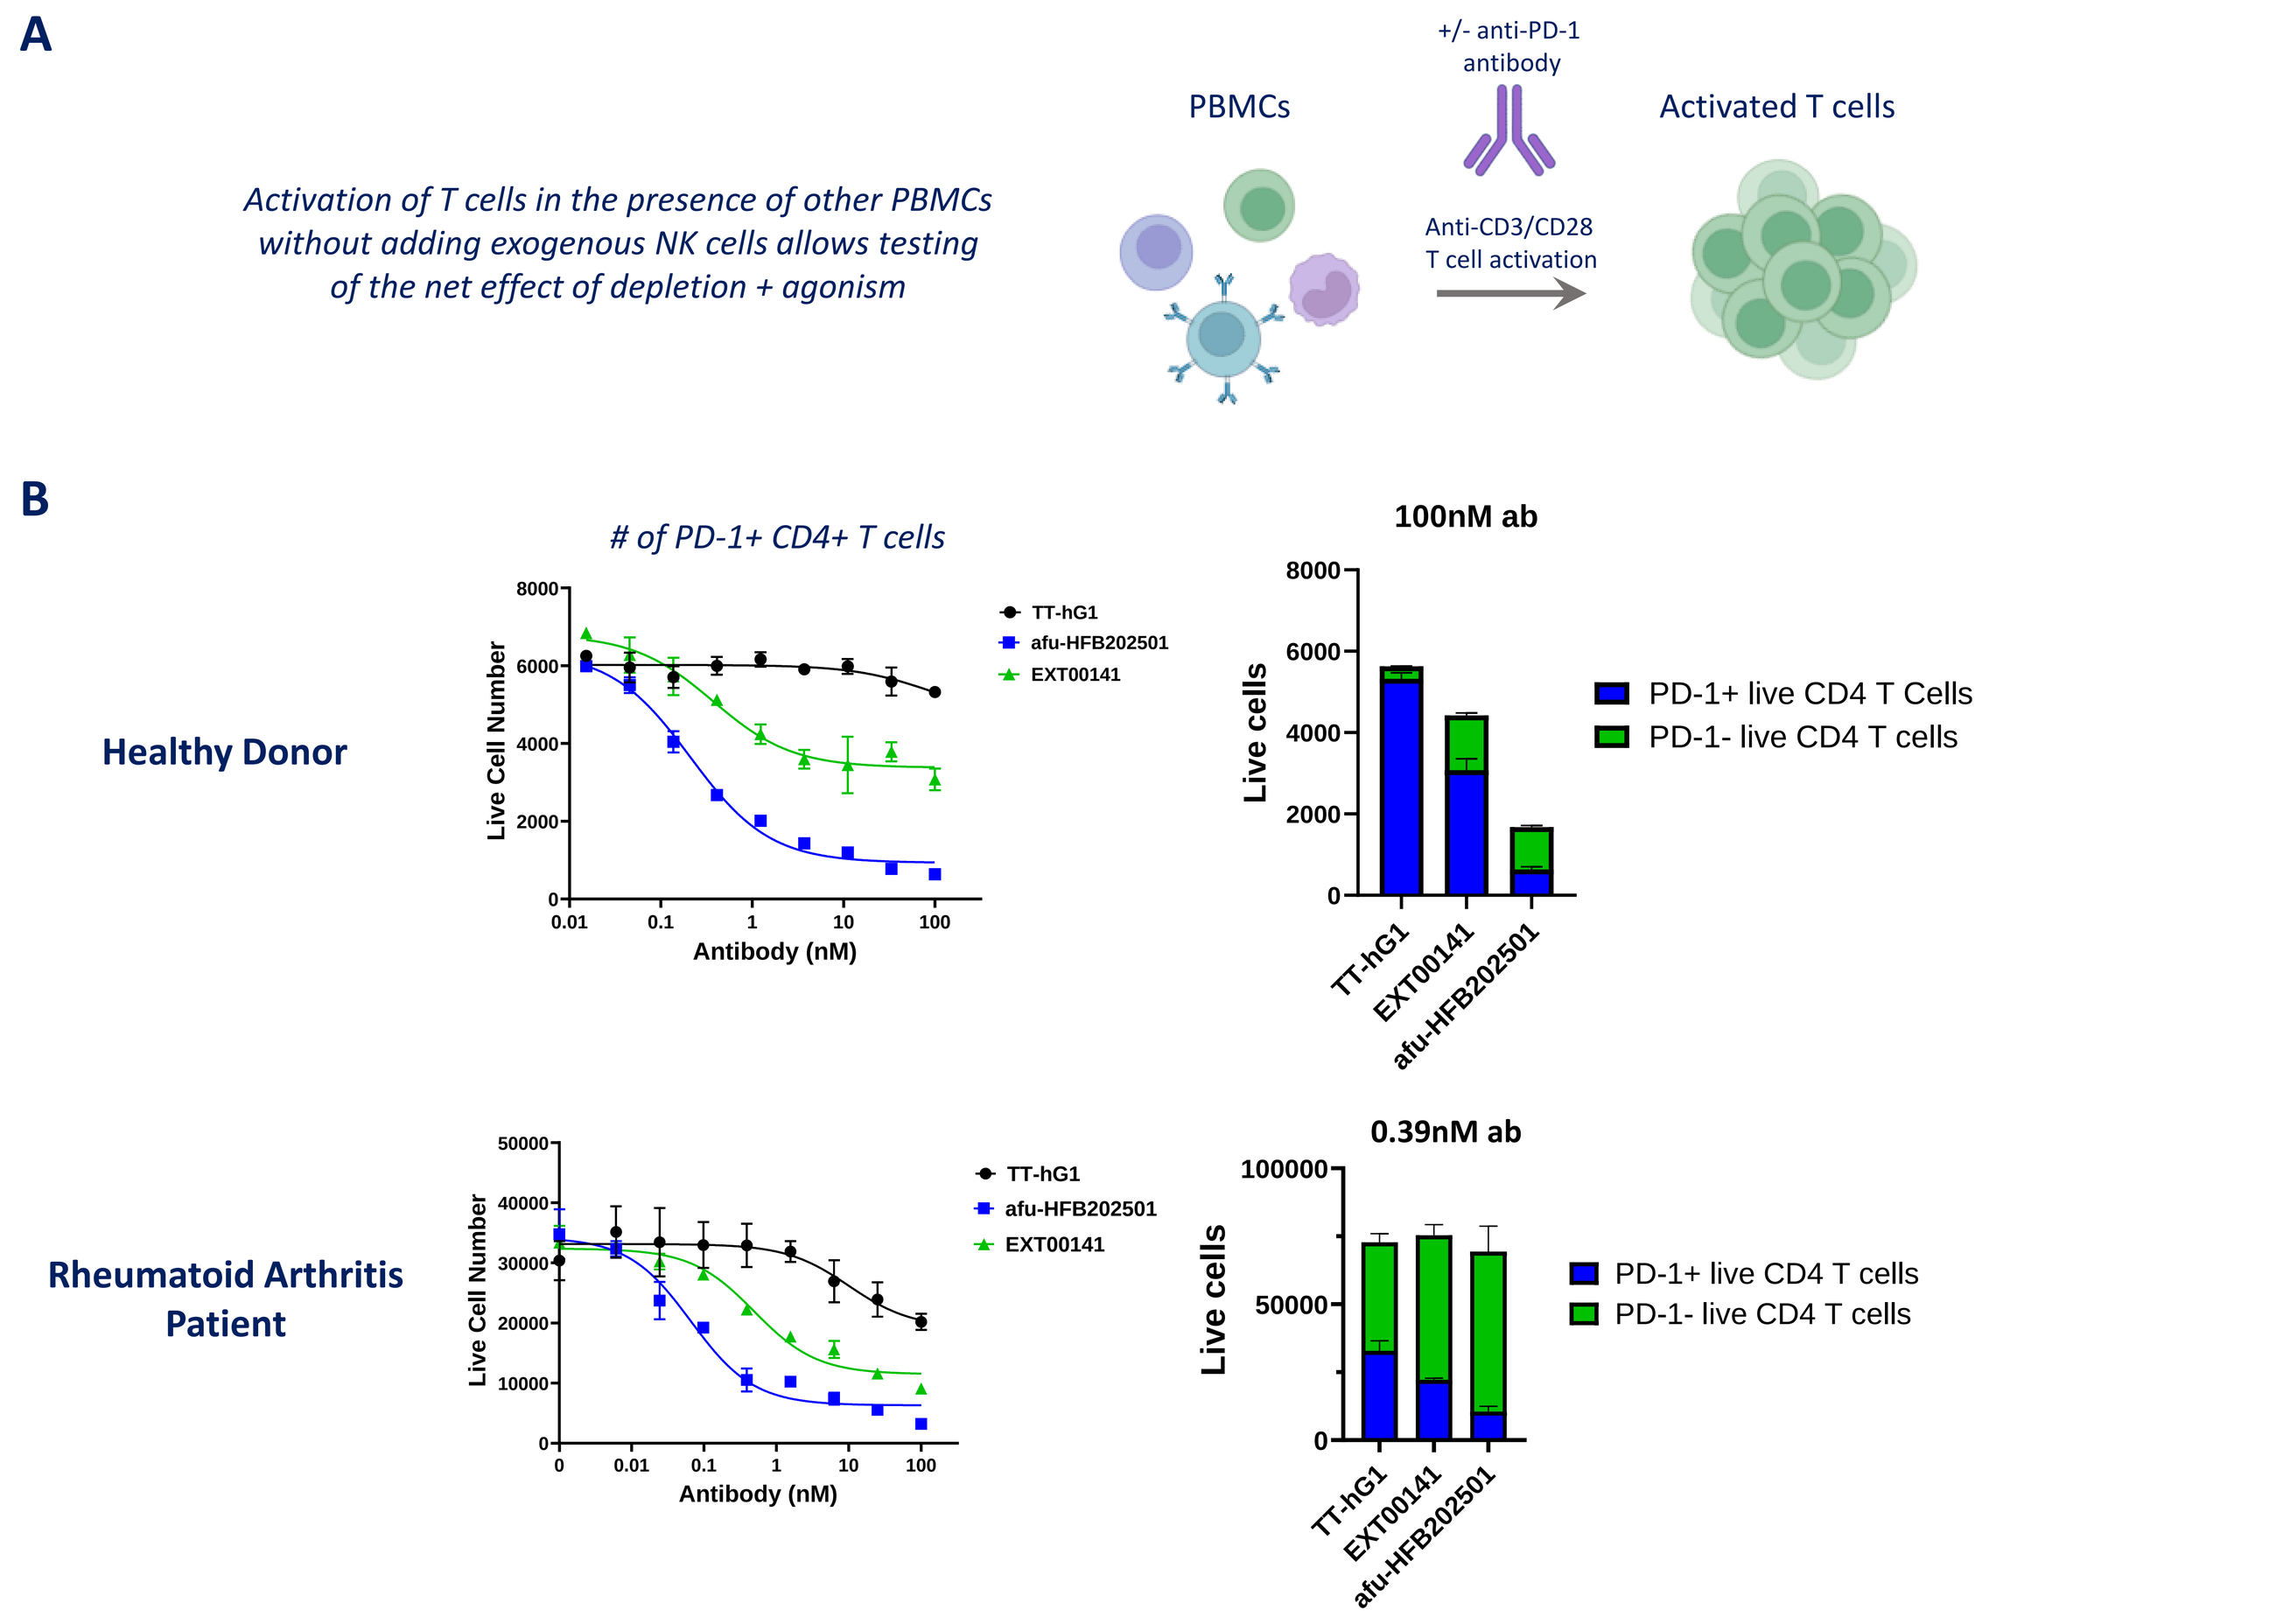
A
Activation of T cells in the presence of other PBMCs without adding exogenous NK cells allows testing of the net effect of depletion + agonism
PBMCs
+/- anti-PD-1 antibody
Anti-CD3/CD28 T cell activation
Activated T cells
B
of PD-1+ CD4+ T cells
Healthy Donor
TT-hG1
afu-HFB202501
EXT00141
Live Cell Number
Antibody (nM)
100nM ab
PD-1+ live CD4 T Cells
PD-1- live CD4 T cells
Live cells
Rheumatoid Arthritis Patient
0.39nM ab

Figure 9. HFB202501 shows a stronger net suppressive and depleting effect on activated T cells than a clinical-stage comparator antibody. (A) T cells from healthy donor or patient PBMCs are activated with anti-CD3 and anti-CD28 antibodies, and treated with selected anti-PD-1 antibodies or controls. PD-1 antibodies can mediate suppression of T cell activation through receptor agonism, or through ADCC or ADCP mediated by endogenous effector cells in the PBMCs. **(B)** HFB202501 shows more potent control of both CD4+ T cell numbers and the proportion of PD-1+ CD4+ T cells in the population compared to rosnilimab, indicating a stronger net suppressive and depleting effect.

RESULTS | In Vivo Efficacy in a Mouse Model of Lupus

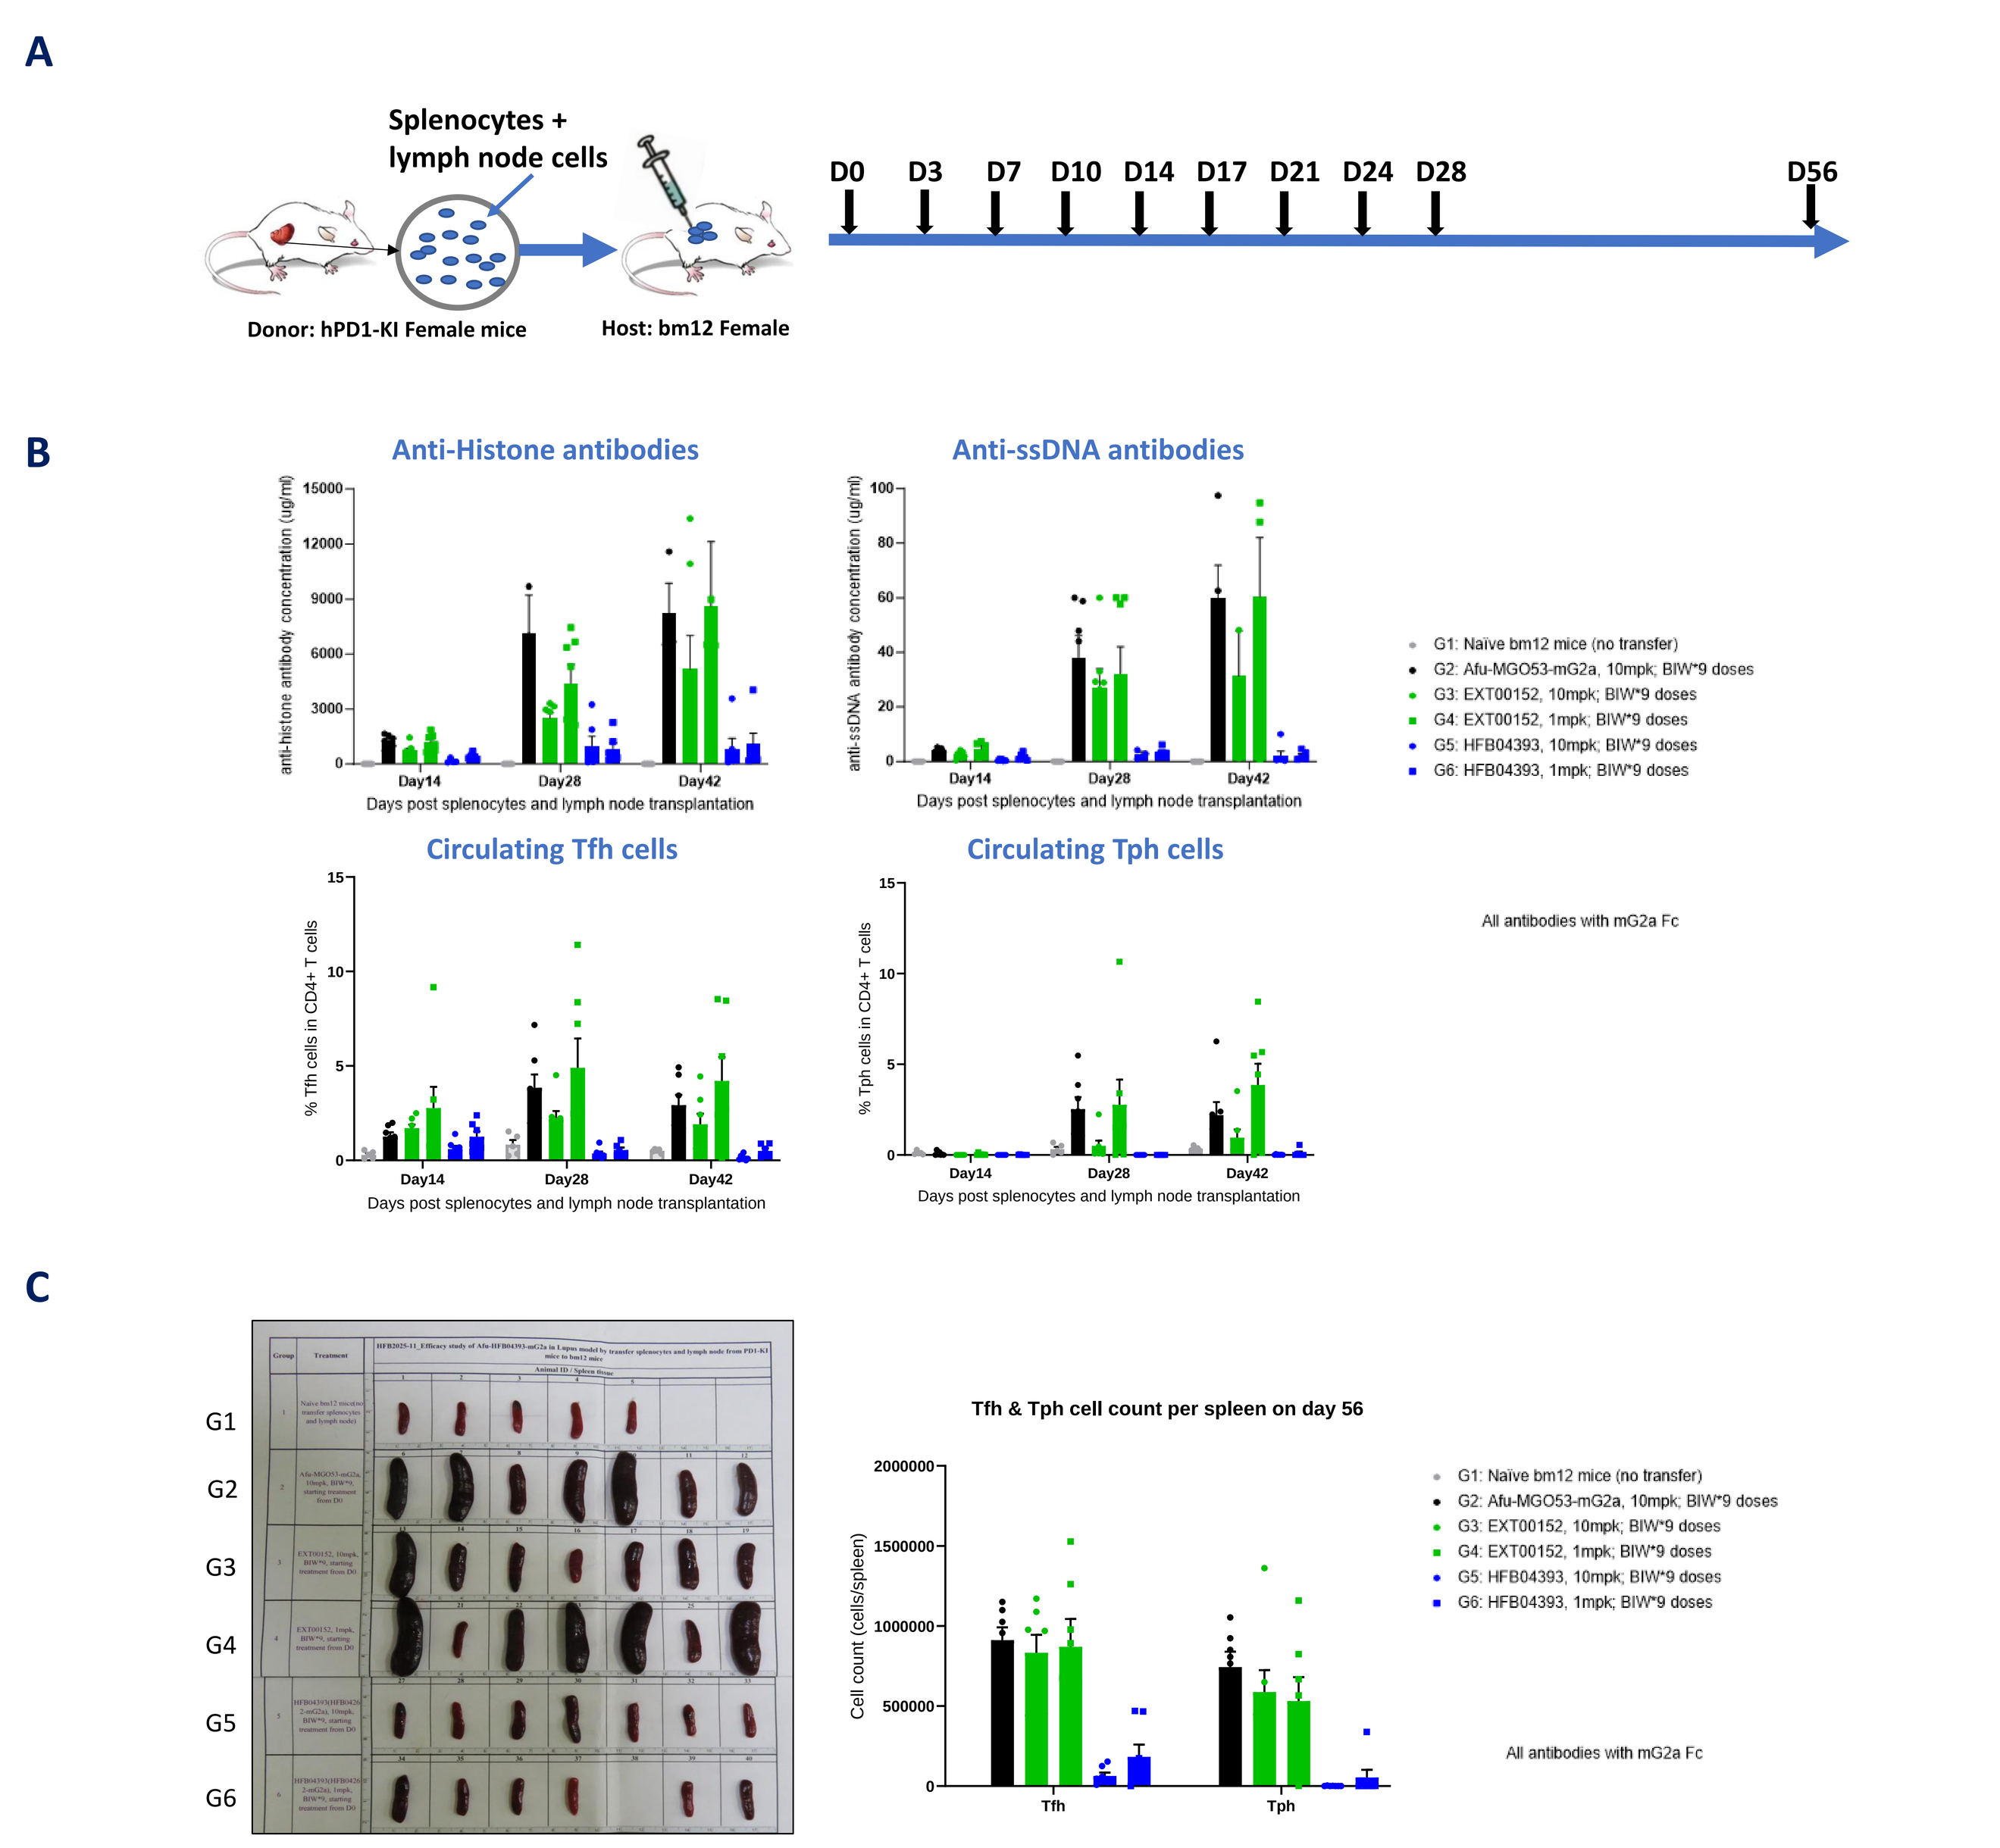
A
Splenocytes + lymph node cells
Donor: hPD1-KI Female mice
Host: bm12 Female
D0
D3
D7
D10
D14
D17
D21
D24
D28
D56
B
Anti-Histone antibodies
Anti-ssDNA antibodies
Circulating Tfh cells
Circulating Tph cells
Days post splenocytes and lymph node transplantation
G1: Naive bm12 mice (no transfer)
G2: Afu-MGO53-mG2a, 10mpk; BIW*9 doses
G3: EXT00152, 10mpk; BIW*9 doses
G4: EXT00152, 1mpk; BIW*9 doses
G5: HFB04393, 10mpk; BIW*9 doses
G6: HFB04393, 1mpk; BIW*9 doses
All antibodies with mG2a Fc
C
G1
G2
G3
G4
G5
G6
Tfh & Tph cell count per spleen on day 56
Cell count (cells/spleen)
Tfh
Tph

Figure 10. HFB202501-mG2a (HFB04393) shows potent disease control in a mouse model of lupus. (A) Disease model and study design. Splenocytes and lymph node cells from female hPD-1 knock-in mice are transplanted into wild-type female bm12 mice, resulting in a lupus-like graft-versus-host disease characterized by autoantibodies and splenomegaly supported by donor-derived hPD-1+ Tph and Tfh cells. **(B)** Control of autoantibodies *(top row)* and circulating Tfh and Tph cells *(bottom row)* by anti-PD-1 or control antibodies. HFB2020501-mG2a shows more potent control of autoantibodies and circulating Tph/Tfh cell levels as compared to rosnilimab-mG2a (EXT00152). **(C)** Size of spleens *(left)* and number of splenic Tfh and Tph cells *(right)* from treated mice at the time of sacrifice (day 56 of the study). HFB202501-mG2a is able to prevent splenomegaly more effectively than rosnilimab-mG2a (EXT00152).

CONCLUSION

We have discovered and are developing a PD-1 agonist antibody with enhanced depleting activity, HFB202501, with best-in-class potential. Enhanced depletion allows for more effective precision targeting of pathogenic Tfh and Tph cells expressing high levels of PD-1, as well as PD-1+ activated effector CD8+ T cells expressing lower levels of PD-1. PD-1 agonists with depleting activity have demonstrated clinical efficacy in rheumatoid arthritis. Given the more potent activity of HFB202501 as compared to these clinical-stage anti-PD-1 antibodies in multiple preclinical models and the potential for depletion of pathogenic PD-1+ T cells to reset the immune response and restore tolerance, HFB202501 is a promising novel treatment for autoantibody-mediated diseases and other autoimmune and inflammatory conditions mediated by PD-1+ T cells.

REFERENCES

1. F Ramirez-Valle *et al.*, 'Sequential immunotherapy: Towards cures for autoimmunity.' *Nat Rev Drug Disc* https://doi.org/10.1038/s41573-024-00959-8.
2. S Grebinoski and DA Vignali. 'Inhibitory receptor agonists: the future of autoimmune disease therapeutics?' *Curr Op Immunol* (2020) **67:** 1–9.
3. K Suzuki *et al.*, 'Anti-PD-1 antibodies recognizing the membrane-proximal region are PD-1 agonists that can down-regulate inflammatory diseases.' *Sci Immunol* (2023) **8:** eadd4947.
4. 'Here come the PD-1 agonists.' (2023) https://www.evaluate.com/node/18854/amp
5. J Tuttle *et al.,* 'A phase 2 trial of peresolimab for adults with rheumatoid arthritis.' *N Engl J Med* (2023) **388:** 1853–1862.
6. AnaptysBio corporate presentation: Rosnilimab updated Phase 2b data in RA (https://ir.anaptysbio.com/static-files/6cd574c5-b4fa-42f8-873f-93e9af2de251).

HiFiBiO THERAPEUTICS

For additional information, please email contact@hifibio.com or visit Hifibio.com