Presentation # 1011P
Presented at European Society for Medical Oncology
September 13 - 17, 2024
Barcelona, Spain

HiFiBiO THERAPEUTICS

Model-informed dose optimization of HFB200301, a TNFR2 agonist monoclonal antibody, in monotherapy and in combination with the anti-PD-1 tislelizumab in patients with advanced solid tumors

Desamparados Roda[1], Jon Zugazagoitia[2], Elena Garralda[3], Alexander I. Spira[4], Anthony B. El-Khoueiry[5], Yujie Zhao[6], Peter J. Oppelt[7], Xi Lin[8], Shaozhen Xie[8], Margaret E. Chen[8], Jinping Gan[8], Gabrielle Wong[8], Jack Russella-Pollard[8], William Hedrich[8], John Pallante[8], Spencer Huggett[8], Francisco Adrian[8], Liang Schweizer[8], Xiaorong Shi[8] and Robert H.I. Andtbacka[8]

Hospital Clínico Universitario de Valencia[1], Hospital Universitario 12 de Octubre[2], Vall d'Hebron Institute of Oncology[3], NEXT Oncology Virginia[4], University of Southern California Norris Comprehensive Cancer Center[5], Mayo Clinic Florida[6], Washington University School of Medicine in St. Louis[7], HiFiBiO Inc, Cambridge, MA USA[8] , Corresponding Author*

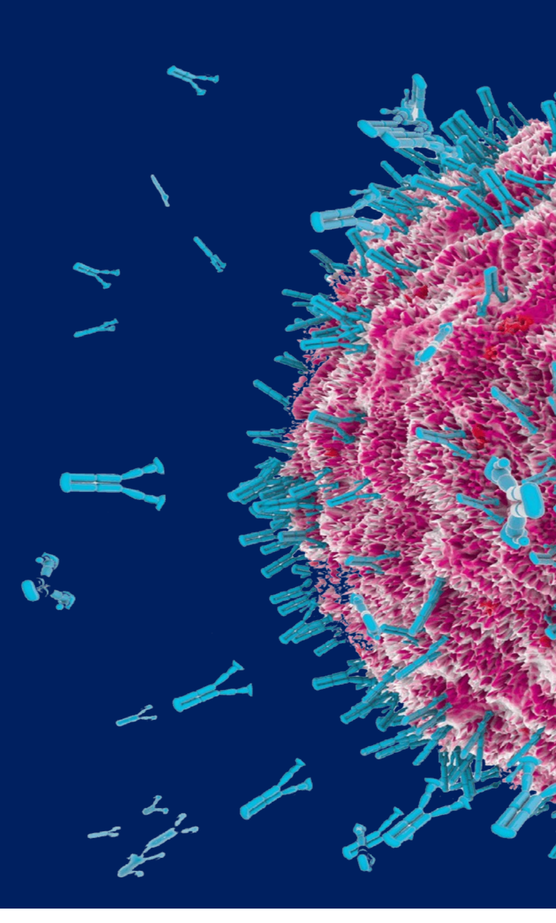

BACKGROUND

- Agonism of tumor necrosis factor receptor-2 (TNFR2) enhances anti-tumor immunity by stimulating T- and NK-cells in the tumor microenvironment.
- HFB200301, a first-in-class anti-TNFR2 agonistic monoclonal antibody, triggers both innate and adaptive immune responses.
- In our Phase I Dose Escalation trial, we explored a Q4W dosing regimen of HFB200301, involving 27 patients receiving monotherapy and 12 patients in combination with tislelizumab (TIS). This regimen demonstrated a tolerable safety profile and showed signs of clinical activity[1].
- Determining the optimal dosing regimen for an agonist requires balancing safety and efficacy. While preclinical evaluation suggested Q4W dosing would provide a suitable regimen for efficacy, based on the assessment of the Q4W data, we hypothesized that more frequent dosing of HFB200301 could enhance agonistic effects and improve clinical activity.
- To test this hypothesis, we used preliminary pharmacokinetic (PK), target engagement (TE), and pharmacodynamic (PD) data from the Q4W monotherapy arm to construct models and simulate various dosing regimens, aiming to optimize HFB200301 exposure and target engagement.
- We also present the initial clinical data from our ongoing Phase I trial of HFB200301 Q2W in monotherapy and in combination with TIS in advanced refractory solid tumors (NCT05238883).

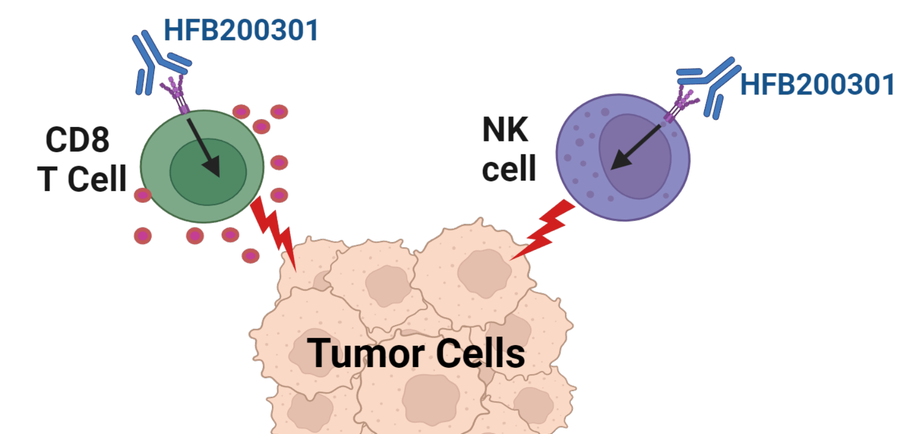
HFB200301
HFB200301
CD8 T Cell
NK cell
Tumor Cells

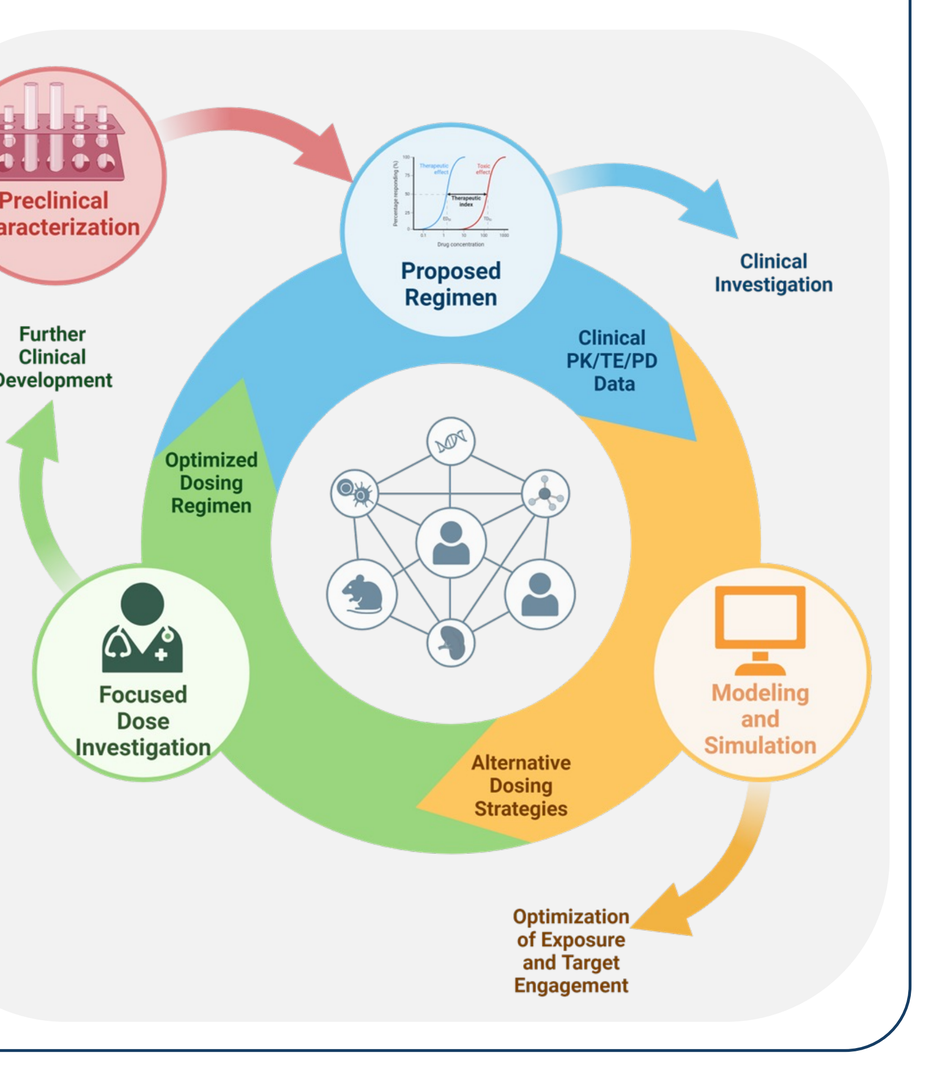
Preclinical Characterization
Proposed Regimen
Clinical Investigation
Clinical PK/TE/PD Data
Further Clinical Development
Optimized Dosing Regimen
Focused Dose Investigation
Modeling and Simulation
Alternative Dosing Strategies
Optimization of Exposure and Target Engagement

OBJECTIVES and STUDY DESIGN

Primary Objectives
- Safety and tolerability of HFB200301 in monotherapy and in combination with TIS

Secondary Objectives
- Assess PK, PD, and immunogenicity of HFB200301
- Establish RDE and RP2D
- Examine preliminary anti-tumor efficacy, ORR using RECIST 1.1, iRECIST, and mRECIST for mesothelioma

Exploratory Objectives
- Establish Proof of Mechanism (POM) in paired tumor biopsies and peripheral blood
- Generate biomarker hypothesis for patient enrichment

Key Eligibility Criteria
- Adult patients with advanced or metastatic solid tumors. Tumor types include:
 - cervical cancer
 - EBV+ gastric cancer
 - head and neck squamous cell carcinoma
 - melanoma
 - pleural mesothelioma
 - non-small cell lung cancer
 - renal cell carcinoma
 - sarcoma
 - testicular germ cell tumor
- Measurable disease - RECIST 1.1 or mRECIST
- ECOG PS 0-1
- Patient must have exhausted standard lines of systemic therapy*

**Other protocol-defined inclusion criteria may apply*

Q4W study design

Monotherapy HFB200301 Q4W (n=27)

DL 5 HFB (n=4)
DL 4 HFB (n=3)
DL 3 HFB (n=5)
DL 2 HFB (n=8)
DL 1 HFB (n=7)

Combination HFB200301 Q4W + tislelizumab Q4W (n=12)

DL 4 HFB + TIS (n=3)
DL 3 HFB + TIS (n=3)
DL 2 HFB + TIS (n=6)

DL (Dose Level); HFB (HFB200301); Q4W (once every 4 weeks); TIS (tislelizumab)

MODELING and SIMULATION

HFB200301 model structure and fitting

PK of HFB200301 is well-described by a two-compartment model with parallel elimination via linear and non-linear clearance. Peripheral and in-tumor RO are described by direct effect models while sTNFR2 is captured by an indirect response model.

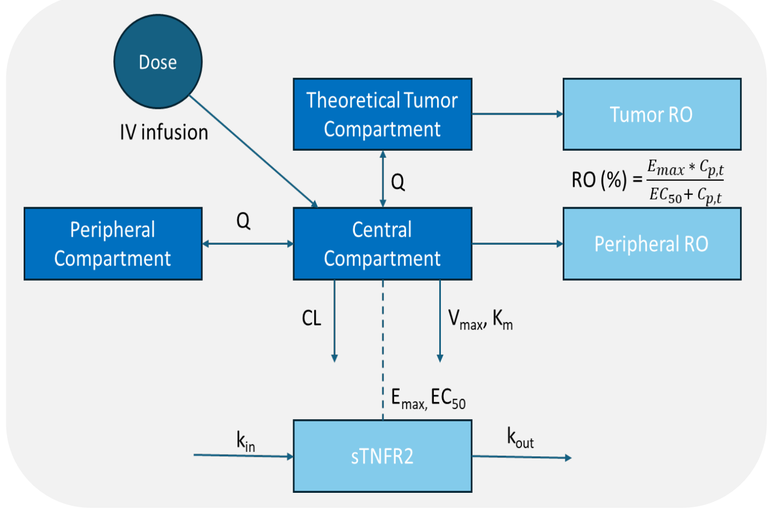
Dose
IV infusion
Theoretical Tumor Compartment
Tumor RO
Q
Peripheral Compartment
Central Compartment
Peripheral RO
CL
sTNFR2

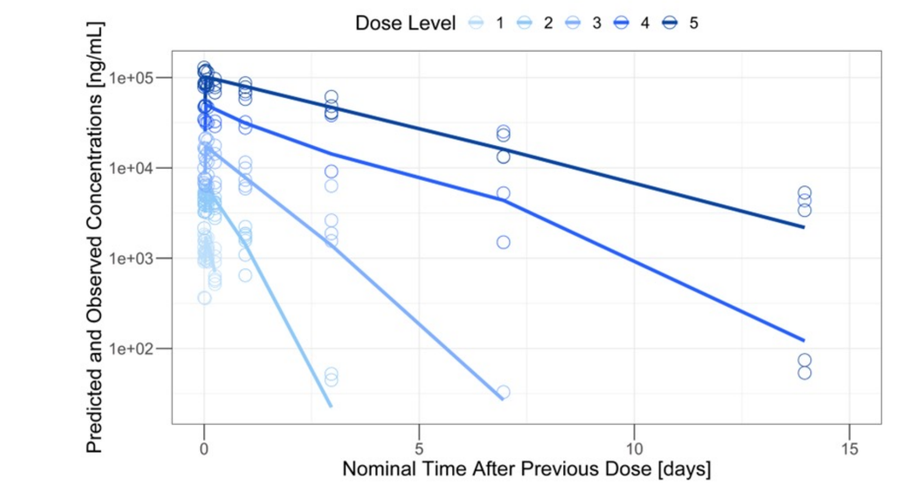
Dose Level 1 2 3 4 5
Predicted and Observed Concentrations [ng/mL]
Nominal Time After Previous Dose [days]

MODEL-INFORMED REGIMEN OPTIMIZATION

Schedule optimization with simulated PK and RO

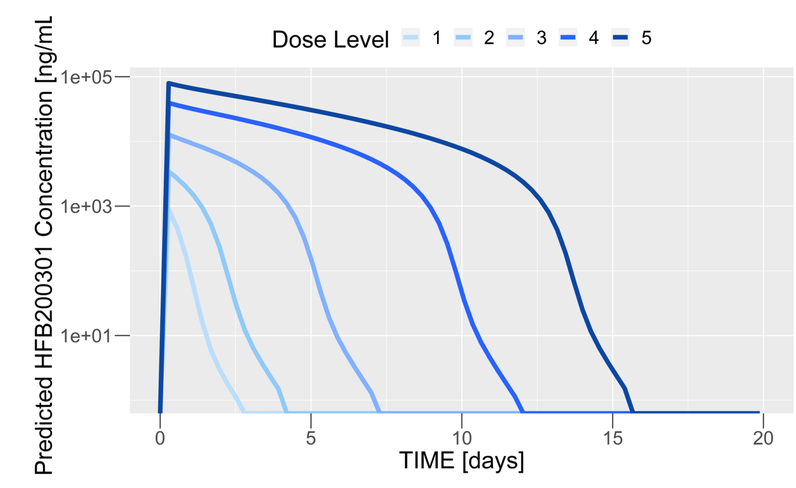
A
Dose Level 1 2 3 4 5
Predicted HFB200301 Concentration [ng/mL]
TIME [days]

A. The fitted PK model of HFB200301 was used to simulate exposures with alternative regimens to explore optimal administration

B. Observed peripheral RO data (shown in open circles) was used to fit a direct response model of systemic (solid line) and on-target (dashed line) RO, demonstrating increased target engagement with more frequent dosing of HFB200301

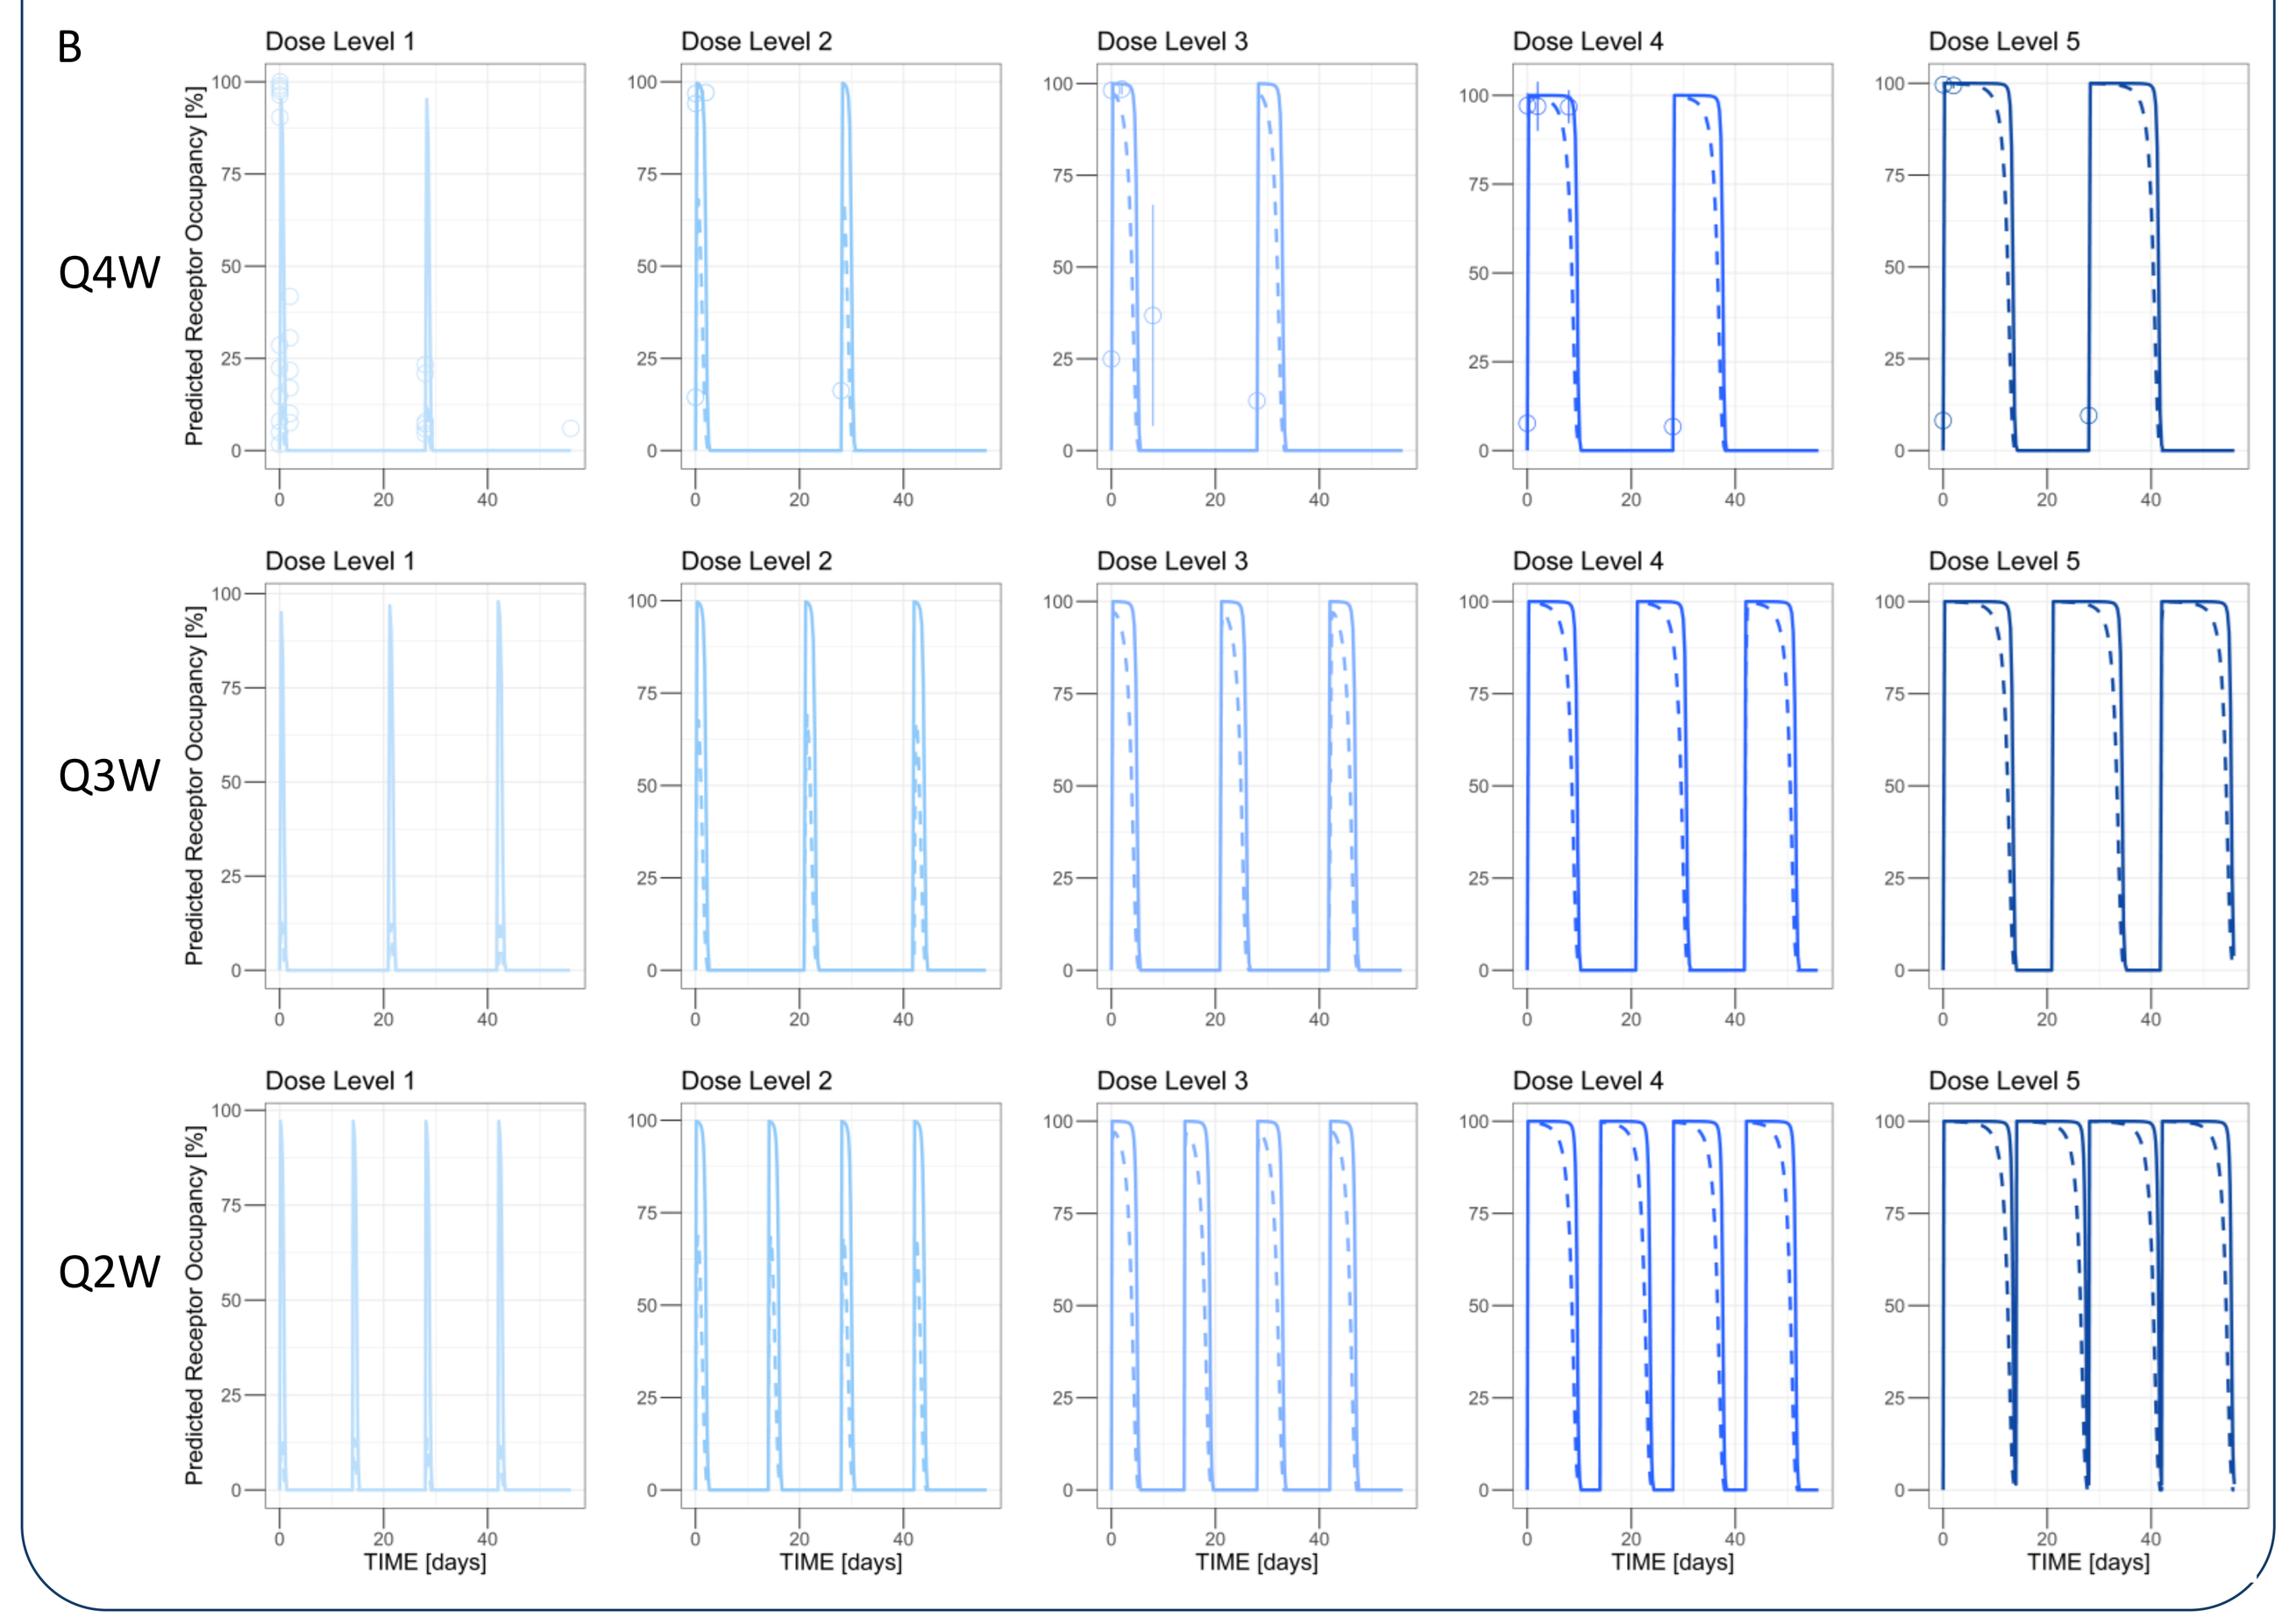
B
Q4W
Q3W
Q2W
Dose Level 1
Dose Level 2
Dose Level 3
Dose Level 4
Dose Level 5
Predicted Receptor Occupancy [%]
TIME [days]

Dose optimization with peripheral PD markers

A. Observed (points) and predicted (lines) sTNFR2 levels in serum following a single dose of HFB200301 demonstrate exposure-dependent target engagement with each administration.

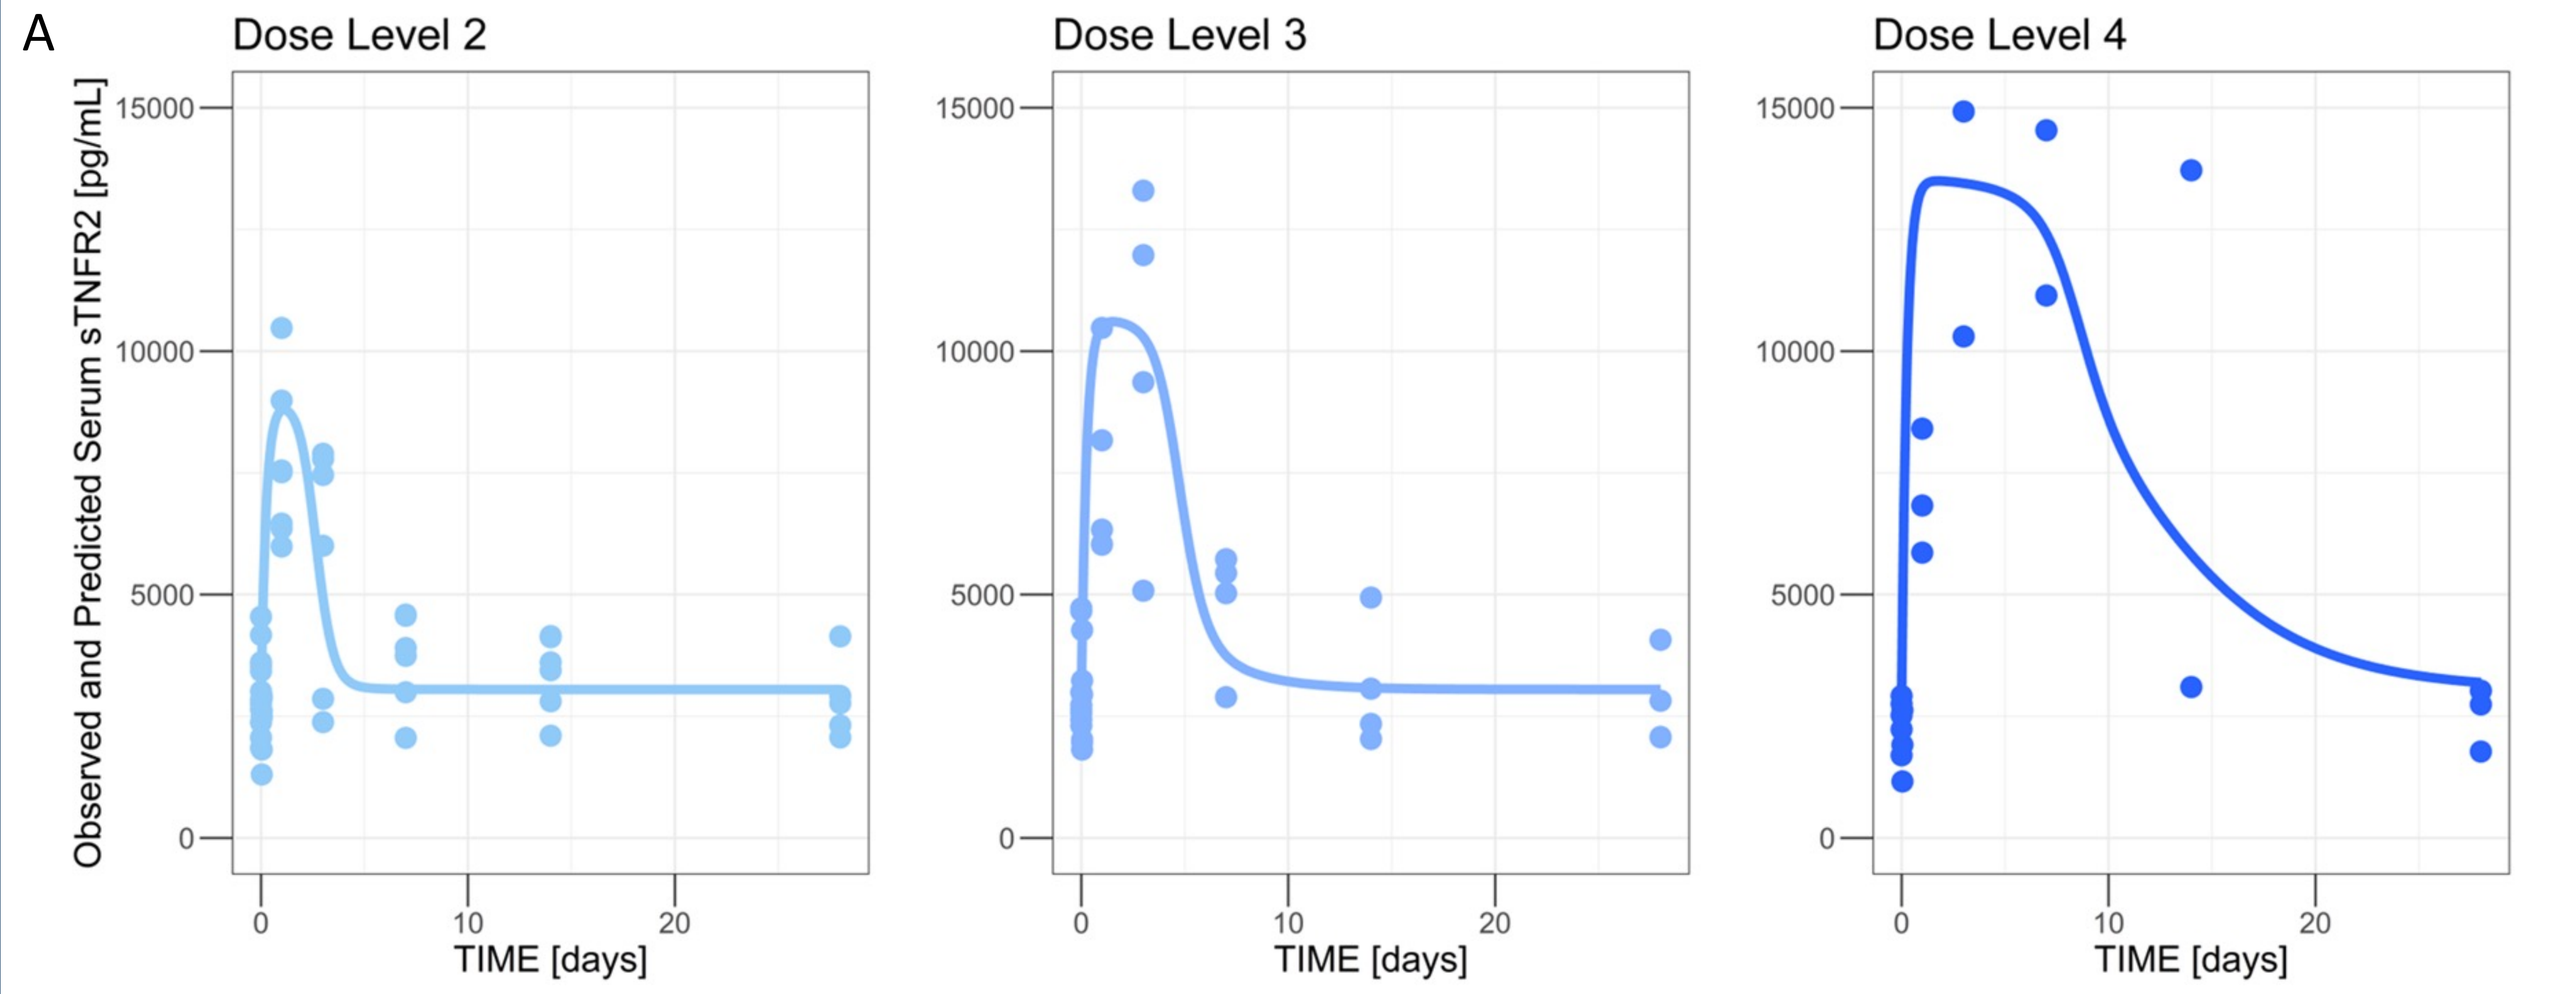
A
Dose Level 2
Dose Level 3
Dose Level 4
Observed and Predicted Serum sTNFR2 [pg/mL]
TIME [days]

B. Peripheral single-cell RNA-sequencing data demonstrated modulation of TNFα RNA expression following a single dose of HFB200301 in relevant immune cell types. Dose-dependent increases in TNFα expression were observed in CD8+ T and NK cell subsets, indicating on-mechanism immune cell activation in the periphery.

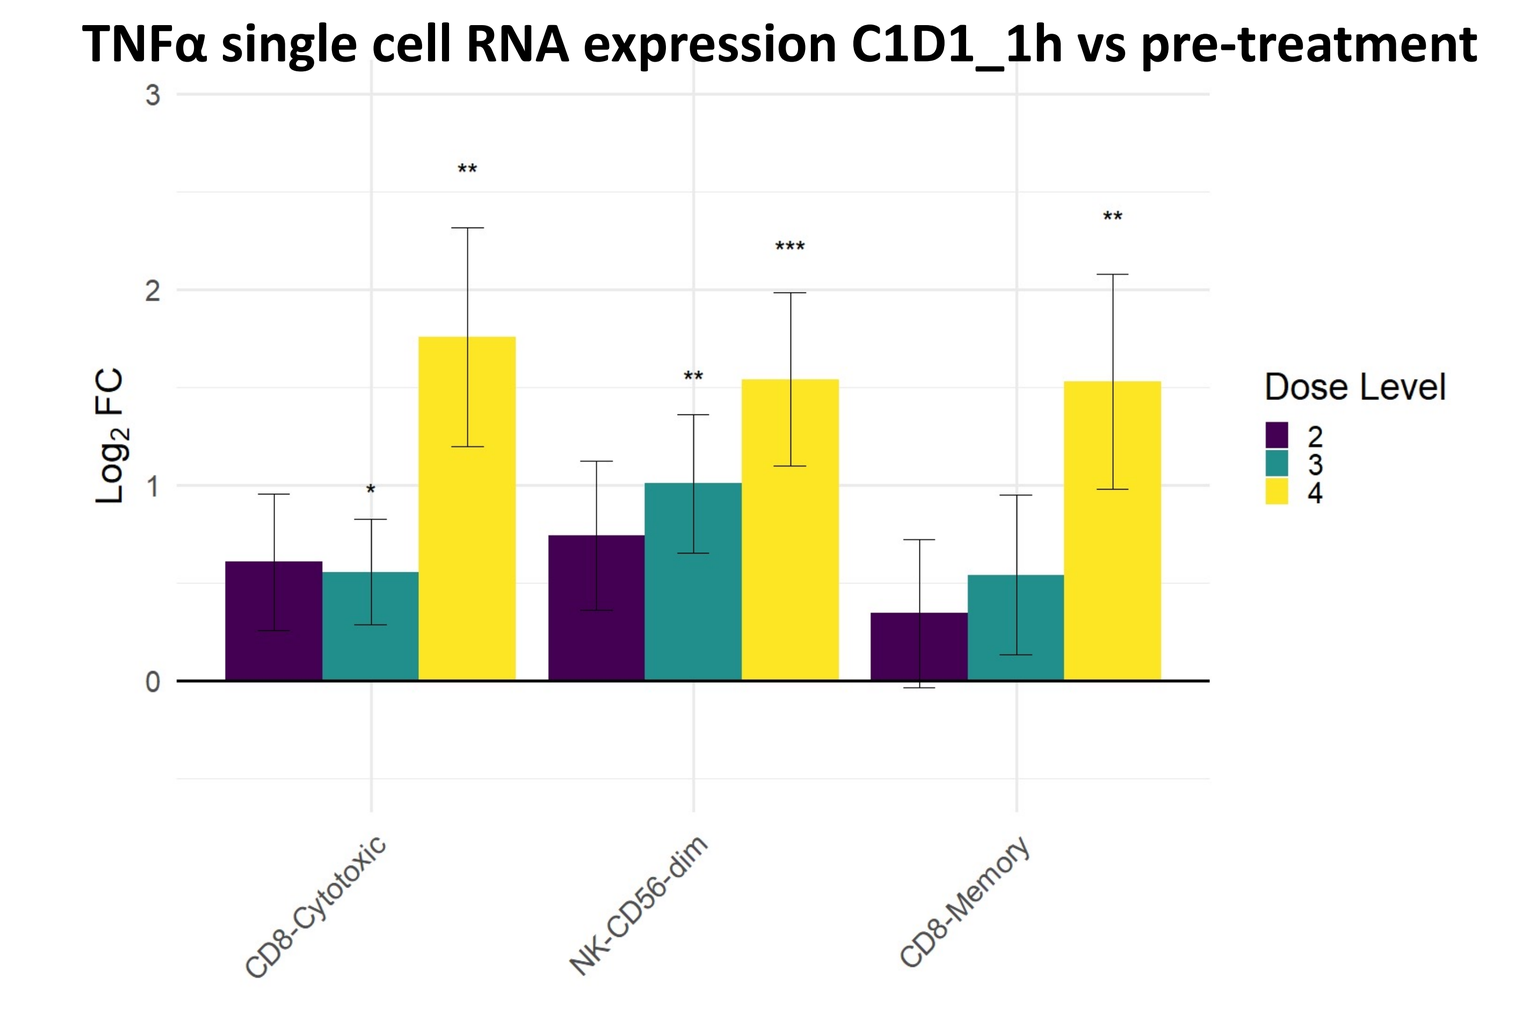
TNFα single cell RNA expression C1D1_1h vs pre-treatment
Log2 FC
Dose Level
2
3
4
CD8-Cytotoxic
NK-CD56-dim
CD8-Memory

~290k cells passing QC across time-points in 16 patients; scRNA-seq data integrated with HiFiBiO generative AI; Log2FC shows change between post- vs pre-treatment
*** p < 0.001; ** p < 0.01; * p < 0.05 via Bayesian multi-level linear modeling

FOLLOW-UP DESIGN and RESULTS

Q2W study design and enrollment status

Monotherapy HFB200301 Q2W (n=11)

DL 4 HFB (n=4)
DL 3 HFB (n=7)

Combination HFB200301 Q2W + tislelizumab Q4W (n=14)

DL 4 HFB + TIS (n=7)
DL 3 HFB + TIS (n=7)

DL (Dose Level); HFB (HFB200301); Q2W (once every 2 weeks); TIS (tislelizumab)

Demographics and clinical characteristics

Characteristic	Monotherapy (n=11)*	Combination (n=14)*
Median age, years (range)	56 (50-71)	62.5 (37-76)
Sex, n (%)		
Women	5 (45)	6 (43)
Men	6 (55)	8 (57)
ECOG PS, n (%)		
0	4 (40)	5 (36)
1	6 (60)	9 (64)
Median time since initial diagnosis (range), years	2.2 (0.4-22.0)	3.2 (0.7-15.8)
Number of prior systemic cancer therapy regimens, n (%)		
Median (range)	2 (1-3)	3 (1-4)
1	2 (18)	1 (8)
2	4 (36)	5 (38)
≥3	5 (46)	7 (54)
Received prior anti-PD-(L)1 therapy, n (%)		
Yes	8 (73)	12 (92)
No	3 (27)	1 (8)
Median follow-up time, months (range)	2.0 (0.2+ -5.2)	0.9 (0.2+ - 3.8+)
Tumor types, n (%)		
Cervical cancer	0 (0)	2 (14)
Gastric cancer, EBV+	0 (0)	0 (0)
Head and neck squamous cell carcinoma	3 (28)	1 (7)
Melanoma	1 (8)	0 (0)
Non-small cell lung cancer	1 (8)	4 (29)
Pleural mesothelioma	0 (0)	0 (0)
Renal cell carcinoma	3 (28)	7 (50)
Sarcoma	3 (28)	0 (0)
Testicular germ cell tumor	0 (0)	0 (0)

EBV+, Epstein-Barr virus positive; ECOG PS, Eastern Cooperative Oncology Group performance status; PD-(L)1, programmed cell death protein (ligand) 1.
* Data from all subjects may not yet be available, hence, the total reported numbers may be less.

Safety profiles of HFB200301 Q2W ± tislelizumab Q4W

- HFB200301 Q2W across all dose levels was well tolerated in monotherapy and combination with TIS Q4W without any DLTs, Grade 3 TRAEs, and TRAEs leading to discontinuation or dose modification.
- Similar incidence rate and severity of TRAEs commonly observed in Q4W regimen[1].

Adverse Events	HFB200301 Monotherapy Q2W (n=11)				HFB200301 Q2W + Tislelizumab Q4W (n=14)			
	All grades n (%)	Grade 1 n (%)	Grade 2 n (%)	Grade 3 n (%)	All grades n (%)	Grade 1 n (%)	Grade 2 n (%)	Grade 3 n (%)
Anemia	-	-	-	-	1 (7)	-	1 (7)	-
Asthenia	2 (18)	1 (9)	1 (9)	-	3 (21)	1 (6)	2 (14)	-
Back pain	1 (9)	1 (9)	-	-	-	-	-	-
Chills, shivers	-	-	-	-	3 (21)	3 (21)	-	-
Cytokine release syndrome	1 (9)	1 (9)	-	-	3 (21)	-	3 (21)	-
Dysthermia	-	-	-	-	1 (7)	1 (7)	-	-
Emesis	1 (9)	1 (9)	-	-	-	-	-	-
Fever	1 (9)	1 (9)	-	-	3 (21)	3 (21)	-	-
Hyperthyroidism	-	-	-	-	1 (7)	1 (7)	-	-
Hypoalbuminaemia	-	-	-	-	1 (7)	1 (7)	-	-
Infusion reaction	-	-	-	-	1 (7)	-	1 (7)	-
Mucositis oral	1 (9)	1 (9)			-	-	-	-
Myalgia	1 (9)	1 (9)	-	-	1 (7)	-	1 (7)	-
Myoclonus	-	-	-	-	1 (7)	-	1 (7)	-
Nausea	1 (9)	1 (9)	-	-	-	-	-	-
Pruritus	-	-	-	-	1 (7)	-	1 (7)	-
Rash	1 (9)	1 (9)	-	-	1 (7)	1 (7)	-	-
WBC decreased	1 (9)	1 (9)	-	-	-	-	-	-
Weight loss	-	-	-	-	1 (7)	1 (7)	-	-
Xerophthalmia	-	-	-	-	1 (7)	-	1 (7)	-

DLT, dose-limiting toxicity; TRAE, treatment-related adverse event

PRELIMINARY ANTI-TUMOR RESPONSE

Best overall response to HFB200301 Q2W ± tislelizumab Q4W

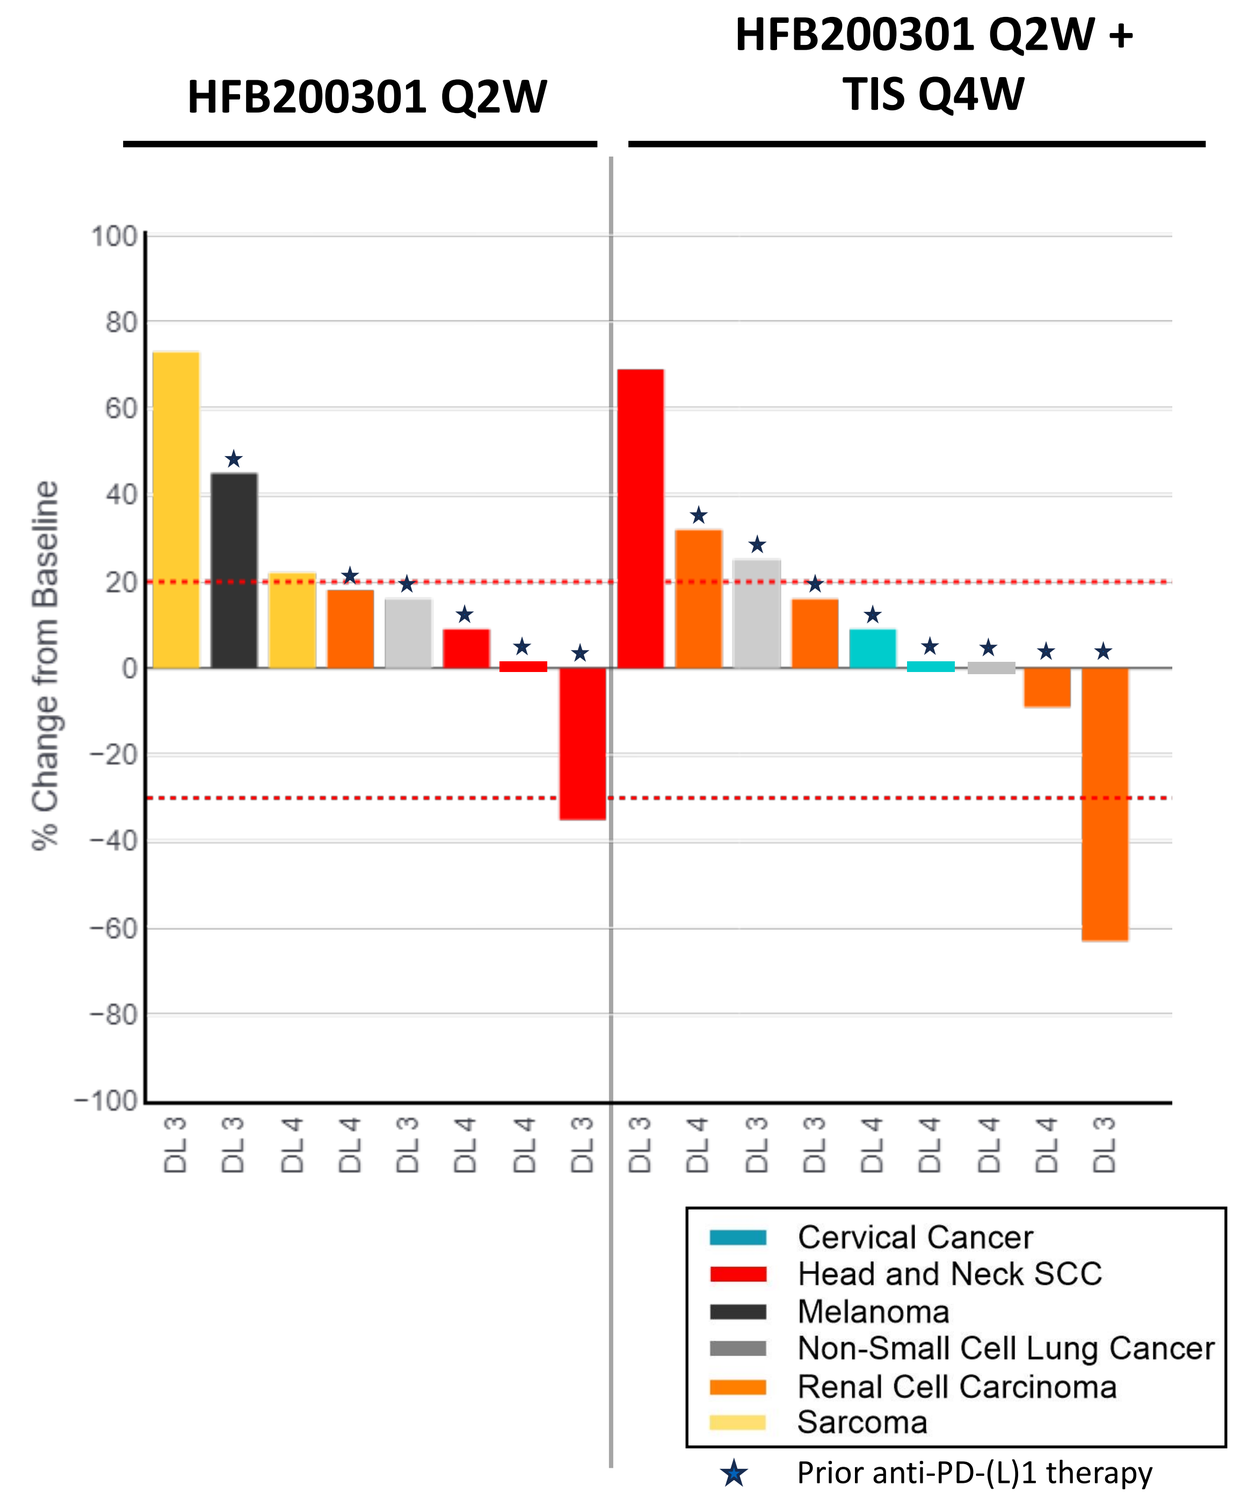
HFB200301 Q2W
HFB200301 Q2W + TIS Q4W
% Change from Baseline
DL 3
DL 4
Cervical Cancer
Head and Neck SCC
Melanoma
Non-Small Cell Lung Cancer
Renal Cell Carcinoma
Sarcoma
Prior anti-PD-(L)1 therapy

HFB200301 Mono Q2W (Evaluable patients, n= 9)
- **Partial response** in **heavily pretreated HNSCC** patient with 4 prior LoT including anti-PD-1 therapy
- In contrast, there were no responses observed in Q4W monotherapy[1]
- Response: 1 PR, 4 SD, 4 PD, DCR* 56%
- 2 Patients still awaiting first scan

HFB200301 Q2W + TIS Q4W (Evaluable patients, n=11)
- **Partial response** confirmed in **anti-PD-1 relapsed RCC** patient, with DCR* 75% in RCC
- Response: 1 PR, 5 SD, 5 PD, DCR* 55%
- 3 Patients still awaiting first scan

*DCR includes stable disease (SD), partial response (PR) and complete response (CR) in target lesion per RECIST 1.1; LoT, lines of treatment

Radiographic responses to HFB200301 Q2W ± tislelizumab Q4W

Head and Neck SCC
Patient & Treatment Information
- 64-year-old female
- Enrolled in HFB200301 DL 3 Q2W monotherapy cohort
- Stage IV, CPS 2
- 4 prior lines of therapy, including chemotherapeutic, targeted, and anti-PD-1 agents.
- Time on treatment HFB200301 trial 5.3 mo

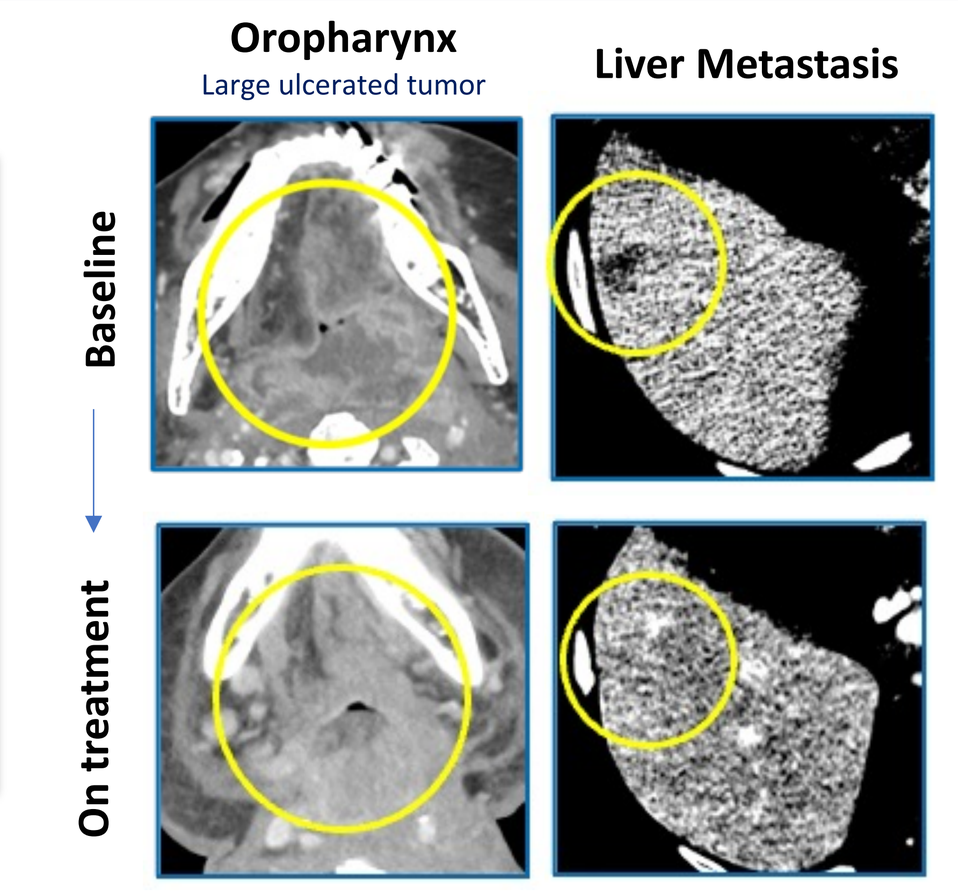
Oropharynx
Large ulcerated tumor
Liver Metastasis
Baseline
On treatment

Renal Cell Carcinoma
Patient & Treatment Information
- 49-year-old male
- Enrolled in HFB200301 DL 3 Q2W in combination with tislelizumab Q4W cohort
- Stage IV
- 2 prior lines of therapy, including anti-PD-1 and targeted agents
- Time on treatment HFB200301 trial > 4 mo (ongoing)

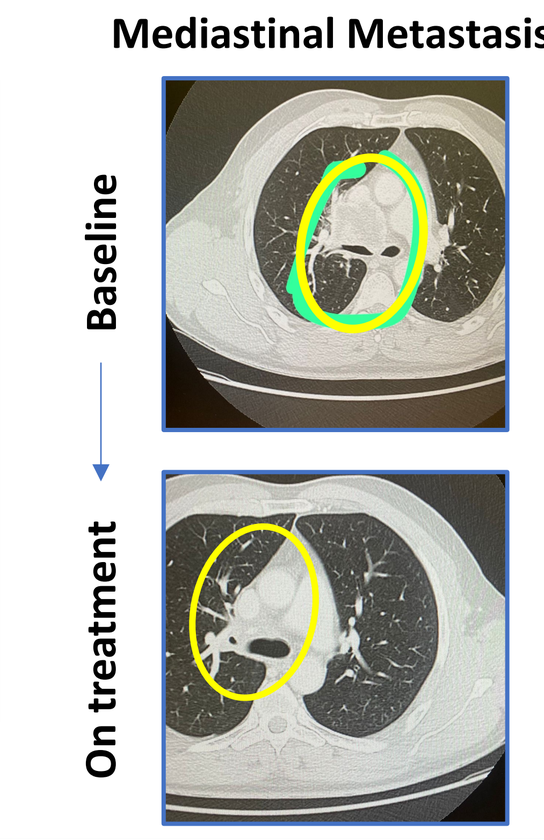
Mediastinal Metastasis
Baseline
On treatment

SUMMARY and FUTURE DIRECTIONS

- Data from HFB200301 Q4W dosing enabled the successful development of PK and target engagement dosing models.
- These models guided the optimization of dosing, leading to the clinical evaluation of HFB200301 at Q2W.
- HFB200301 Q2W dosing showed a favorable safety profile similar to that of Q4W dosing.
- Additionally, Q2W dosing demonstrated preliminary efficacy, both as monotherapy and in combination with TIS, in heavily pre-treated tumors.
 - Preliminary efficacy with Q2W monotherapy hints potentially improved efficacy compared to Q4W monotherapy.
- The potentially improved efficacy with Q2W dosing regimen will be further evaluated in the Dose Expansion part of the clinical trial.
- This trial underscores the effectiveness of model-informed development of agonist drugs.

Acknowledgments and references

We extend our thanks to the patients, their families, and the investigators and staff members who made this trial possible.

1. Roda, D. et al., (2024). *Phase 1 Dose Escalation Trial of the First-in-class TNFR2 Agonist Monoclonal Antibody HFB200301 in Monotherapy and in Combination with tislelizumab, an anti-PD-1 Monoclonal Antibody, in Adult Patients with Advanced Solid Tumors*. ASCO 2024. Chicago, IL.

Study sponsored by HiFiBiO Inc.

For additional information, please email contact@hifibio.com or visit HiFiBiO.com