Poster #783TiP
ESMO Congress 2022
Paris, France
September 9-13, 2022

Phase I study of HFB301001, a novel OX40 agonist monoclonal antibody, in patients with solid tumors selected via Drug Intelligence Science (DIS™)

Alexander I. Spira[1], Ranee Mehra[2], Charlene Mantia[3], Hani Babiker[4], Mitesh Borad[5], Andrés Cervantes[6], Elena Garralda Cabanas[7], Amit Mahipal[8], Luis Paz-Ares[9], Christos Hatzis[10], Angela Liu[10], Andreas Raue[10], Jinping Gan[10], Francisco Adrian[10], Luigi Manenti[10], Anthony El-Khoueiry[11]

[1]Virginia Cancer Specialists, Fairfax, VA; [2]University of Maryland, Greenbaum Comprehensive Cancer Center, Baltimore, MD; [3]Dana Farber Cancer Institute, Boston, MA; [4]Mayo Clinic, Jacksonville, FL; [5]Mayo Clinic, Scottsdale, AZ; [6]University of Valencia, Instituto de Salud Carlos III, CIBERONC, Valencia, Spain; [7]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [8]Mayo Clinic, Rochester MN; [9]Hospital Universitario 12 de Octubre, Madrid, Spain; [10]HiFiBiO Inc., Cambridge, MA; [11]University of Southern California, Norris Comprehensive Cancer Center, Los Angeles, CA

BACKGROUND

- First generation of OX40 agonist antibodies have shown promising preclinical activity but limited clinical success thus far[1-4]. To increase the probability of success for a next generation OX40 agonist, HFB301001, we optimized the i) pharmacological profile, ii) dosing regimen, and iii) biomarker strategy for patient selection.

i) Pharmacological Profile

- HFB301001, a novel fully human IgG1 OX40 agonist antibody
- Binds to a unique epitope on OX40, allowing for agonistic activity without competing with endogenous OX40 ligand binding, and does not result in decreased OX40 surface levels upon co-stimulation of T cells[5].
- Enhances effector T cell activity and depletes regulatory T cells[6].
- Demonstrates more potent *in vivo* anti-tumor activity than a benchmark OX40 agonist, suggesting potentially superior T cell stimulation[5].

➡ **Optimized OX40 agonist characteristics, translated in superior activity preclinically**

ii) Dosing Regimen

- A mechanistic modeling approach was used to integrate available PK, PD, and efficacy data from an MC38 hOX40 knock-in mouse model.
- The PK/PD model was employed together with PK data in cynomolgus monkeys to predict the optimal human dosing regimen[6].

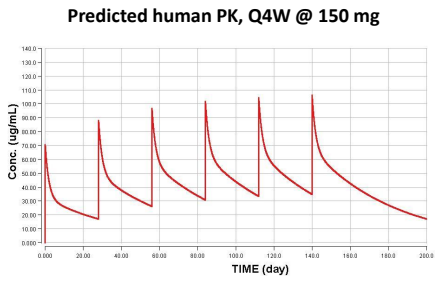
Predicted human PK, Q4W @ 150 mg
Conc. (ug/mL)
TIME (day)

➡ **Human dose & schedule derived from all available preclinical PK, PD, and efficacy data to optimize maximize immune stimulation**

iii) Biomarker Strategy for Patient Selection

- Leveraging unique, in-house single-cell immuno-profiling platform Drug Intelligence Science (DIS™) to identify most promising indications

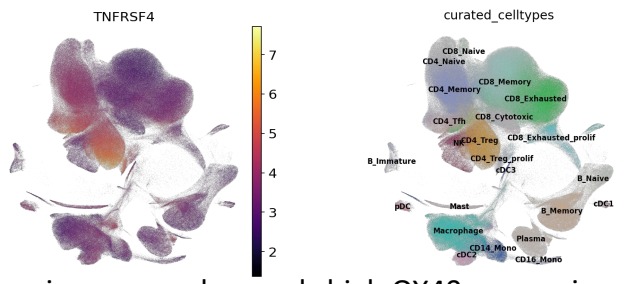
TNFRSF4
curated_celltypes
CD8_Naive
CD4_Naive
CD8_Memory
CD4_Memory
CD8_Exhausted
CD4_Tfh
CD8_Cytotoxic
CD4_Treg
CD8_Exhausted_prolif
CD4_Treg_prolif
cDC3
B_Immature
B_Naive
pDC
Mast
B_Memory
cDC1
Macrophage
Plasma
CD14_Mono
cDC2
CD16_Mono

- Immune cell profiling through deep learning approach reveals high OX40-expression in effector T cells (T_{eff}) and regulatory T cells (T_{reg})
- The following cancer indications have been identified based on the prevalence of high OX40 signature in T_{eff} and T_{reg} cells from The Cancer Genome Atlas (TCGA)

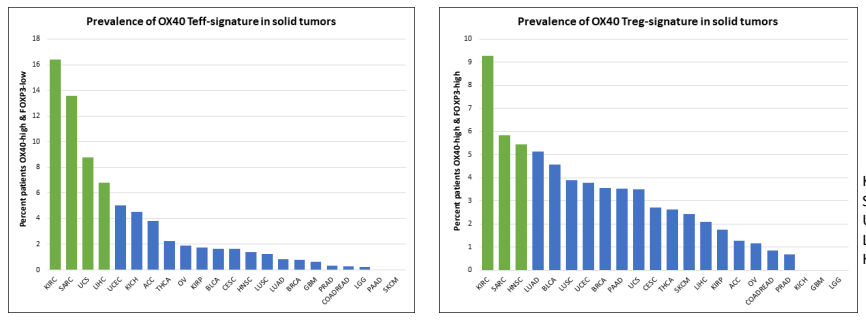
Prevalence of OX40 Teff-signature in solid tumors
Percent patients OX40-high & FOXP3-low
KIRC SARC UCS LIHC UCEC KICH ACC THCA OV KIRP BLCA CESC HNSC LUSC LUAD BRCA GBM PRAD COADREAD LGG PAAD SKCM
Prevalence of OX40 Treg-signature in solid tumors
Percent patients OX40-high & FOXP3-high
KIRC SARC HNSC LUAD BLCA LUSC UCEC BRCA PAAD UCS CESC THCA SKCM LIHC KIRP ACC OV COADREAD PRAD KICH GBM LGG

KIRC: clear cell renal cell carcinoma
SARC: sarcoma
USC: uterine carcinoma
LIHC: hepatocellular carcinoma
HNSC: head and neck squamous cell carcinoma

- The integrated single-cell atlas will be complemented by scRNA-seq from treated patients in our trial to guide precision biomarkers

➡ **The DIS™ guided OX40 indications may enable enrichment of OX40 responding patients, helping to identify predictive biomarkers**

STUDY DESIGN

First-in-human, multicenter, open-label, dose escalation and dose expansion study

Dose Escalation

- Using modified Toxicity Probability Interval 2 (mTPI-2) design[7] with target dose-limiting toxicity (DLT) rate of approximately 30%
- Dosing initiated at 5 mg with provisional dose levels of 15, 50, and 150 mg
- At least 3 patients (up to 6) must be enrolled in a cohort for dose escalation to occur. Back-filling cohorts is permitted
- HFB301001 is administered as a 30-minute IV infusion every 4 weeks
- DLT period is the first 28 days after cycle 1 day 1 treatment

Dose Expansion

- Expansions of up to 5 cohorts with up to 20 patients each.
 - Soft tissue sarcoma (STS), renal cell carcinoma (RCC), hepatocellular carcinoma (HCC), head and neck squamous cell carcinoma (HNSCC), and potentially uterine carcinosarcoma (UCS)

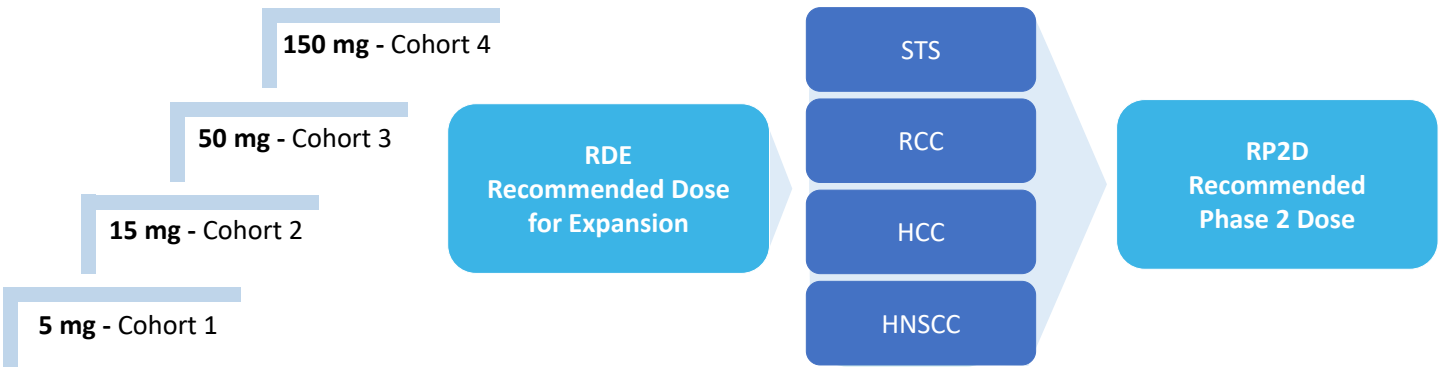
150 mg - Cohort 4
50 mg - Cohort 3
15 mg - Cohort 2
5 mg - Cohort 1
RDE
Recommended Dose for Expansion
STS
RCC
HCC
HNSCC
RP2D
Recommended Phase 2 Dose

KEY ELIGIBILITY CRITERIA

- Male/female adult patients with histologically documented and advanced or metastatic solid tumors. Permitted tumor types:
 - **Soft tissue sarcoma**
 - **Renal cell carcinoma**
 - **Hepatocellular carcinoma**
 - **Head and neck squamous cell carcinoma**
 - **Uterine carcinosarcoma**
- Patient must have exhausted standard lines of systemic therapy*
- Patient must be willing to undergo pre-treatment and on-treatment biopsies
- Patient must have measurable disease based on RECIST 1.1
- Patient cannot have hemoglobin <9.0 g/dL or equivalent

*Other protocol defined inclusion criteria may apply

KEY OBJECTIVES

Primary

- Dose escalation:
 - Characterize safety and tolerability of single agent HFB301001
 - Determine RDE(s)
- Dose expansion:
 - Determine RP2D

Secondary

- Assess the PK profile and pharmacodynamic effects of HFB301001 in the blood and tumor
- Evaluate immunogenicity
- Determine biologically active dose(s) and anti-tumor efficacy of HFB301001

Exploratory

- Assess relationship between PK, baseline and on treatment biomarkers, and/or anti-tumor efficacy
- Characterize immune modulation in the tumor microenvironment

ENROLLMENT

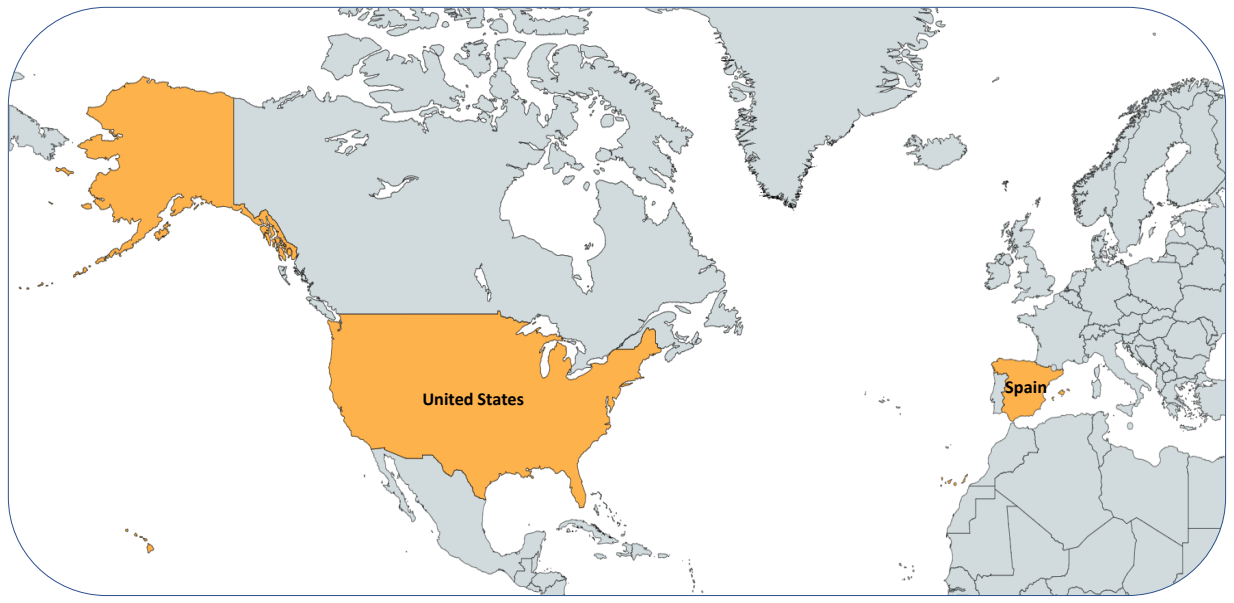
United States
Spain

Enrollment opened in February 2022
United States: 7 sites
Spain: 3 sites

Virginia Cancer Specialists	Mayo Clinic Scottsdale
University of Maryland Cancer Center	Mayo Clinic Rochester
Dana Farber Cancer Institute	Vall d'Hebron Institute of Oncology
USC Norris Comprehensive Cancer Center, LA	Universitario de Valencia
Mayo Clinic Jacksonville	Hospital Universitario 12 de Octubre

REFERENCES

1. Glisson et al. Clin Cancer Res. 2020;26(20):5358-5367
2. Kim et al. Clin Cancer Res. 2022; clincanres.4020.2021
3. Postel-Vinay et al. Cancer Res 2020; 80 (16_Supplement):CT150
4. Diab et al. Clin Cancer Res. 2022; 28(1): 71-83
5. AACR 2020, E-Poster #2285
6. AACR 2021, E-Poster #1882
7. Guo et al. *Contemp Clin Trials*. 2017;58:23-33

ACKNOWLEDGEMENTS

We extend our thanks to the patients, their families, and the investigators and their site staff members who are making this trial possible. Thank you to Olja Rapaic, John Pallante, and Medpace, Inc. for their efforts and collaboration in this trial.

The presenter, Dr. Anthony El-Khoueiry, declares no conflict of interest in the context of this clinical trial.

This study is supported by HiFiBiO Inc.

CONTACT INFORMATION

This poster was presented at 2022 ESMO Congress (Sept 9-13, 2022); Paris, France.

Corresponding Author: elkhouei@med.usc.edu

Clinicaltrials.gov Identifier: NCT05229601

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

SCAN ME