Poster 7176
Presented at the AACR
April 10, 2024

Optimization of T cell co-stimulatory agonists: A semi-mechanistic PKPD model integrating drug properties and tumor-immune interactions

Jinping Gan, William Hedrich, Yun-Yueh Lu, Robert Andtbacka, Francisco Adrian, Liang Schweizer

BACKGROUND

Unlike checkpoint blockade, achieving clinical success with T cell co-stimulation has been challenging. This is primarily due to the complexities surrounding the optimal engagement of agonistic antibodies to co-stimulatory receptors and the lack of biomarkers for patient selection. Understanding various factors such as agonist-binding characteristics, pharmacokinetics (PK), activation and differentiation kinetics of T cell subpopulations, T cell lifespan in the tumor microenvironment, and the impact of T cell/tumor interactions on tumor growth kinetics is crucial for achieving positive clinical outcomes with T cell agonists. Presently, there is no suitable model to fully describe these intricate interactions.

Our novel TNFR2 agonist, HFB3-1, has demonstrated anti-tumor efficacy both alone and in combination with anti-PD-1 in syngeneic mouse models. Preclinical pharmacodynamics (PD) studies have revealed an increase in CD4 and CD8 T cells, as well as NK cells, in the tumor microenvironment upon treatment with HFB3-1, without affecting Tregs. Similarly, our novel OX40 agonist, HFB10-1, has shown anti-tumor efficacy in syngeneic mouse models. Notably, treatment with HFB10-1 results in the depletion of Tregs cells in the tumor microenvironment, in addition to increasing CD4 and CD8 T cells.

We are currently advancing the humanized versions of these agents, HFB200301 and HFB301001, respectively, through clinical development for the treatment of cancer.

MATERIALS AND METHODS

We investigated the PK, PD, and anti-tumor efficacy of the anti-TNFR2 agonist HFB3-1, and the anti-OX40 agonist HFB10-1 in syngeneic tumor models. Flow cytometry was employed to profile post-treatment tumor infiltrating lymphocytes (TILs) in dissociated tumor tissues. In vitro binding affinity was determined using Biolayer Interferometry. A semi-mechanistic model, integrating PK, tumor growth, and immune interaction networks (interactions among Teff, Treg, and tumor cells), was developed in Berkeley Madonna (version 10.5.1).

Semi-mechanistic TMDD Model

The two compartmental PK model parameters, including V1, V2, k12, and K21, were derived from PK profiles from wild-type mice.

Upon binding to membrane receptor, the antibody can undergo shedding and internalization, leading to target mediated drug disposition (TMDD), resulting in increased clearance.

Adapted from Cheng et al., Antibodies (Basel), 2020, 9, 49.

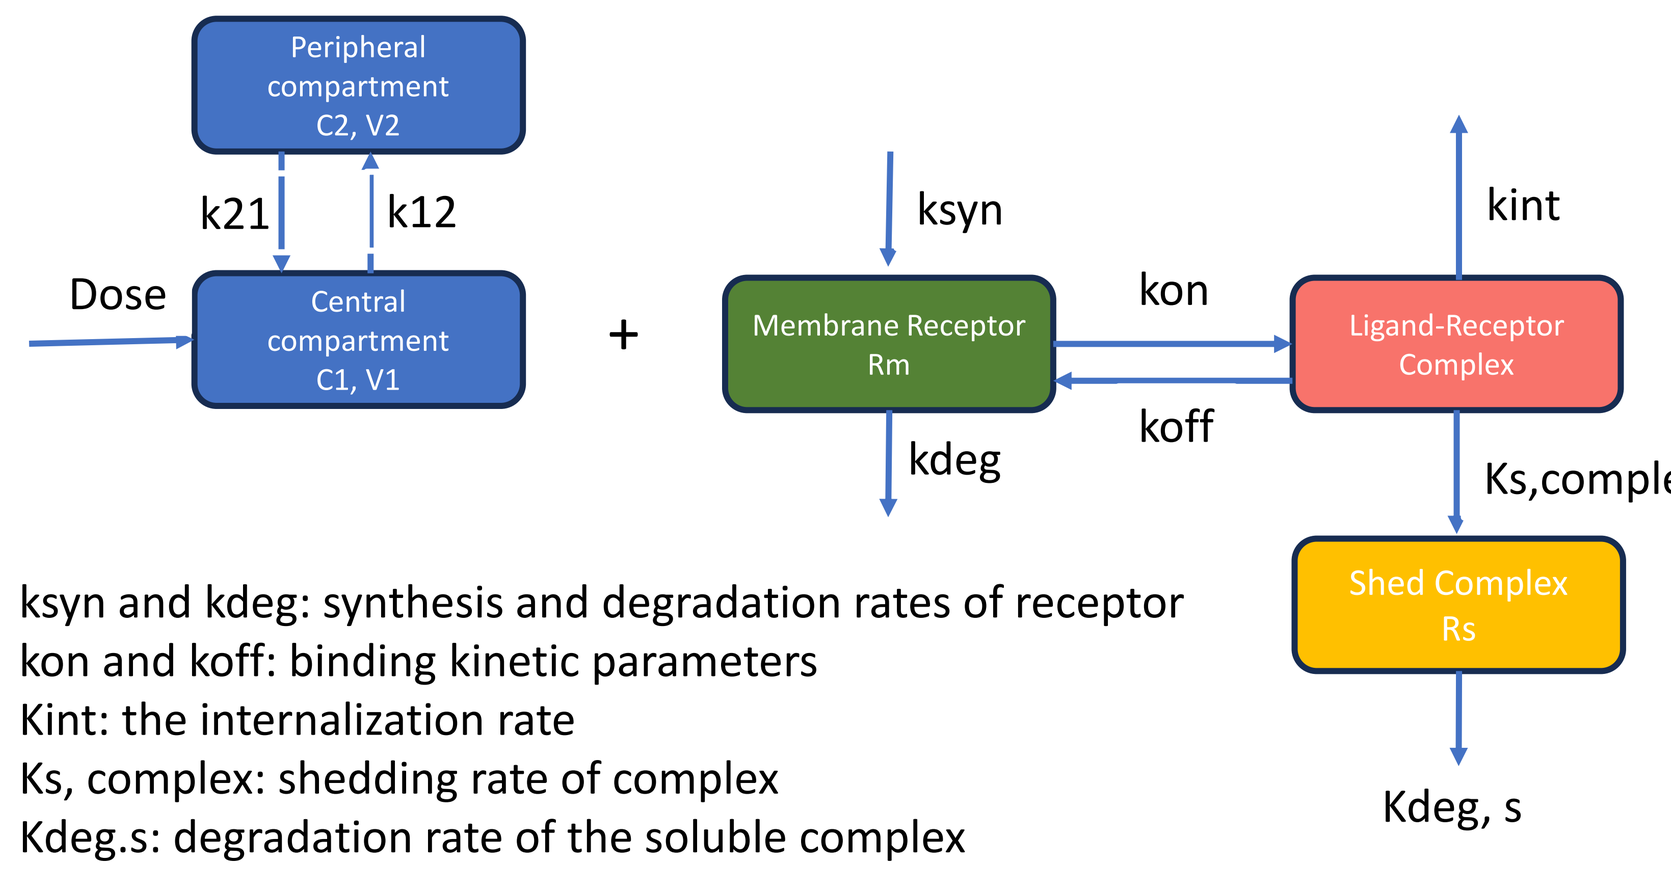
Peripheral compartment C2, V2
k21
k12
Dose
Central compartment C1, V1
+
ksyn
Membrane Receptor Rm
kdeg
kon
koff
Ligand-Receptor Complex
kint
Ks,complex
Shed Complex Rs
Kdeg, s

ksyn and kdeg: synthesis and degradation rates of receptor
kon and koff: binding kinetic parameters
Kint: the internalization rate
Ks, complex: shedding rate of complex
Kdeg.s: degradation rate of the soluble complex

Tumor Growth Model with Dynamic Immune Interaction

The tumor growth parameters are estimated from fitting tumor growth curves in vehicle treated animals along with measured CD8+ T cells and Treg counts on Day 4 after dosing.

Drug effect function follows an Emax model for Teff cell proliferation (HFB3-1), and for both Treg depletion and Teff proliferation (HFB10-1).

Adapted from Sontag, 2017, Cell Systems 4, 231.

$$\frac{dTV}{dt} = kg * TV * \left(1 - \frac{TV}{TV + TG50}\right) - p * Teff * TV$$

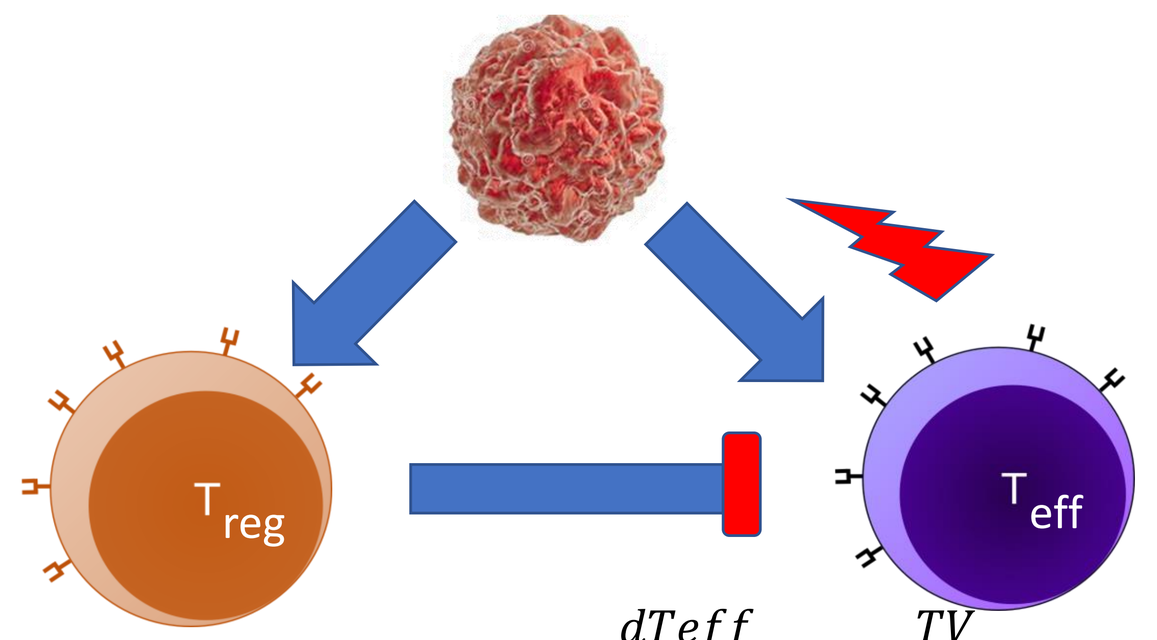
T reg
T eff

$$\frac{dTreg}{dt} = b * TV - kdeg,reg * Treg \qquad \frac{dTeff}{dt} = a * \frac{TV}{Treg} - kapop * Teff - e * Teff^2$$

kg: tumor growth rate; TV: tumor volume; p: tumor killing rate of Teff cells
b: tumor dependent Treg proliferation rate; kdeg,reg: Treg degradation rate
a: tumor and Treg dependent Teff proliferation rate; kapop: Teff apoptosis rate
e: Teff fratricidal rate

The PKPD model was subsequently adapted to a human tumor growth model with T cell interaction networks to assess the impact of doses and dosing regimens on tumor growth inhibition.

RESULTS

PKPD Modeling of HFB3-1 Tumor Growth Inhibition

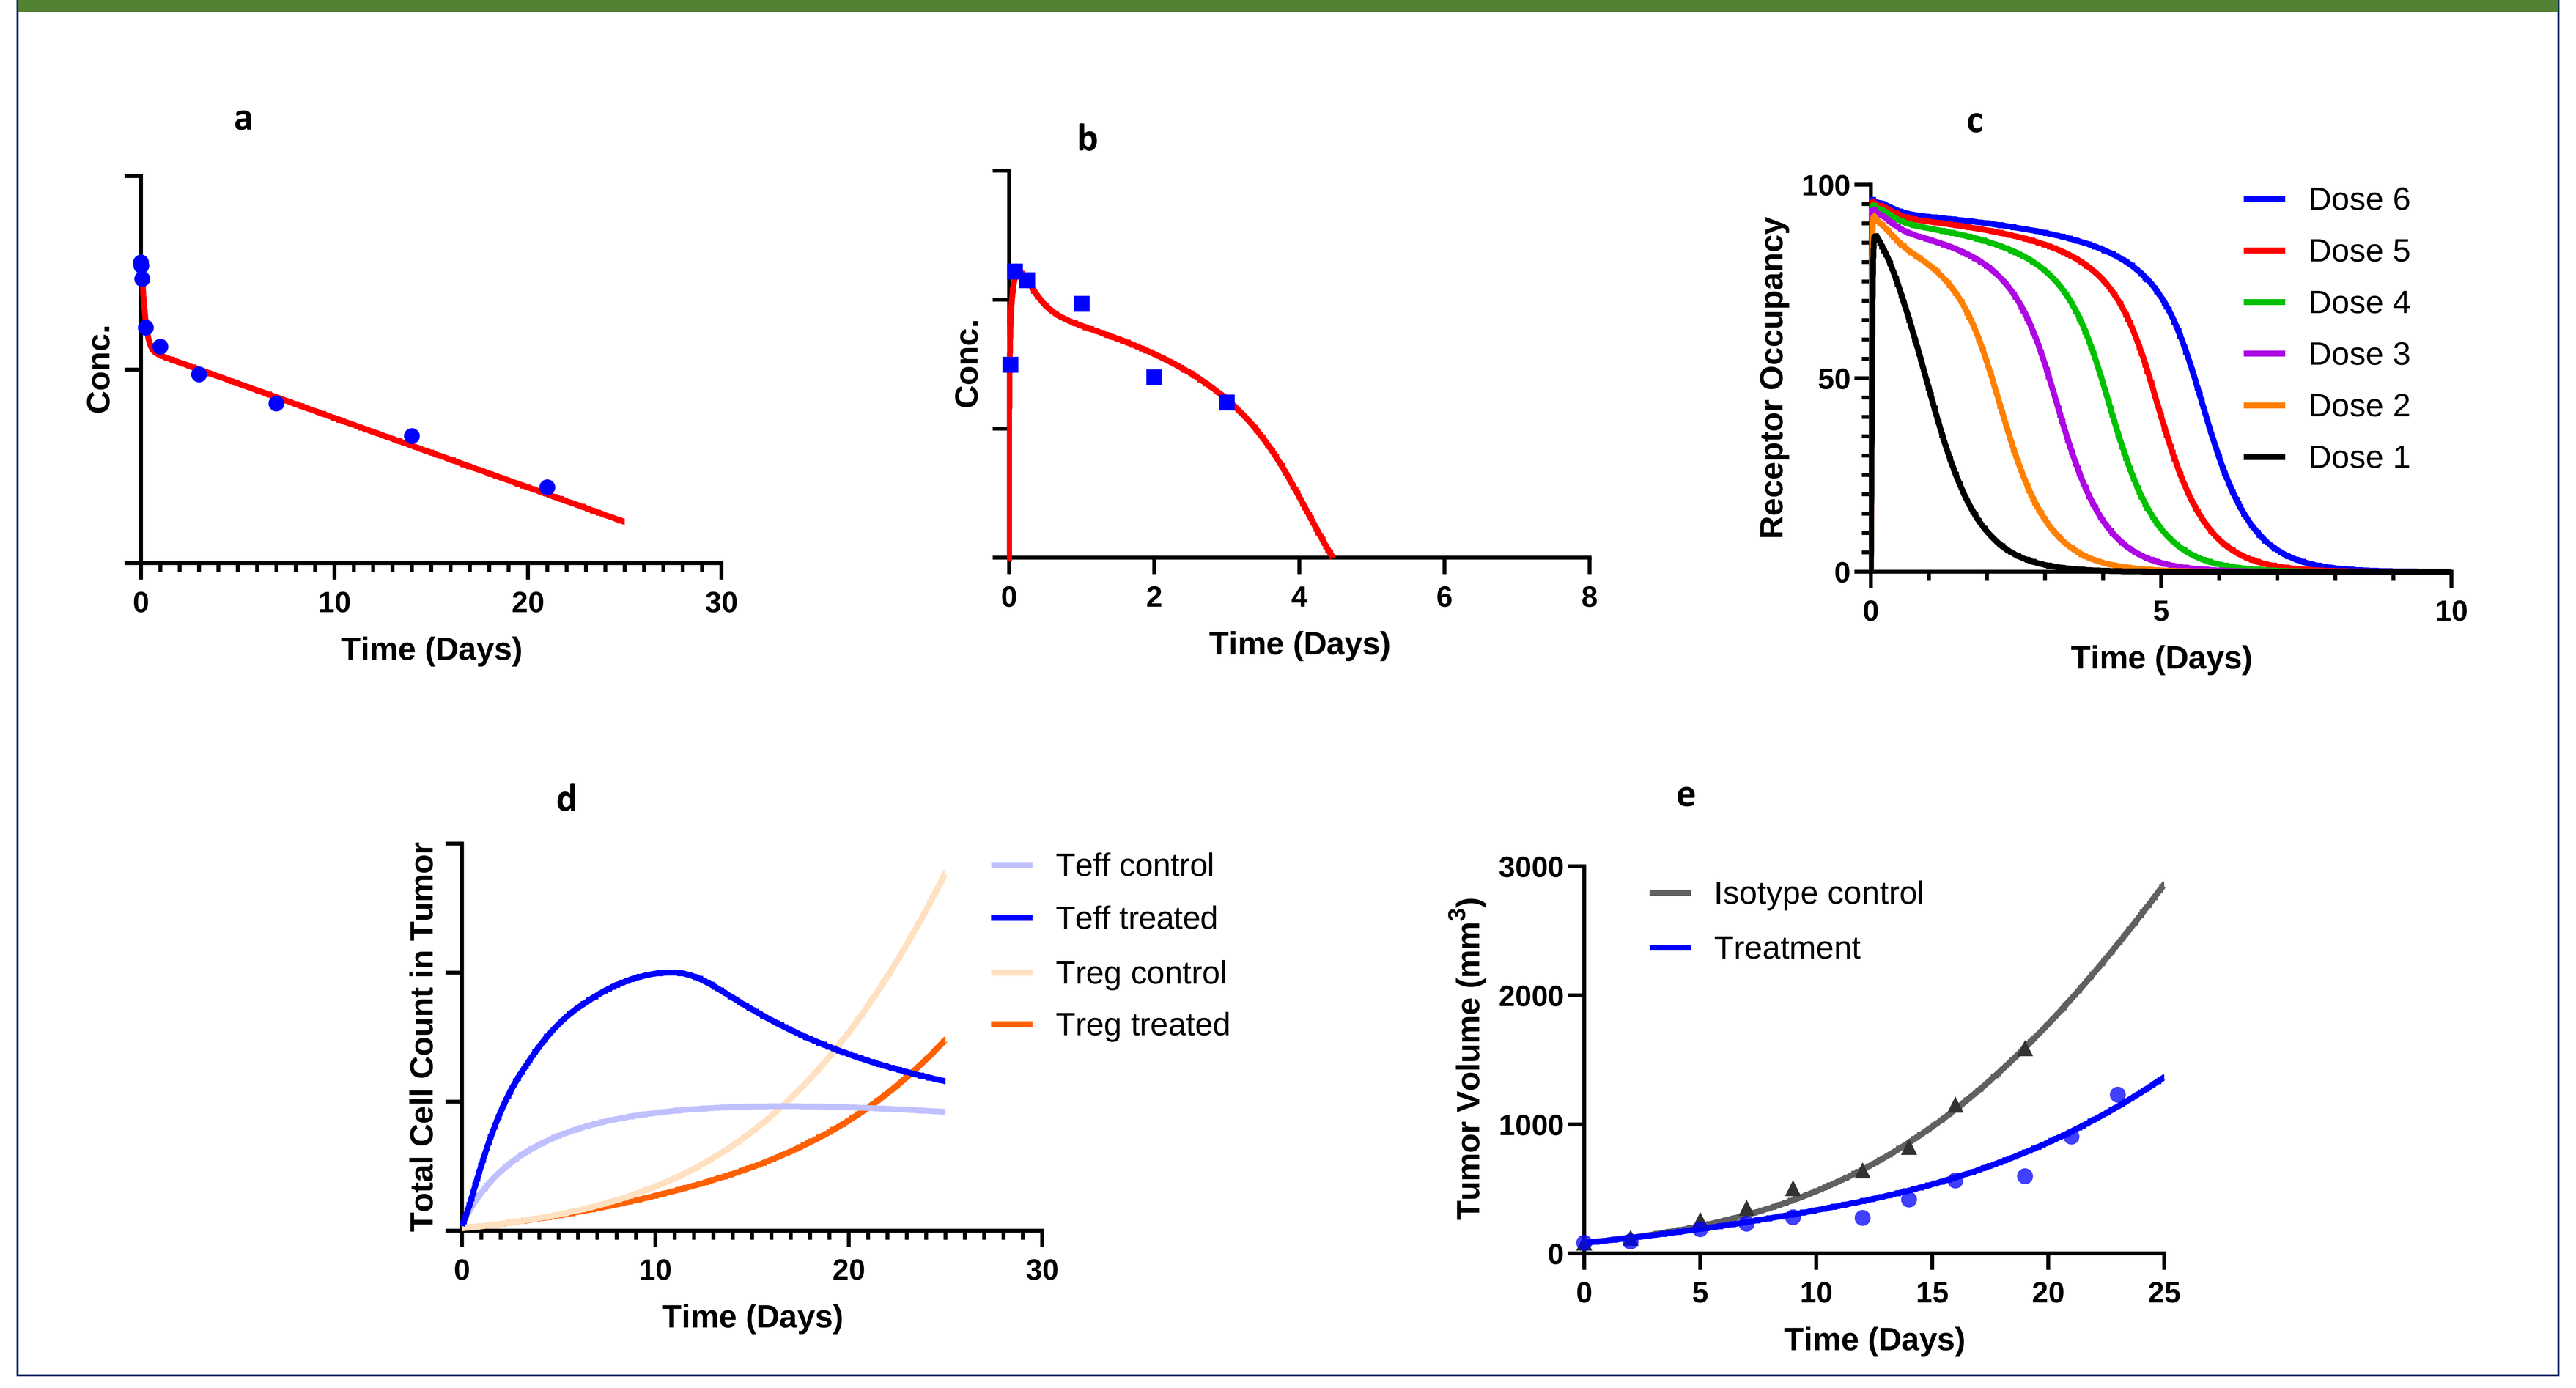
a
Conc.
Time (Days)
b
Conc.
Time (Days)
c
Receptor Occupancy
Time (Days)
Dose 6
Dose 5
Dose 4
Dose 3
Dose 2
Dose 1
d
Total Cell Count in Tumor
Time (Days)
Teff control
Teff treated
Treg control
Treg treated
e
Tumor Volume (mm³)
Time (Days)
Isotype control
Treatment

Figure 1. a. Fitting of HFB3-1 PK in wild type mice; b. Fitting of HFB3-1 PK at the same dose in TNFR2 knock-in (KI) mice; c. Simulated receptor occupancy at different doses in TNFR2 KI mice; d. Simulated profiles of Teff and Treg cells in tumor following the treatment with HFB3-1; e. Fitting of tumor growth profiles in MC38 tumor-bearing TNFR2 KI mice treatment with isotype control and HFB3-1.

Note in panel c, near 100% peripheral RO was achieved even at the lowest dose. RO coverage increased with increase of doses.

PKPD Modeling of HFB10-1 Tumor Growth Inhibition

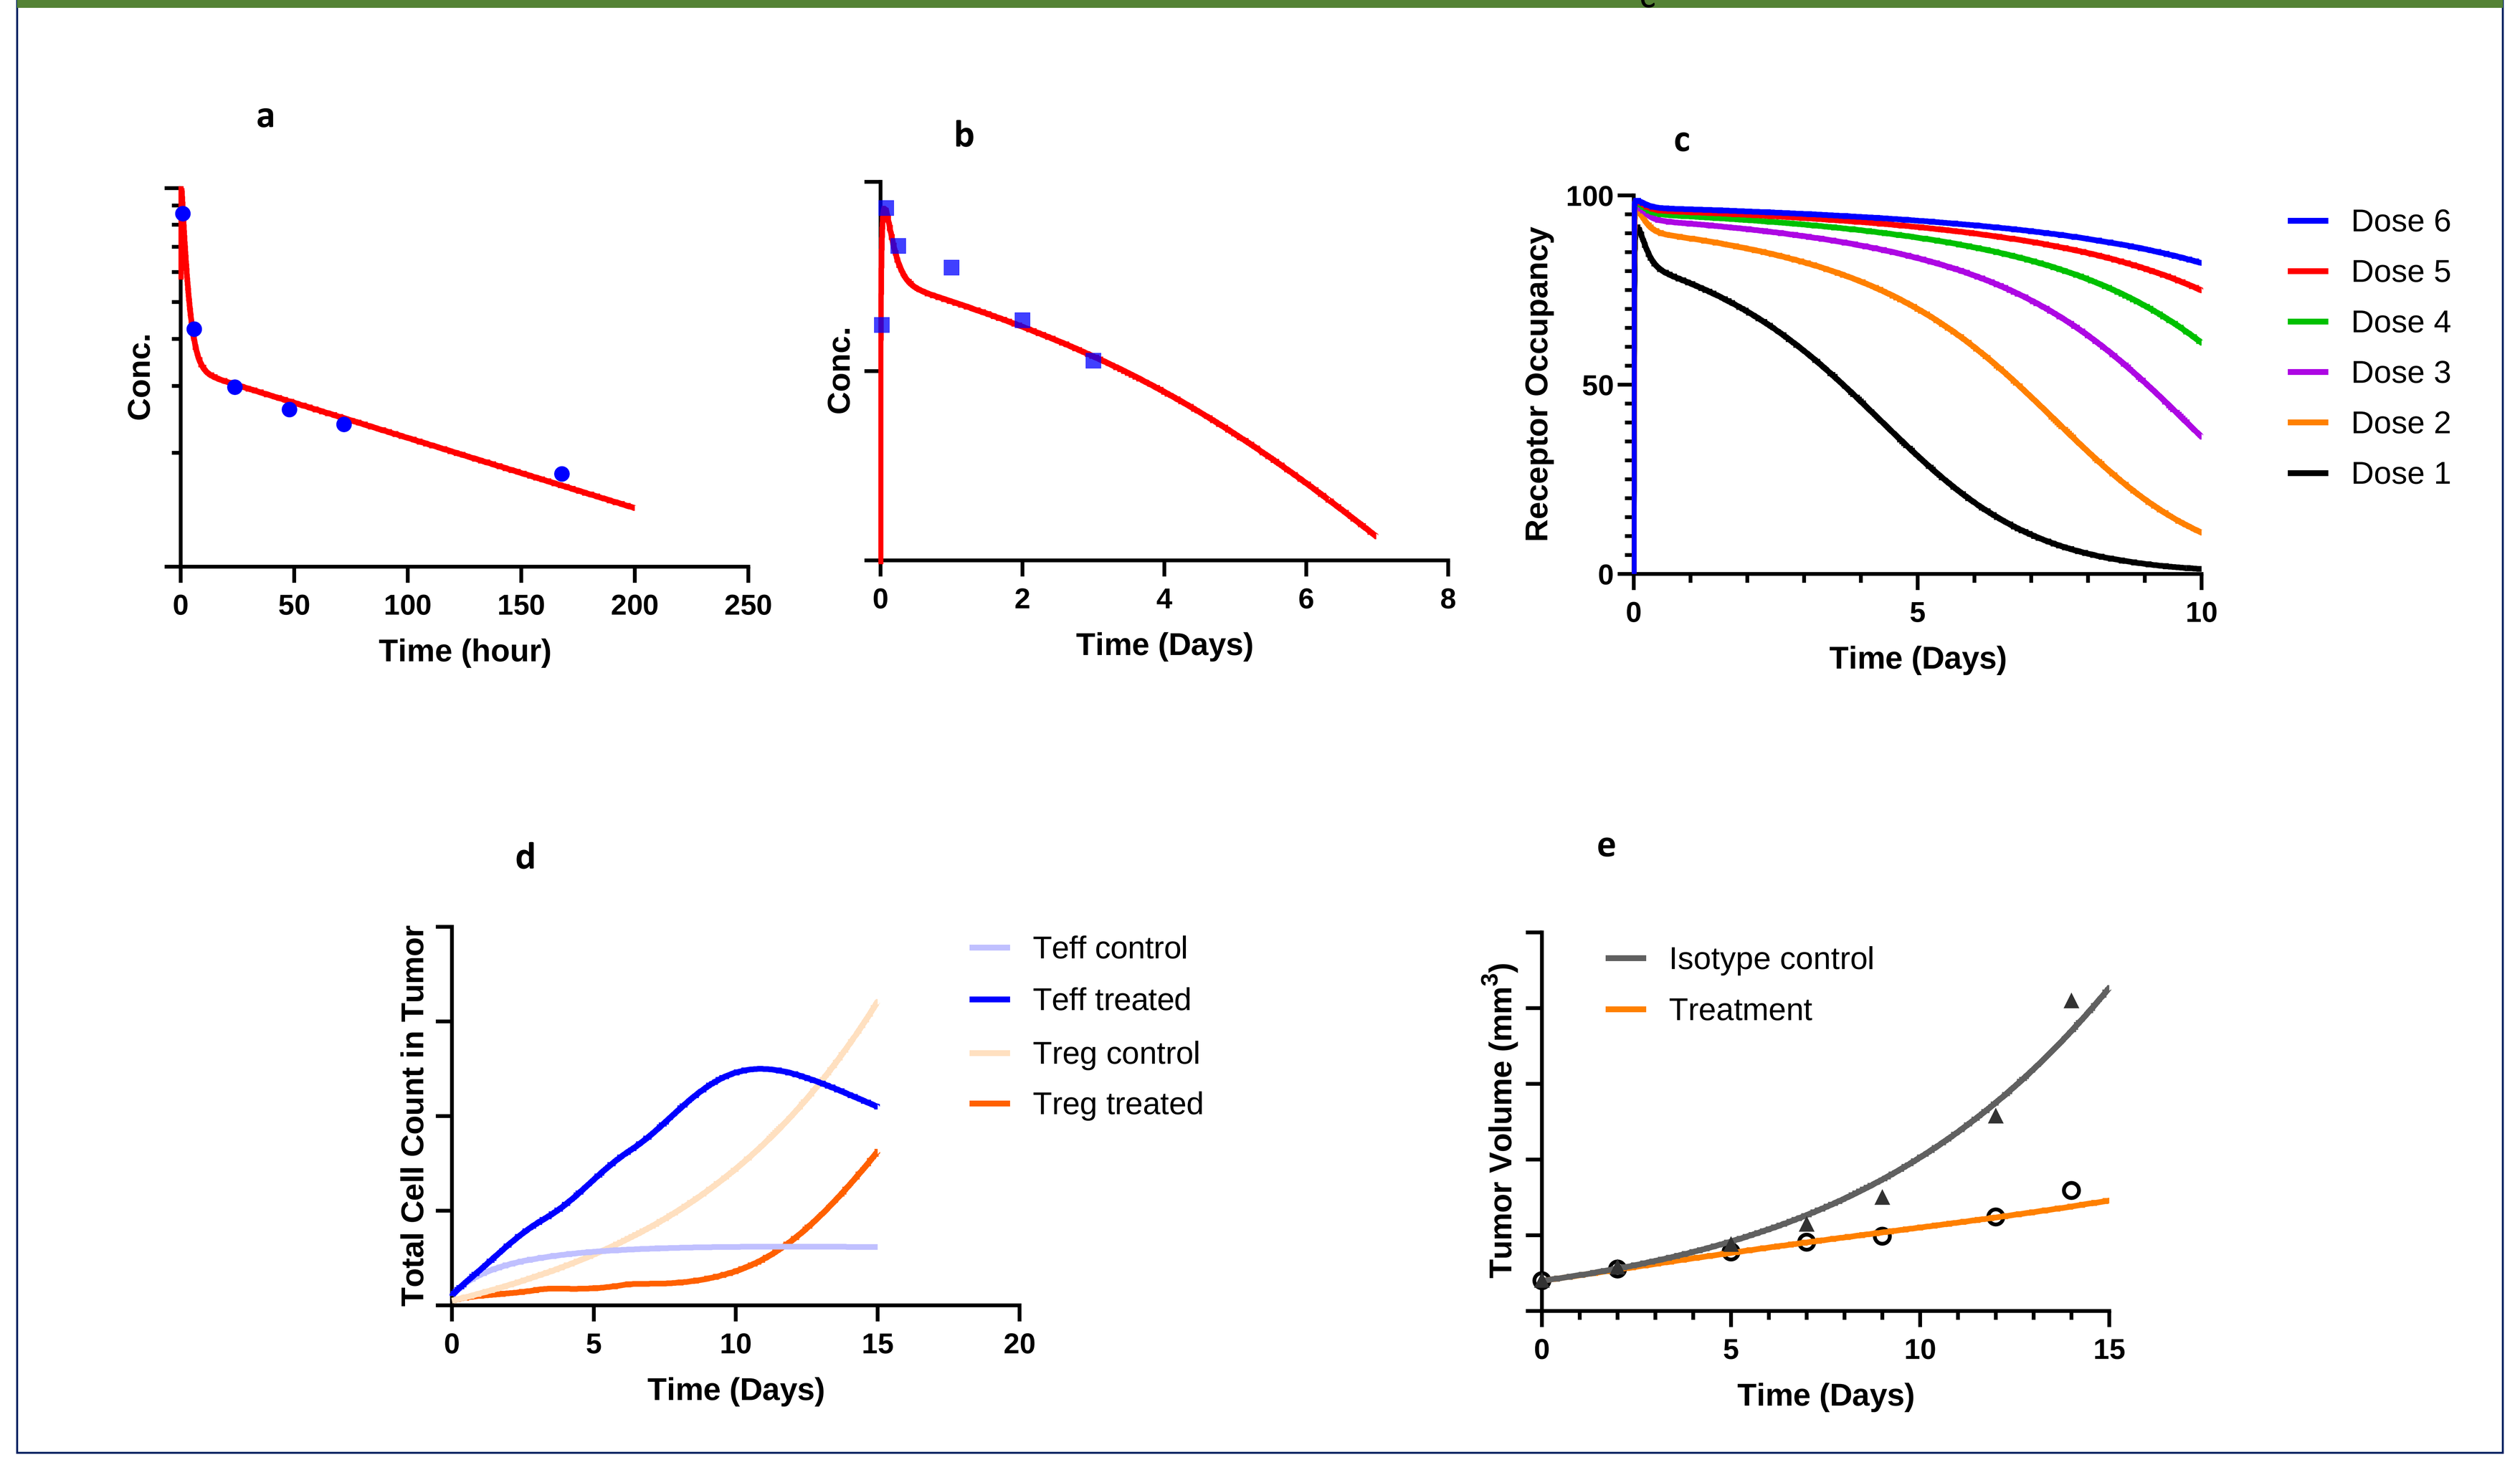
a
Conc.
Time (hour)
b
Conc.
Time (Days)
c
Receptor Occupancy
Time (Days)
Dose 6
Dose 5
Dose 4
Dose 3
Dose 2
Dose 1
d
Total Cell Count in Tumor
Time (Days)
Teff control
Teff treated
Treg control
Treg treated
e
Tumor Volume (mm³)
Time (Days)
Isotype control
Treatment

Figure 2. a. Fitting of HFB10-1 PK in wild type mice; b. Fitting of HFB10-1 PK at the same dose in OX40 knock-in (KI) mice; c. Simulated receptor occupancy at different doses in OX40 KI mice; d. Simulated profiles of Teff and Treg cells in tumor following the treatment of HFB10-1; e. Fitting of tumor growth profiles in MC38 tumor-bearing OX40 KI mice treatment with isotype control and HFB10-1.
In this model, Teff proliferation and Treg depletion are both taken into consideration.

RESULTS (cont.)

Impact of Tumor Microenvironment on anti-Tumor Efficacy

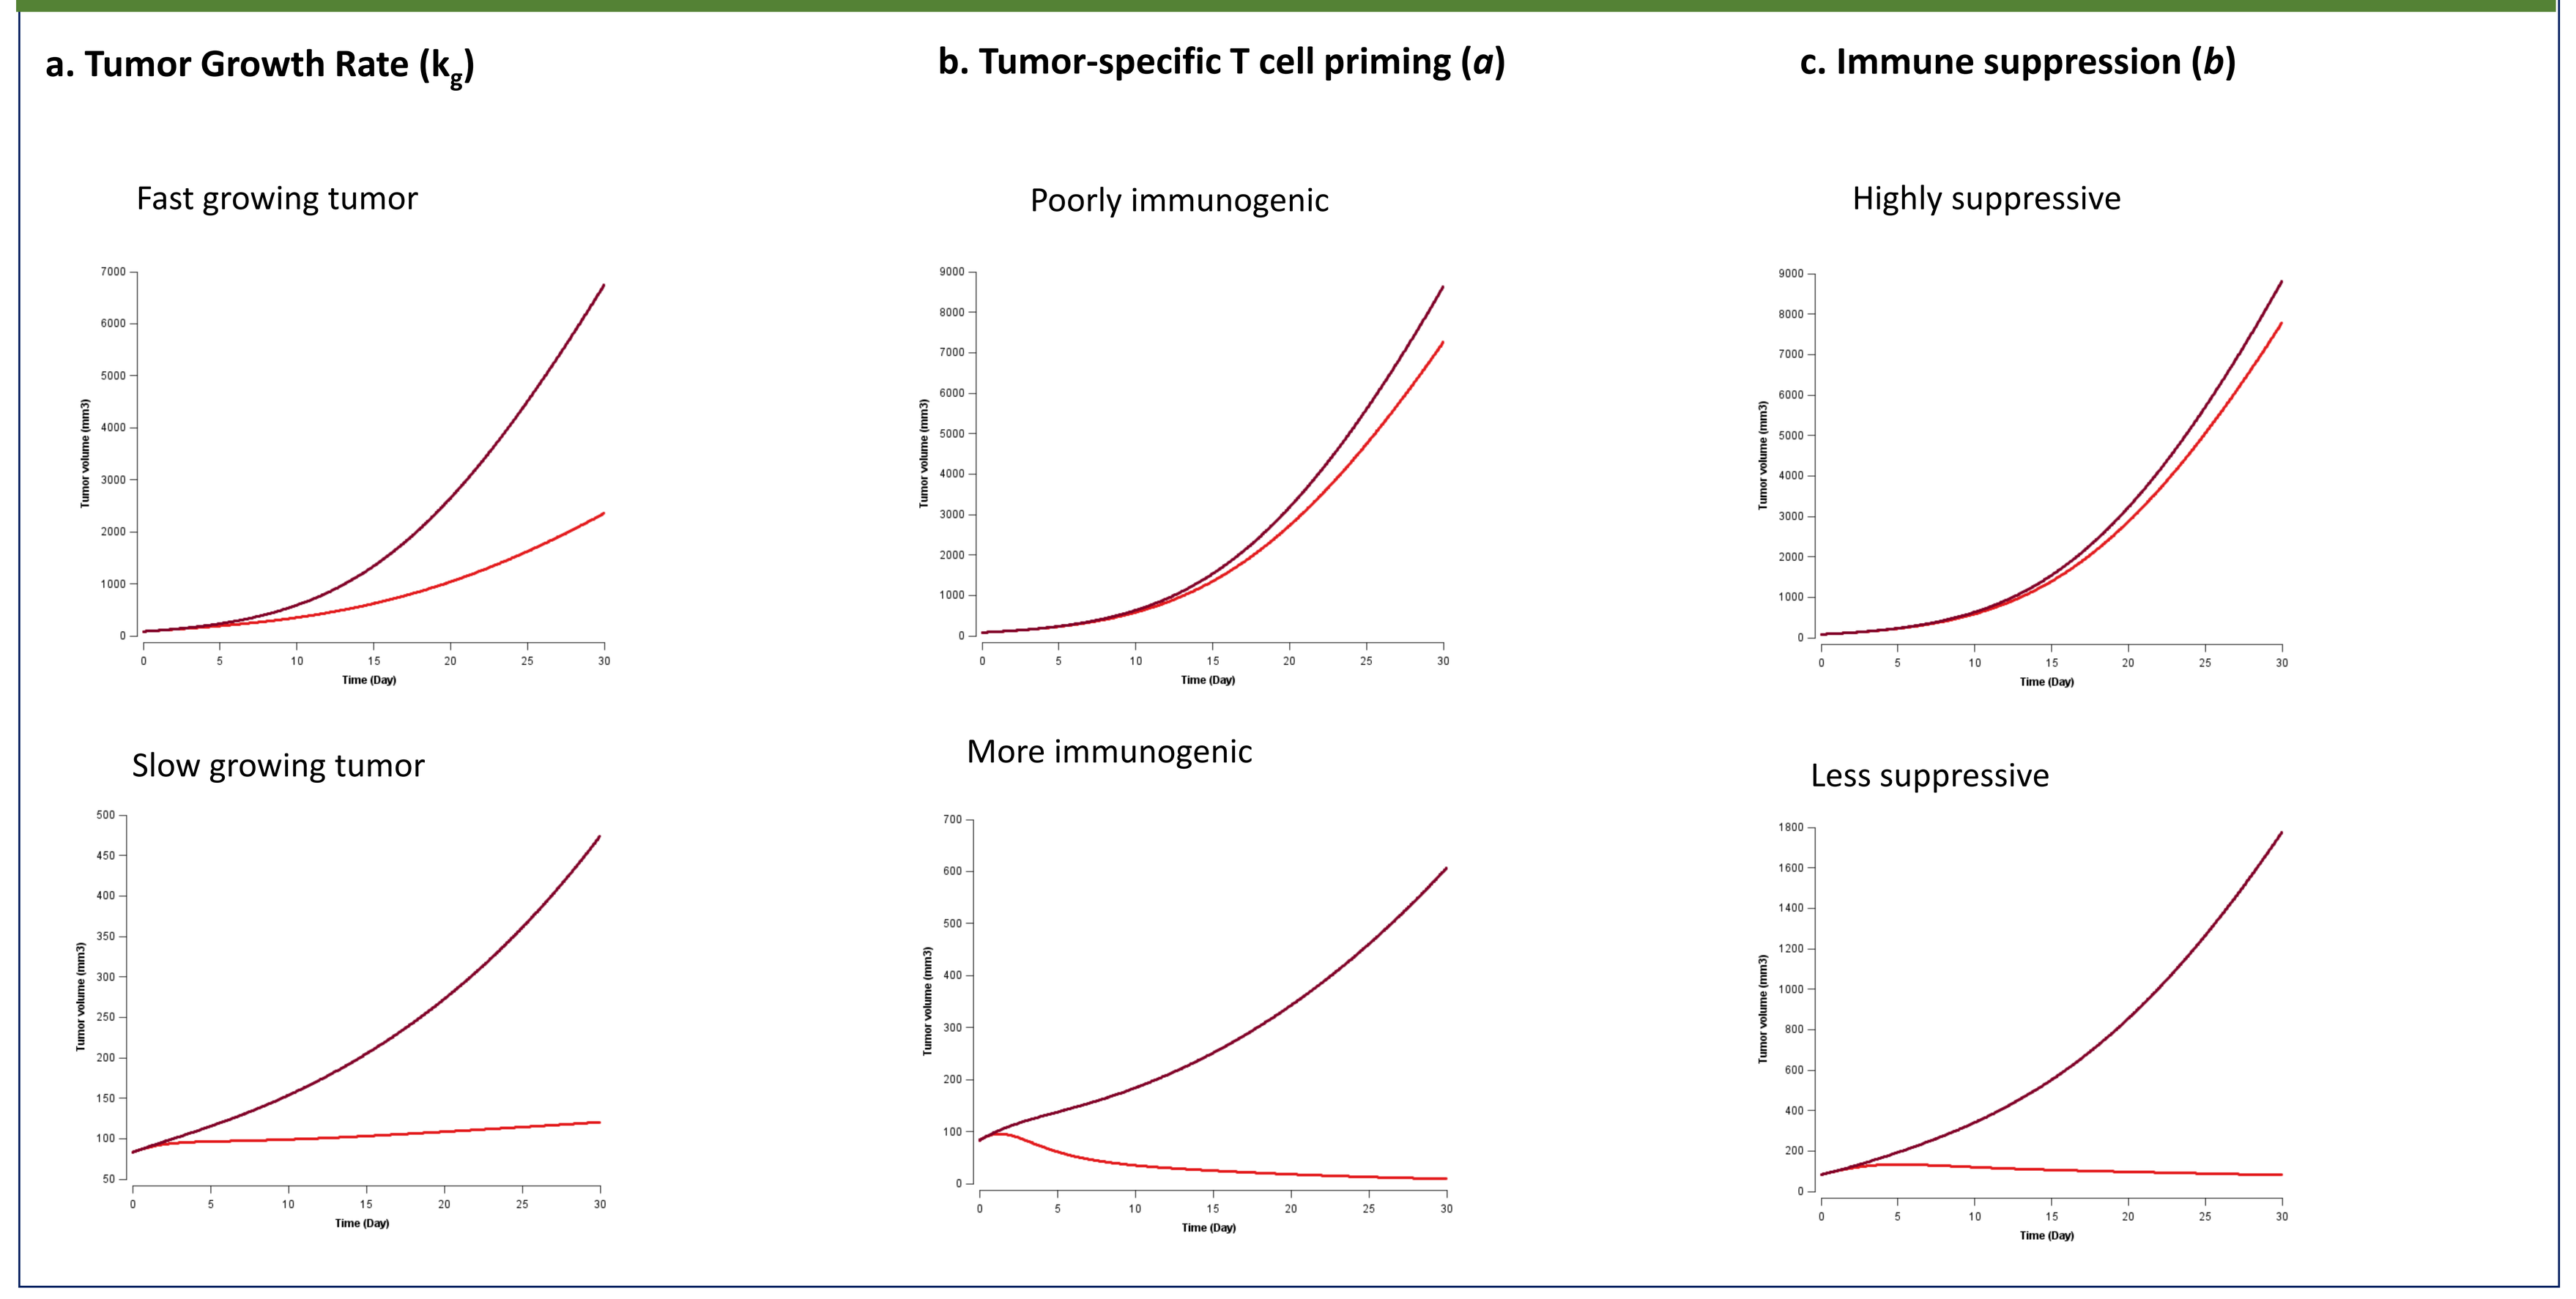
a. Tumor Growth Rate (kg)
b. Tumor-specific T cell priming (a)
c. Immune suppression (b)
Fast growing tumor
Poorly immunogenic
Highly suppressive
Slow growing tumor
More immunogenic
Less suppressive
Tumor volume (mm3)
Time (Day)

Figure 3. Simulations of the impact of tumor microenvironment on anti-tumor efficacy of HFB3-1 in TNFR2 KI mice. a. Improved tumor growth control can be achieved in slower growing tumors; b. Greater T cell priming and infiltration results in better tumor growth control; c. Less immune suppressive environment results in better tumor growth control. Brown trace is isotype control, red trace is HFB3-1 treatment.
Simulations with HFB10-1 produced similar effect.

Human Tumor Growth Inhibition Simulations

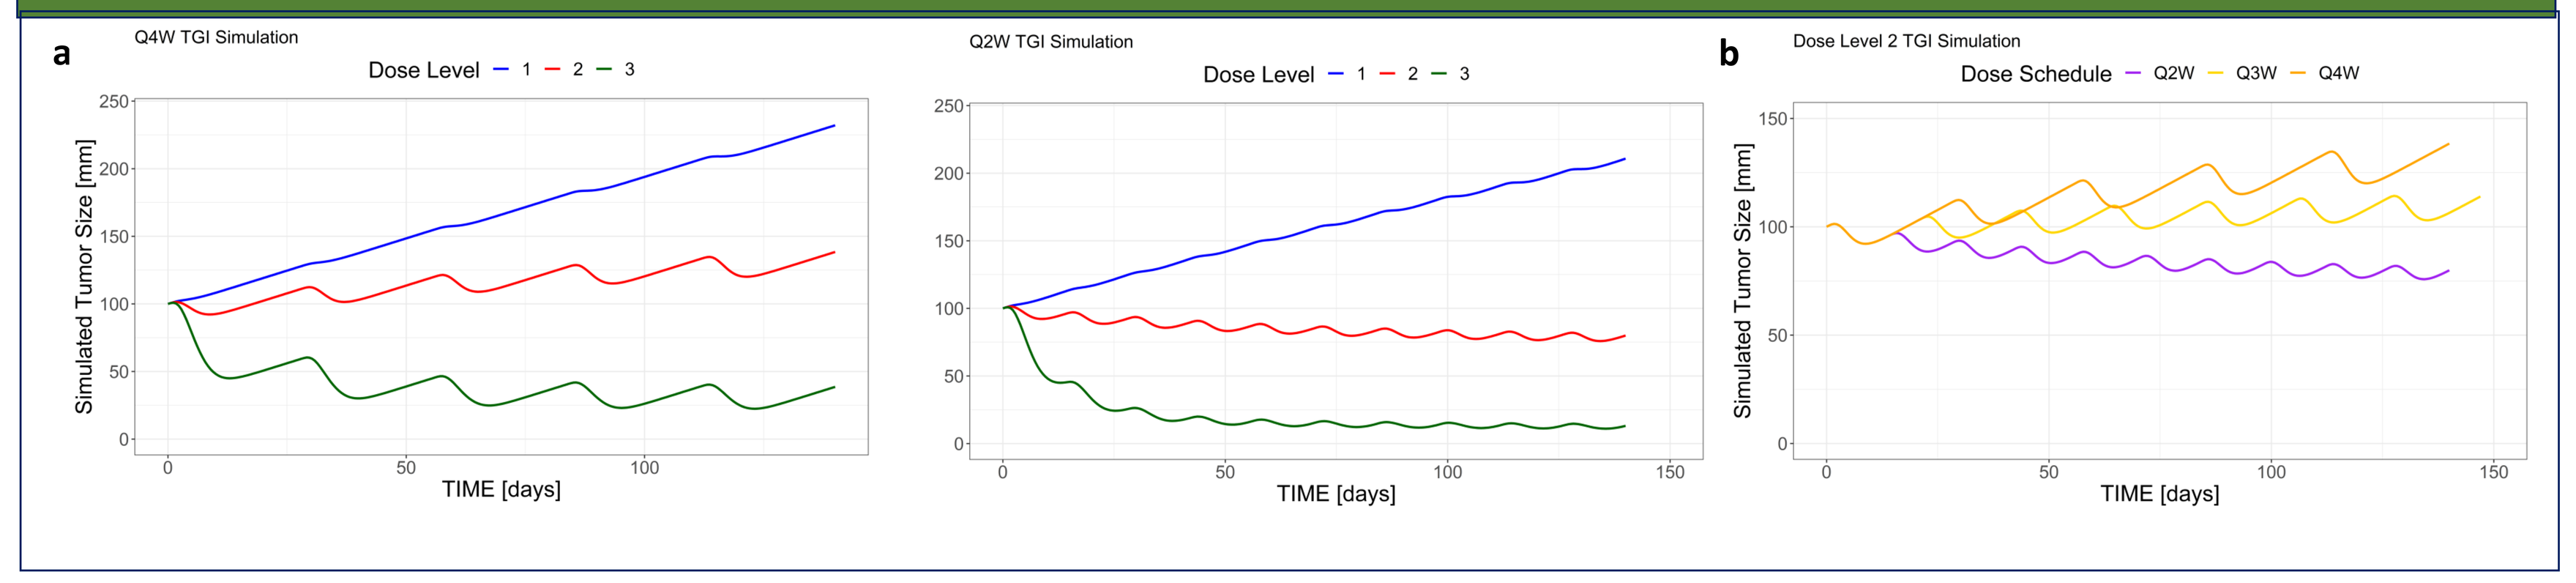
a
Q4W TGI Simulation
Dose Level 1 2 3
Simulated Tumor Size [mm]
TIME [days]
Q2W TGI Simulation
Dose Level 1 2 3
TIME [days]
b
Dose Level 2 TGI Simulation
Dose Schedule Q2W Q3W Q4W
Simulated Tumor Size [mm]
TIME [days]

Figure 4. Simulation of tumor growth inhibition at multiple doses and dosing frequencies of HFB200301. **a.** Simulated tumor growth inhibition at various doses at fixed frequencies; b. simulated tumor growth inhibition at various dosing schedules at a fixed dose.

DISCUSSION

- The anti-tumor efficacy of an immune agonist depends on many factors including agonist exposure and tumor penetration, target expression and engagement, as well as tumor characteristics such as growth rate, immune infiltration, and suppressive signaling in the TME.
- By incorporation of all these factors, our model reasonably described the PK, PD, and anti-tumor efficacy of two immune agonists.
- Simulations of tumor growth inhibition in humans indicate that both dose and frequency are important determinants in the anti-tumor activity of HFB200301, and this methodology may prove useful for optimal dose selection for further development.
- We believe this model has general applicability to immune modulators for cancer treatment.

SUMMARY

A semi-mechanistic PKPD model was successfully established:
- This model integrates PK (TMDD), PD (immune cell counts in the TME, receptor shedding and internalization), and tumor growth inhibition;
- This model confirms the importance of various factors in the TME for anti-tumor efficacy of immune agonists;
- This model allows for evaluation of different doses and dosing schedules of T cell agonists in cancer treatment;
- Future work will focus on the integration of TME and growth characteristics in individual tumor types, as well as modeling of combination therapy.

For additional information, please email contact@hifibio.com or visit Hifibio.com