E-Poster #2285
Presented at the
AACR Virtual Annual Meeting II
June 22-24, 2020

HFB301001, a novel OX40 agonistic antibody with a unique pharmacological profile and innovative biomarker strategy

Andreas Raue[1], Yun-Yueh Lu[1], Ouyang Li[1], Minmin Lu[1], Joyce Pi[1], Jia Wu[1], Mingfang Feng[1], Qian Zhang[1], Ross Fulton[1], Matthieu Delince[1], Juliana Crivello[1], Zachary Duda[1], Alexandra Staskus[1], Charina Ortega[1], Surendar Arumugam[1], Yuan Wang[2], Ruina Jin[2], Hongkai Zhang[2], Pascaline Mary[1], Nicola Beltraminelli[1], Francisco Adrian[1], Liang Schweizer[1]

[1]HiFiBiO Therapeutics; [2]College of Life Sciences, Nankai University, China

HIGHLIGHTS

Lead Antibody	Target cells	MOA	Indications
Fully human IgG1 with cyno cross-reactivity	T Cells	T cells stimulation	Advanced solid tumors

Unique pharmacological profile addresses limitations of first-generation OX40 antibodies

- Avoids interference with endogenous signaling and possibility of synergy with the ligand signal
- Minimizes receptor downregulation and potential for better target engagement
- ➢ Demonstrates more potent anti-tumor activity than benchmark in vivo

Innovative predictive biomarker strategy leverages single-cell technology to define responding patients

- PD-1 research suggests importance of specific T cell clonotypes for responder patients
- Single-cell profiling can identify T cell phenotypes and clonotypes associated with activity of HFB301001
- ➢ Provides hypothesis for biomarker strategy in clinical trial to enrich for responding patients

SUMMARY

OX-40 (CD134, TNFRSF4) is a tumor necrosis factor (TNF) receptor expressed primarily on activated CD4+ and CD8+ T cells and transmits a potent costimulatory signal when engaged. Targeting OX40 with an agonistic antibody has been demonstrated to increase the activity of T cells leading to anti-tumor responses. Several agonistic antibodies against OX40 have been evaluated in the clinical trials with good tolerability. However, so far, limited clinical activities have been observed in the reported clinical trials.

We have developed a novel OX40 antibody (HFB301001) with a unique pharmacological profile and biomarker strategy to address the limitations of previous OX40 agonistic antibodies. Unlike other OX40 antibodies, HFB301001 does not block the binding of OX40 ligand (OX40L) and therefore does not compete with the endogenous signaling. Furthermore, in contrast to other anti-OX40 antibodies, treatment with HFB301001 does not result in significant reduced expression of OX40 on T cells providing a potential for better target engagement. HFB301001 demonstrated more potent in vivo anti-tumor activity in a preclinical mouse model as compared to a previously published anti-OX40 antibody that is in the clinical stage. Our data suggests that HFB301001 may provide superior benefit for patients compared to first generation of OX40 antibodies.

Additionally, we present a novel concept for identifying potential responding patient to HFB301001 using HiFiBiO's proprietary Drug Intelligent Science (DIS™) platform. The DIS approach for discovery of predictive response biomarkers combines high-throughput single-cell profiling of a patient's T cell repertoire with functional read-outs to characterize tumor-specific T cell clonotypes associated with response to HFB301001. Our results provide the foundation for the implementation of the DIS™ platform to guide the clinical development of HFB301001 for selected patients that are most likely to benefit from the treatment.

HFB301001 is being developed as a potential novel treatment option for cancer coupled with a patient stratification biomarker.

LIMITATIONS OF PREVIOUS OX40 ANTIBODIES

1) Pharmacological profile did not optimally leverage the target for activity

Targeting OX40 with antibodies is a well described therapeutic strategy that can lead to increased activity of T cells and significant anti-tumor activity. Most of the previous OX40 antibodies that entered the clinics compete with the endogenous ligand signaling which is counterproductive for the goal of providing a better stimulation of T cells. In addition, receptor downregulation has been described as limiting factor of previous antibodies that made appropriate dose and schedule selection difficult in the clinic[1]. We have identified an OX40 antibody with a unique binding epitope that addresses these limitations.

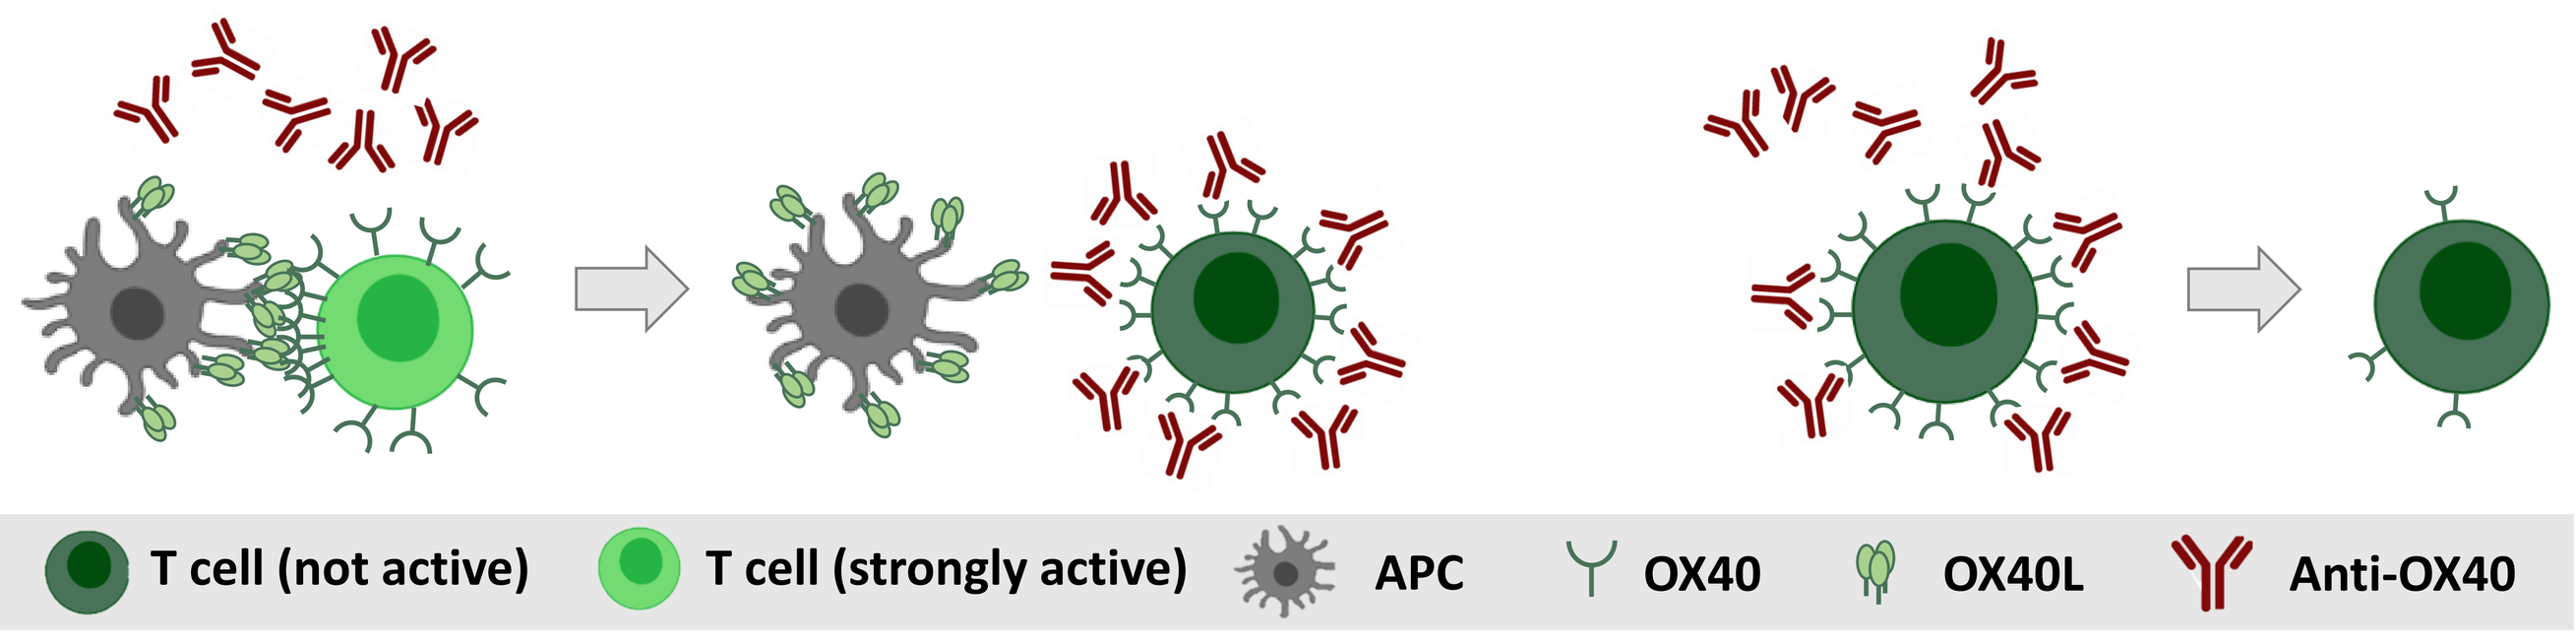
Interference with endogenous ligand signaling
Receptor downregulation
T cell (not active)
T cell (strongly active)
APC
OX40
OX40L
Anti-OX40

2) Trials in unselected patient population, no biomarker strategy

It has been hypothesized that clinical response to OX40 agonism may be driven by the expansion of select anti-tumor T cell clones rather than a broad expansion of T cell clones in the peripheral blood[2]. However, to date no predictive response biomarker has been identified and validated.

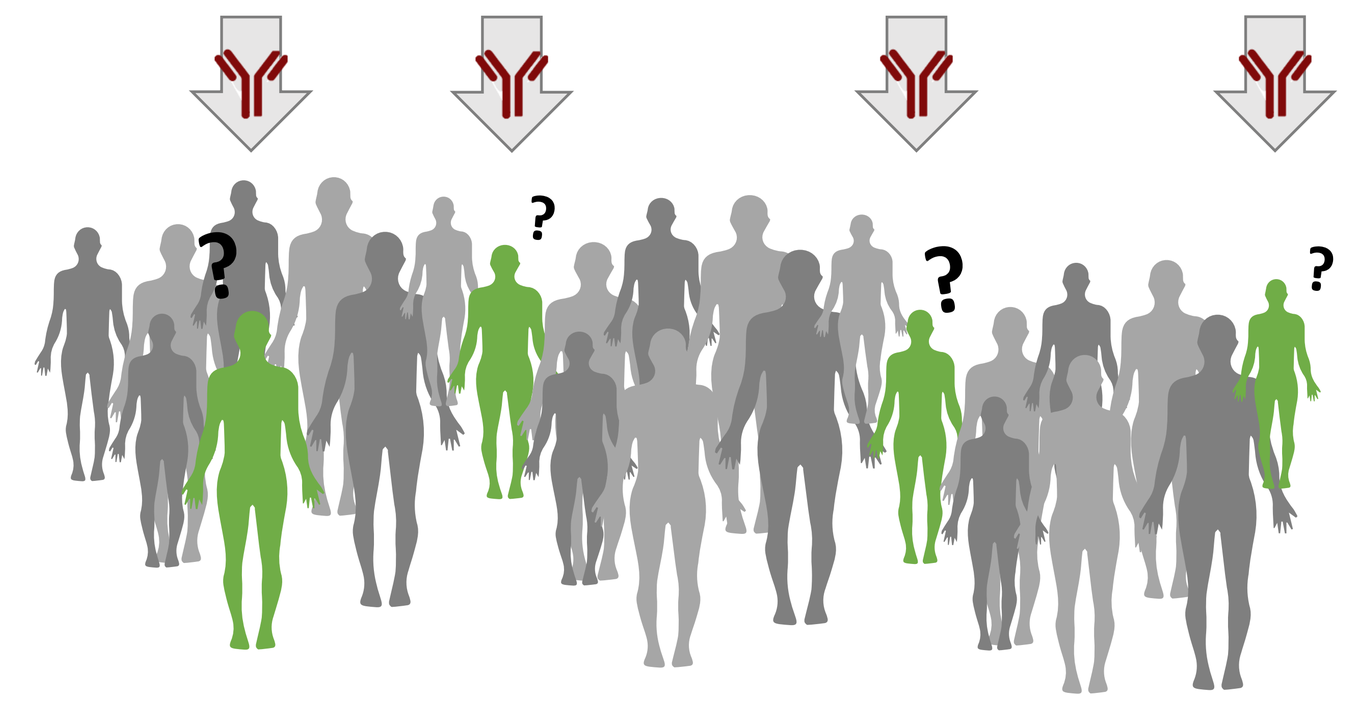

1. Wang, R. et al. Clinical Cancer Research (2019) An Integrative Approach to Inform Optimal Administration of OX40 Agonist Antibodies in Patients with Advanced Solid Tumors. doi:10.1158/1078-0432.CCR-19-0526.

2. Diab, A. *et al.* Cancer Res (2018) Abstract CT010: Pharmacodynamic (PD) changes in tumor RNA expression and the peripheral blood T cell receptor (TCR) repertoire in a phase I study of OX40 agonistic monoclonal antibody (mAb) PF-04518600 (PF-8600). doi:10.1158/1538-7445.am2018-ct010

RESULTS

HFB301001 binds to a unique epitope on OX40

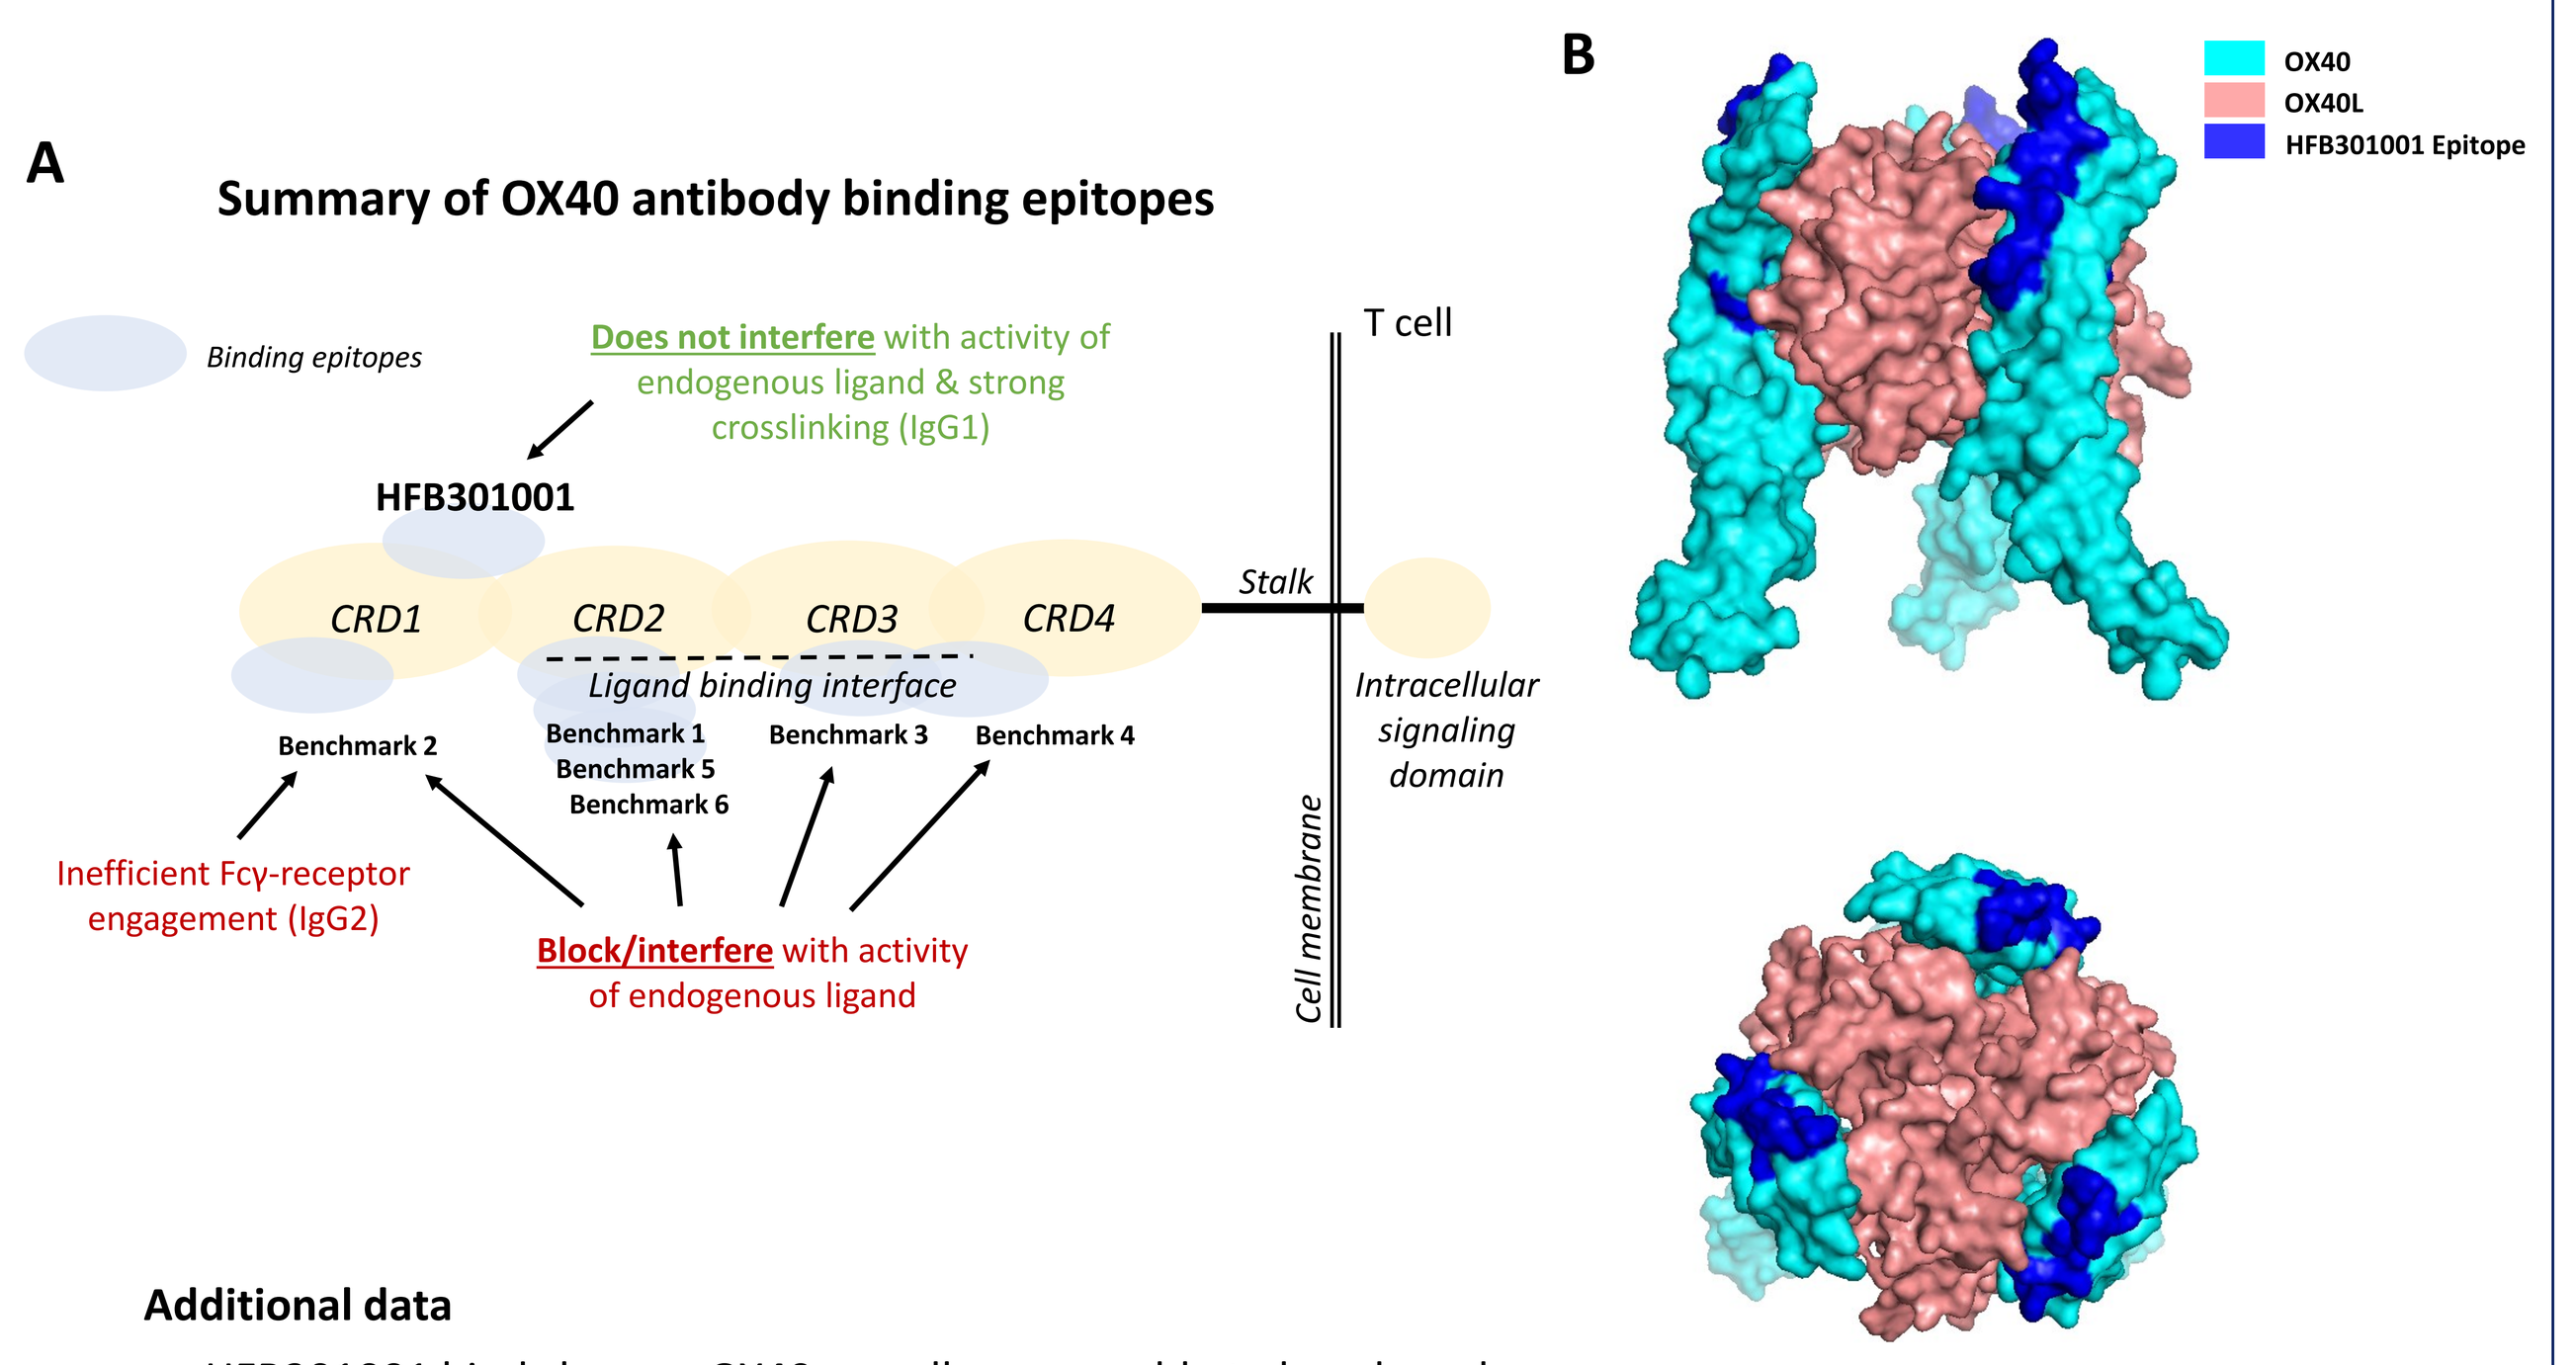
A
Summary of OX40 antibody binding epitopes
Binding epitopes
Does not interfere with activity of endogenous ligand & strong crosslinking (IgG1)
HFB301001
CRD1
CRD2
CRD3
CRD4
Stalk
T cell
Ligand binding interface
Benchmark 2
Benchmark 1
Benchmark 5
Benchmark 6
Benchmark 3
Benchmark 4
Intracellular signaling domain
Cell membrane
Inefficient Fcγ-receptor engagement (IgG2)
Block/interfere with activity of endogenous ligand
B
OX40
OX40L
HFB301001 Epitope

Additional data

- HFB301001 binds human OX40 on cells comparable to benchmarks

Figure 1. (A) Illustration of the OX40 receptor domains and binding epitopes of different OX40 antibodies. Binding epitopes were characterized by epitope binning using a competitive ELISA assay to explore the OX40 binding epitopes of different anti-OX40 antibodies and OX40L. **(B)** We used hydrogen deuterium exchange mass spectrometry to get a more detailed resolution on the binding epitopes. The data shows the binding epitope of HFB301001 on the outer face of OX40 towards the C-terminus, not overlapping with the ligand binding interface.

HFB301001 does not interfere with OX40L, unlike benchmarks

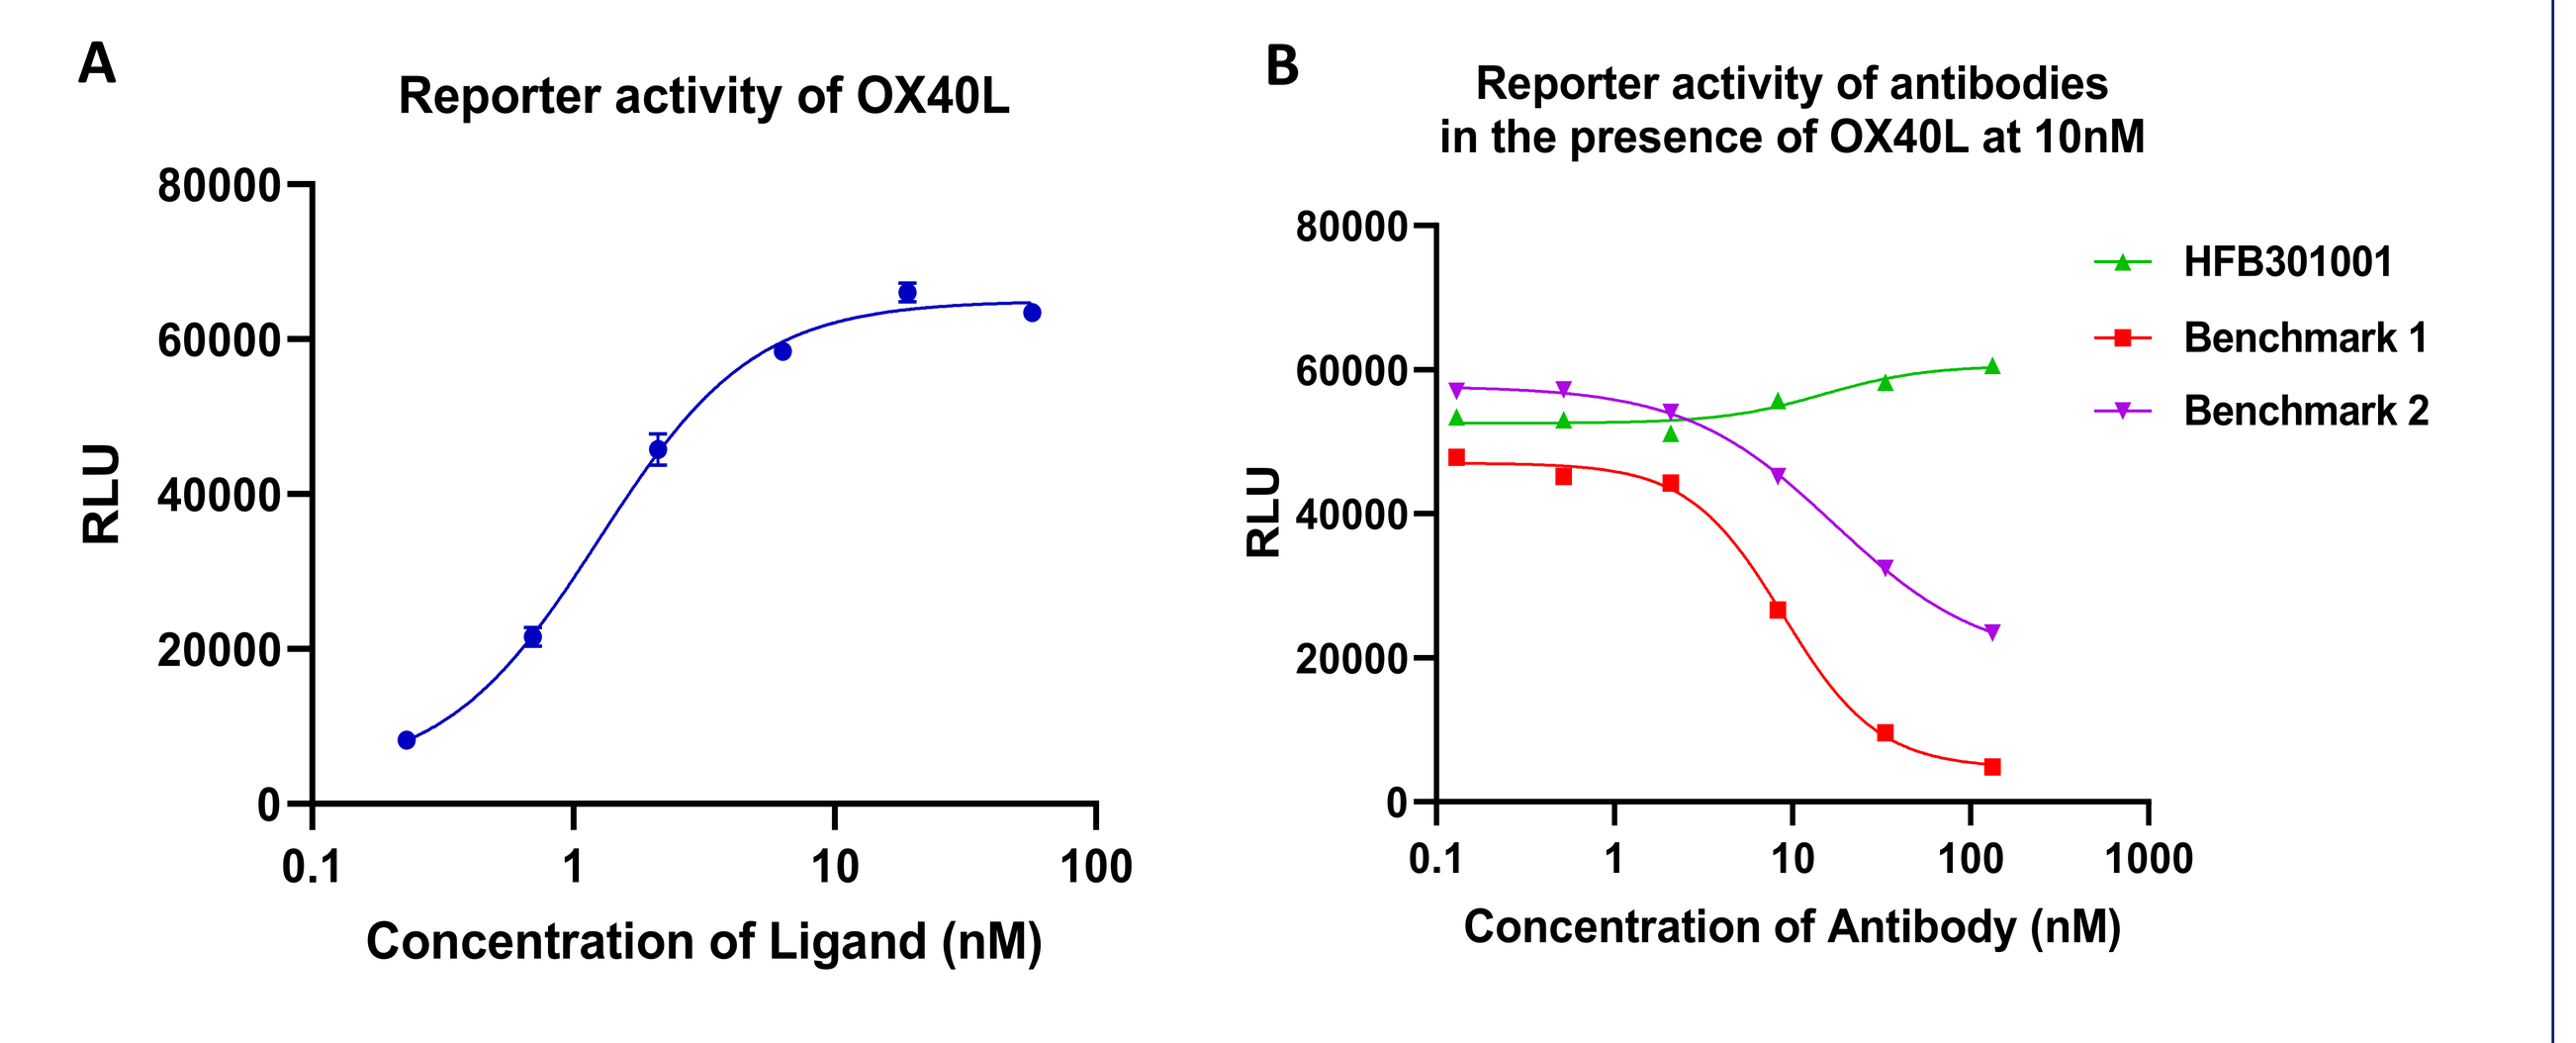
A
Reporter activity of OX40L
RLU
Concentration of Ligand (nM)
B
Reporter activity of antibodies in the presence of OX40L at 10nM
HFB301001
Benchmark 1
Benchmark 2
Concentration of Antibody (nM)

Additional data

- HFB301001 demonstrated desired agonistic activities using a reporter cell assay or peripheral human T cells

Figure 2. (A) OX40 activity assay using a luciferase NF-κB reporter system. OX40 ligand activity was demonstrated in this system with EC80 of approximately 10 nM. **(B)** To investigate the impact of anti-OX40 antibodies on agonistic activity of OX40L, cells were inculcated with anti-OX40 antibodies in the presence of 10nM OX40L. Both Benchmark 1 and Benchmark 2 blocked the agonistic effect of OX40L in a dose-dependent manner, but HFB301001 did not.

HFB301001 does not lead to receptor downregulation, unlike benchmarks

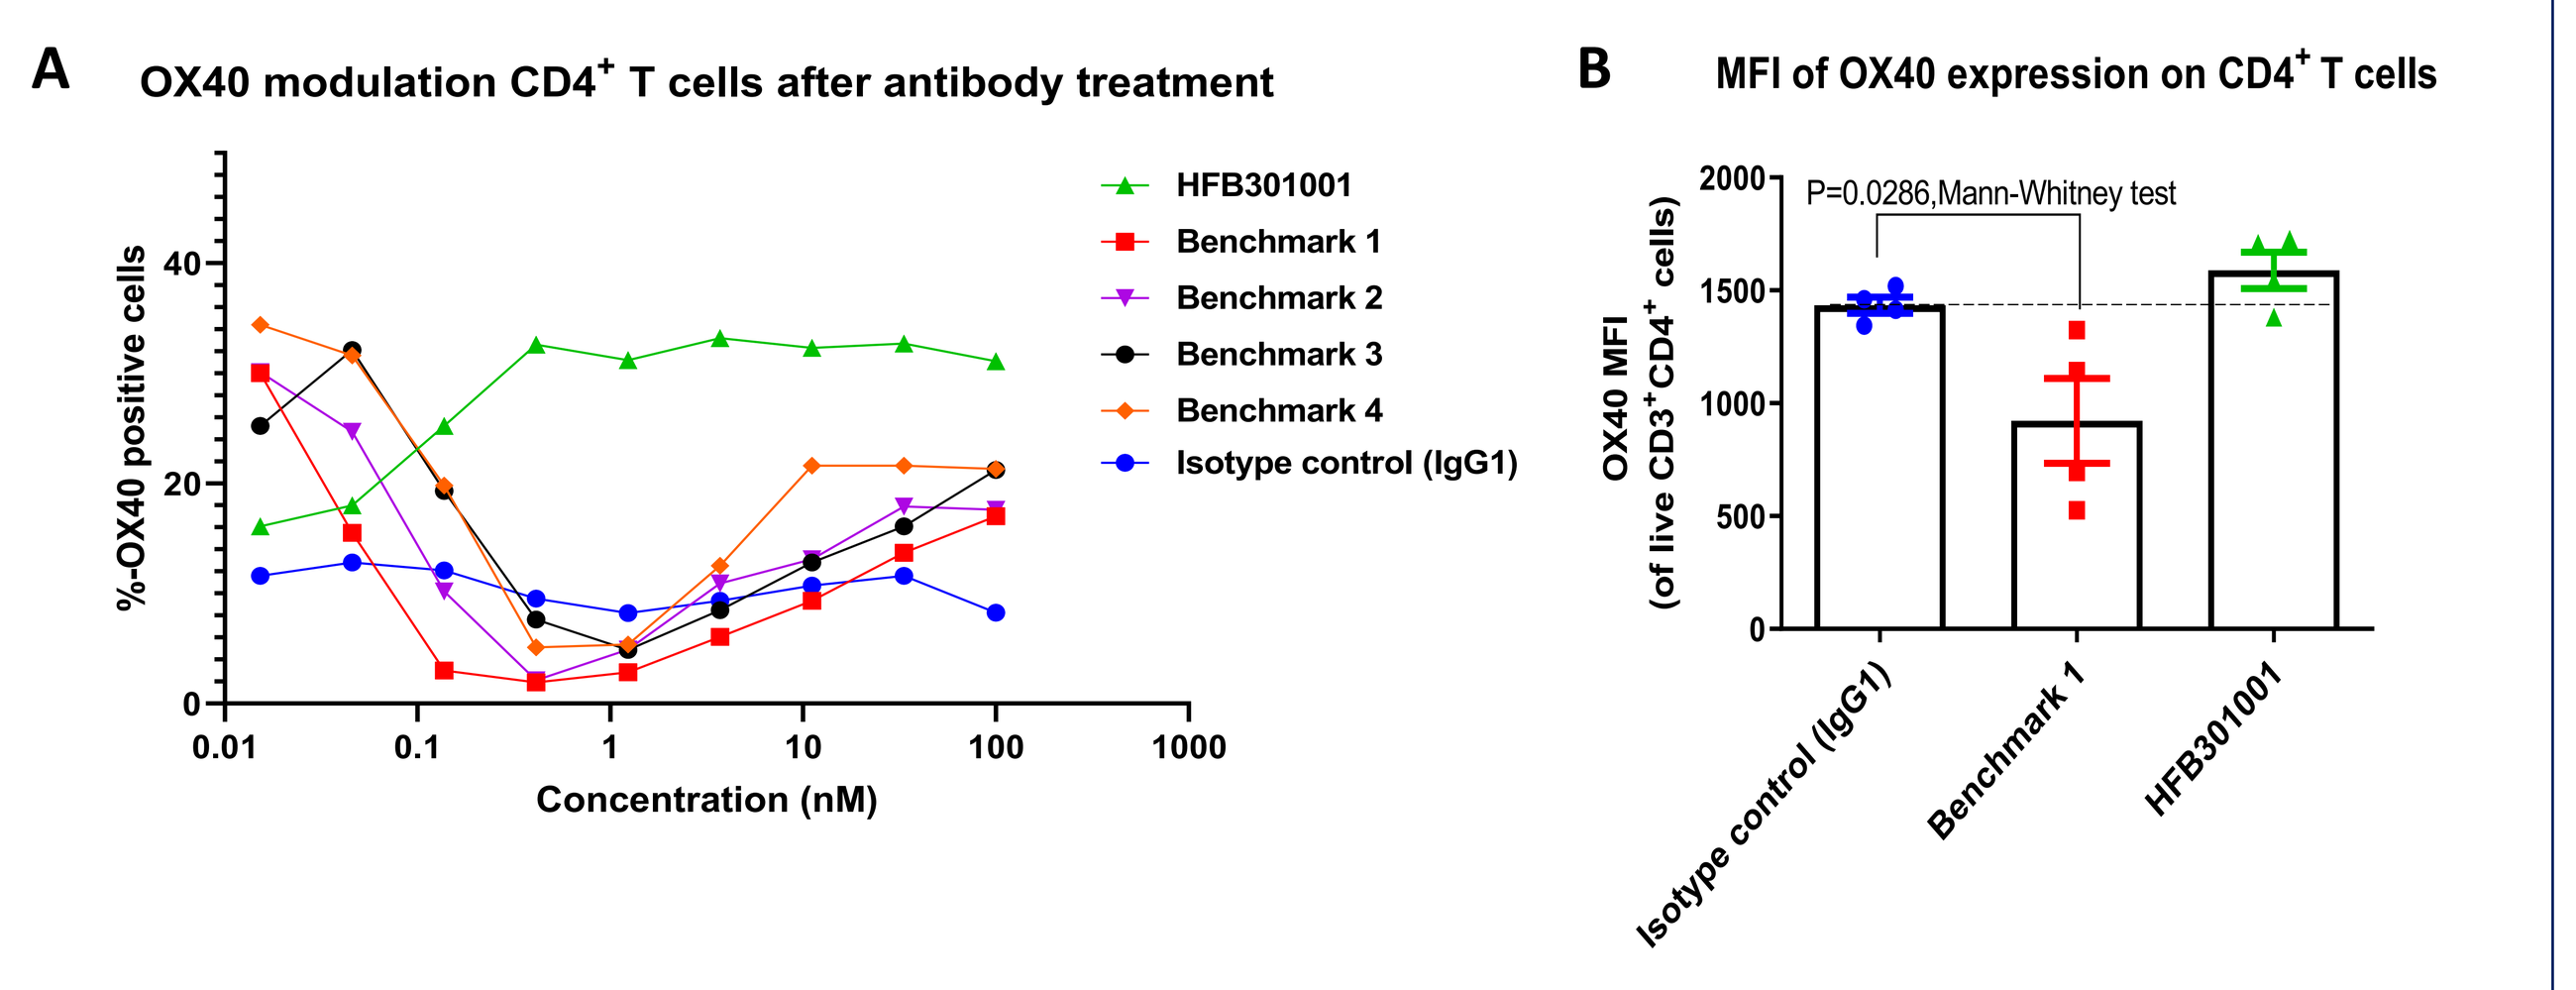
A
OX40 modulation CD4+ T cells after antibody treatment
%-OX40 positive cells
Concentration (nM)
HFB301001
Benchmark 1
Benchmark 2
Benchmark 3
Benchmark 4
Isotype control (IgG1)
B
MFI of OX40 expression on CD4+ T cells
P=0.0286,Mann-Whitney test
OX40 MFI (of live CD3+CD4+ cells)
Isotype control (IgG1)
Benchmark 1
HFB301001

C Reduced receptor downregulation may be a results of binding parameters and epitope

Antibodies	Kinetic Binding Parameters					
	Exp A			Exp B		
	k_a (1/Ms)	k_d (1/s)	K_D (M)	k_a (1/Ms)	k_d (1/s)	K_D (M)
HFB301001	9.7E+04	4.3E-04	4.5E-09	9.7E+04	4.3E-04	4.4E-09
Benchmark 1	1.5E+05	7.4E-05	4.9E-10	9.8E+04	5.9E-05	6.0E-10
Benchmark 2	1.1E+05	6.1E-05	5.8E-10	1.5E+05	7.6E-05	5.1E-10

Figure 3. (A) %-OX40 positive T cells were measured after treatment with anti-OX40 antibodies by flow cytometry. Compared benchmarks, HFB301001 enhanced OX40 expression which stably remained on the cell surface at higher concentrations. **(B)** OX40 expression on CD4+ T cells measured by flow cytometry 24 hours after third treatment with 10 mg/Kg of anti-OX40 antibodies in MC-38 murine colorectal cancer model in human OX40 (hOX40) knock-in (KI) mice. Interestingly, Benchmark 1, but not HFB301001, induced significant downregulation of OX40 expression on CD4+ T cells in blood. **(C)** Kinetic binding parameters for the interaction of HFB301001 with human OX40 was assessed using Bio-Layer Interferometry. HFB301001 bound specifically to recombinant human OX40-mFc protein with single digit nM affinity and showed faster dissociation rate compared to benchmark antibodies.

HFB301001 is more active in MC38 tumor model than benchmark

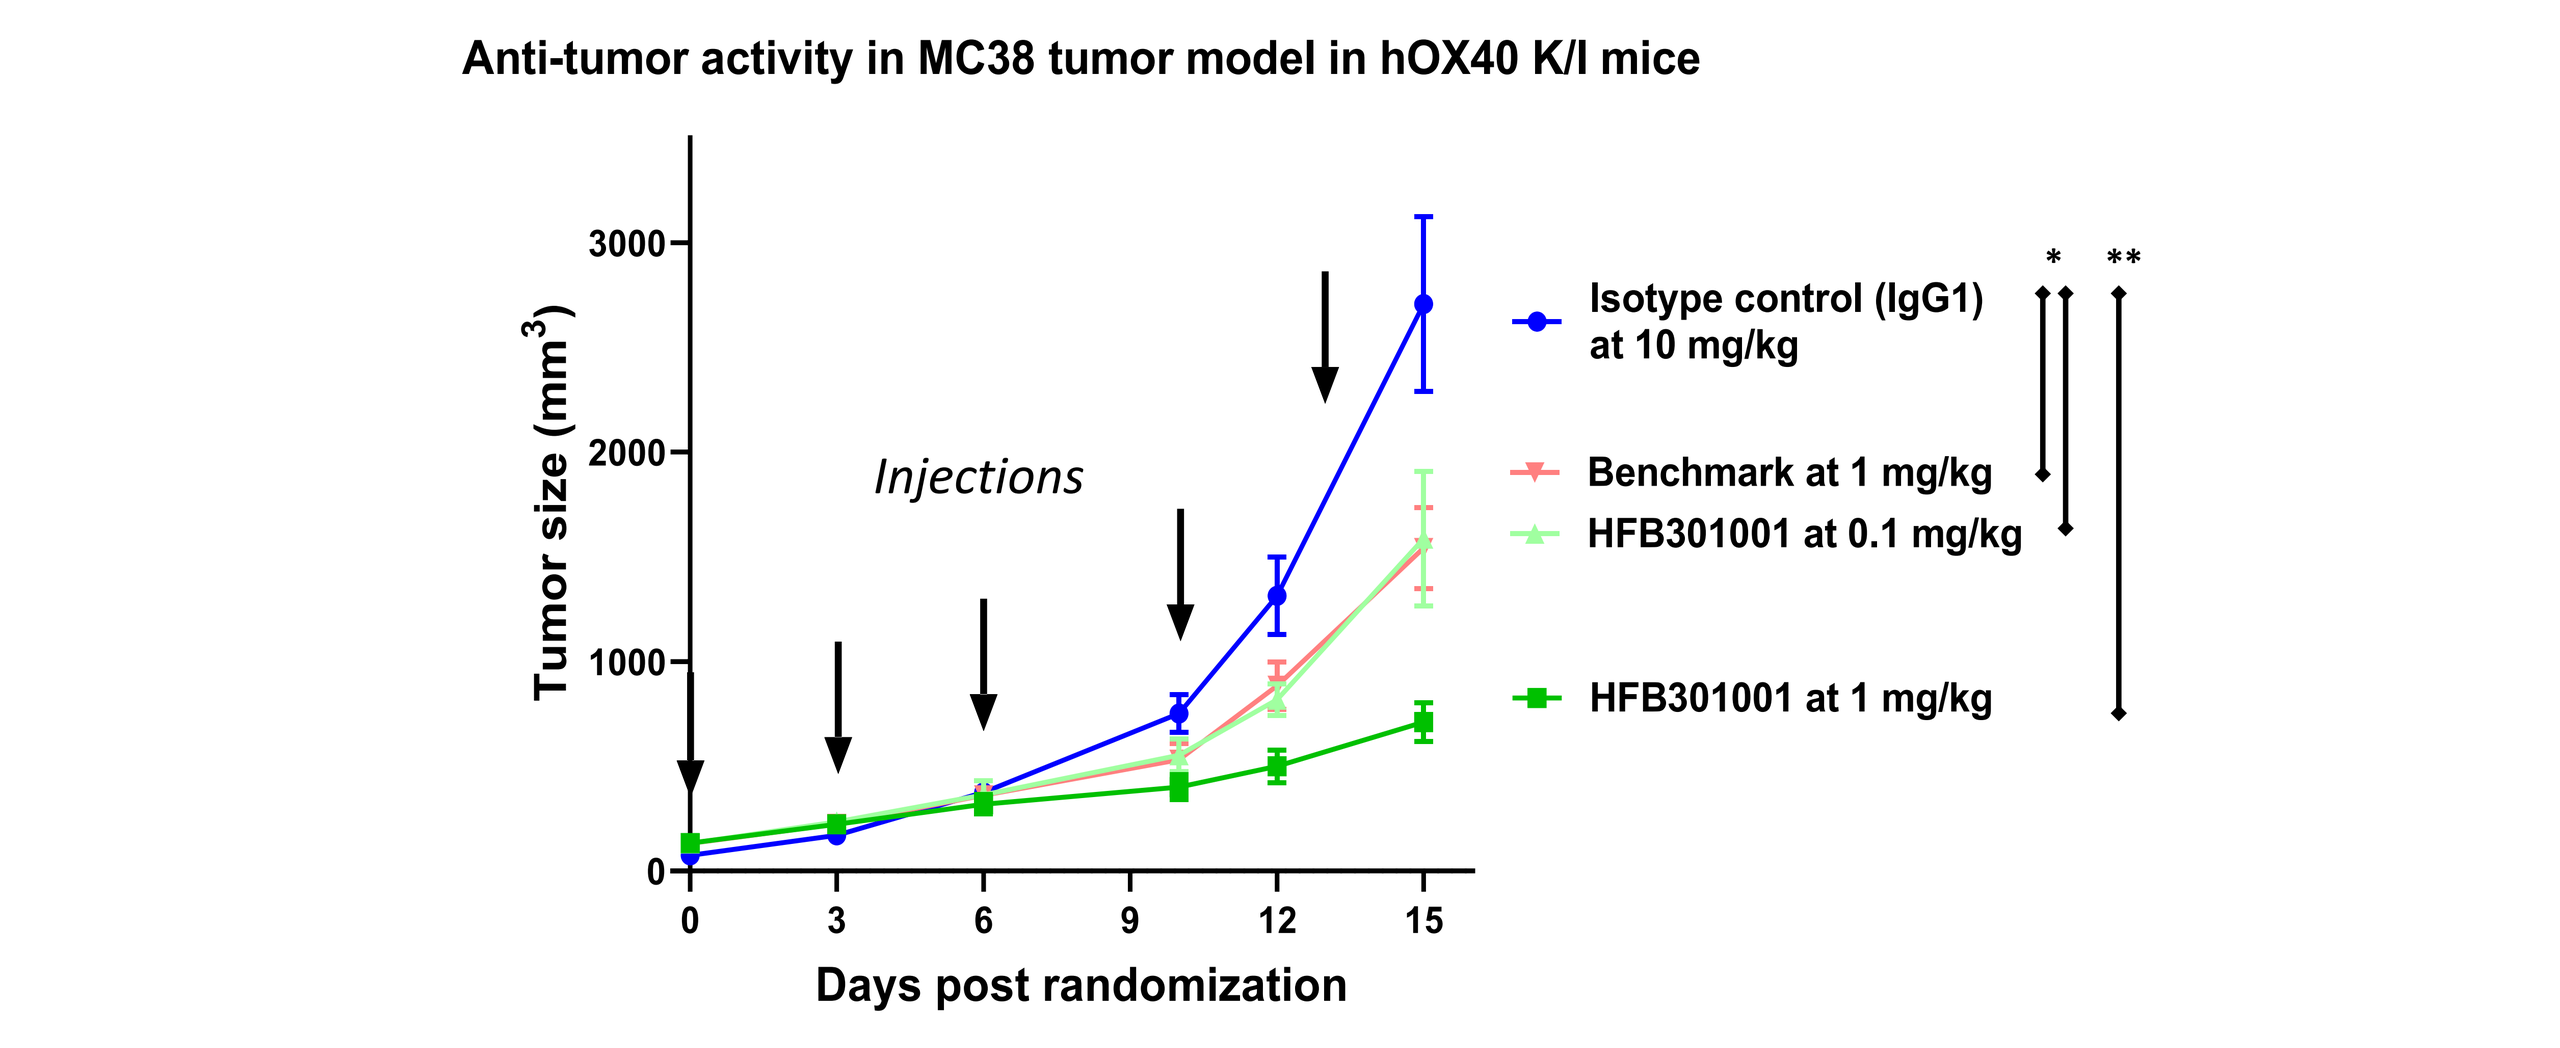
Anti-tumor activity in MC38 tumor model in hOX40 K/I mice
Tumor size (mm3)
Injections
Days post randomization
Isotype control (IgG1) at 10 mg/kg
Benchmark at 1 mg/kg
HFB301001 at 0.1 mg/kg
HFB301001 at 1 mg/kg
*
**

Figure 4. In vivo anti-tumor activity of anti-OX40 antibodies in MC-38 tumor model in hOX40 K/I mice. The treatment time points are indicated by arrows, error bars indicate standard error of mean, significance calculated by one-way ANOVA on last time point (*: $p_{val}<0.05$, **: $p_{val}<0.01$). Under these conditions, HFB301001 lead to significant tumor growth inhibition compared to control and was superior to Benchmark 1.

HFB301001 leads to increased survival in MC38 tumor model compared to benchmark

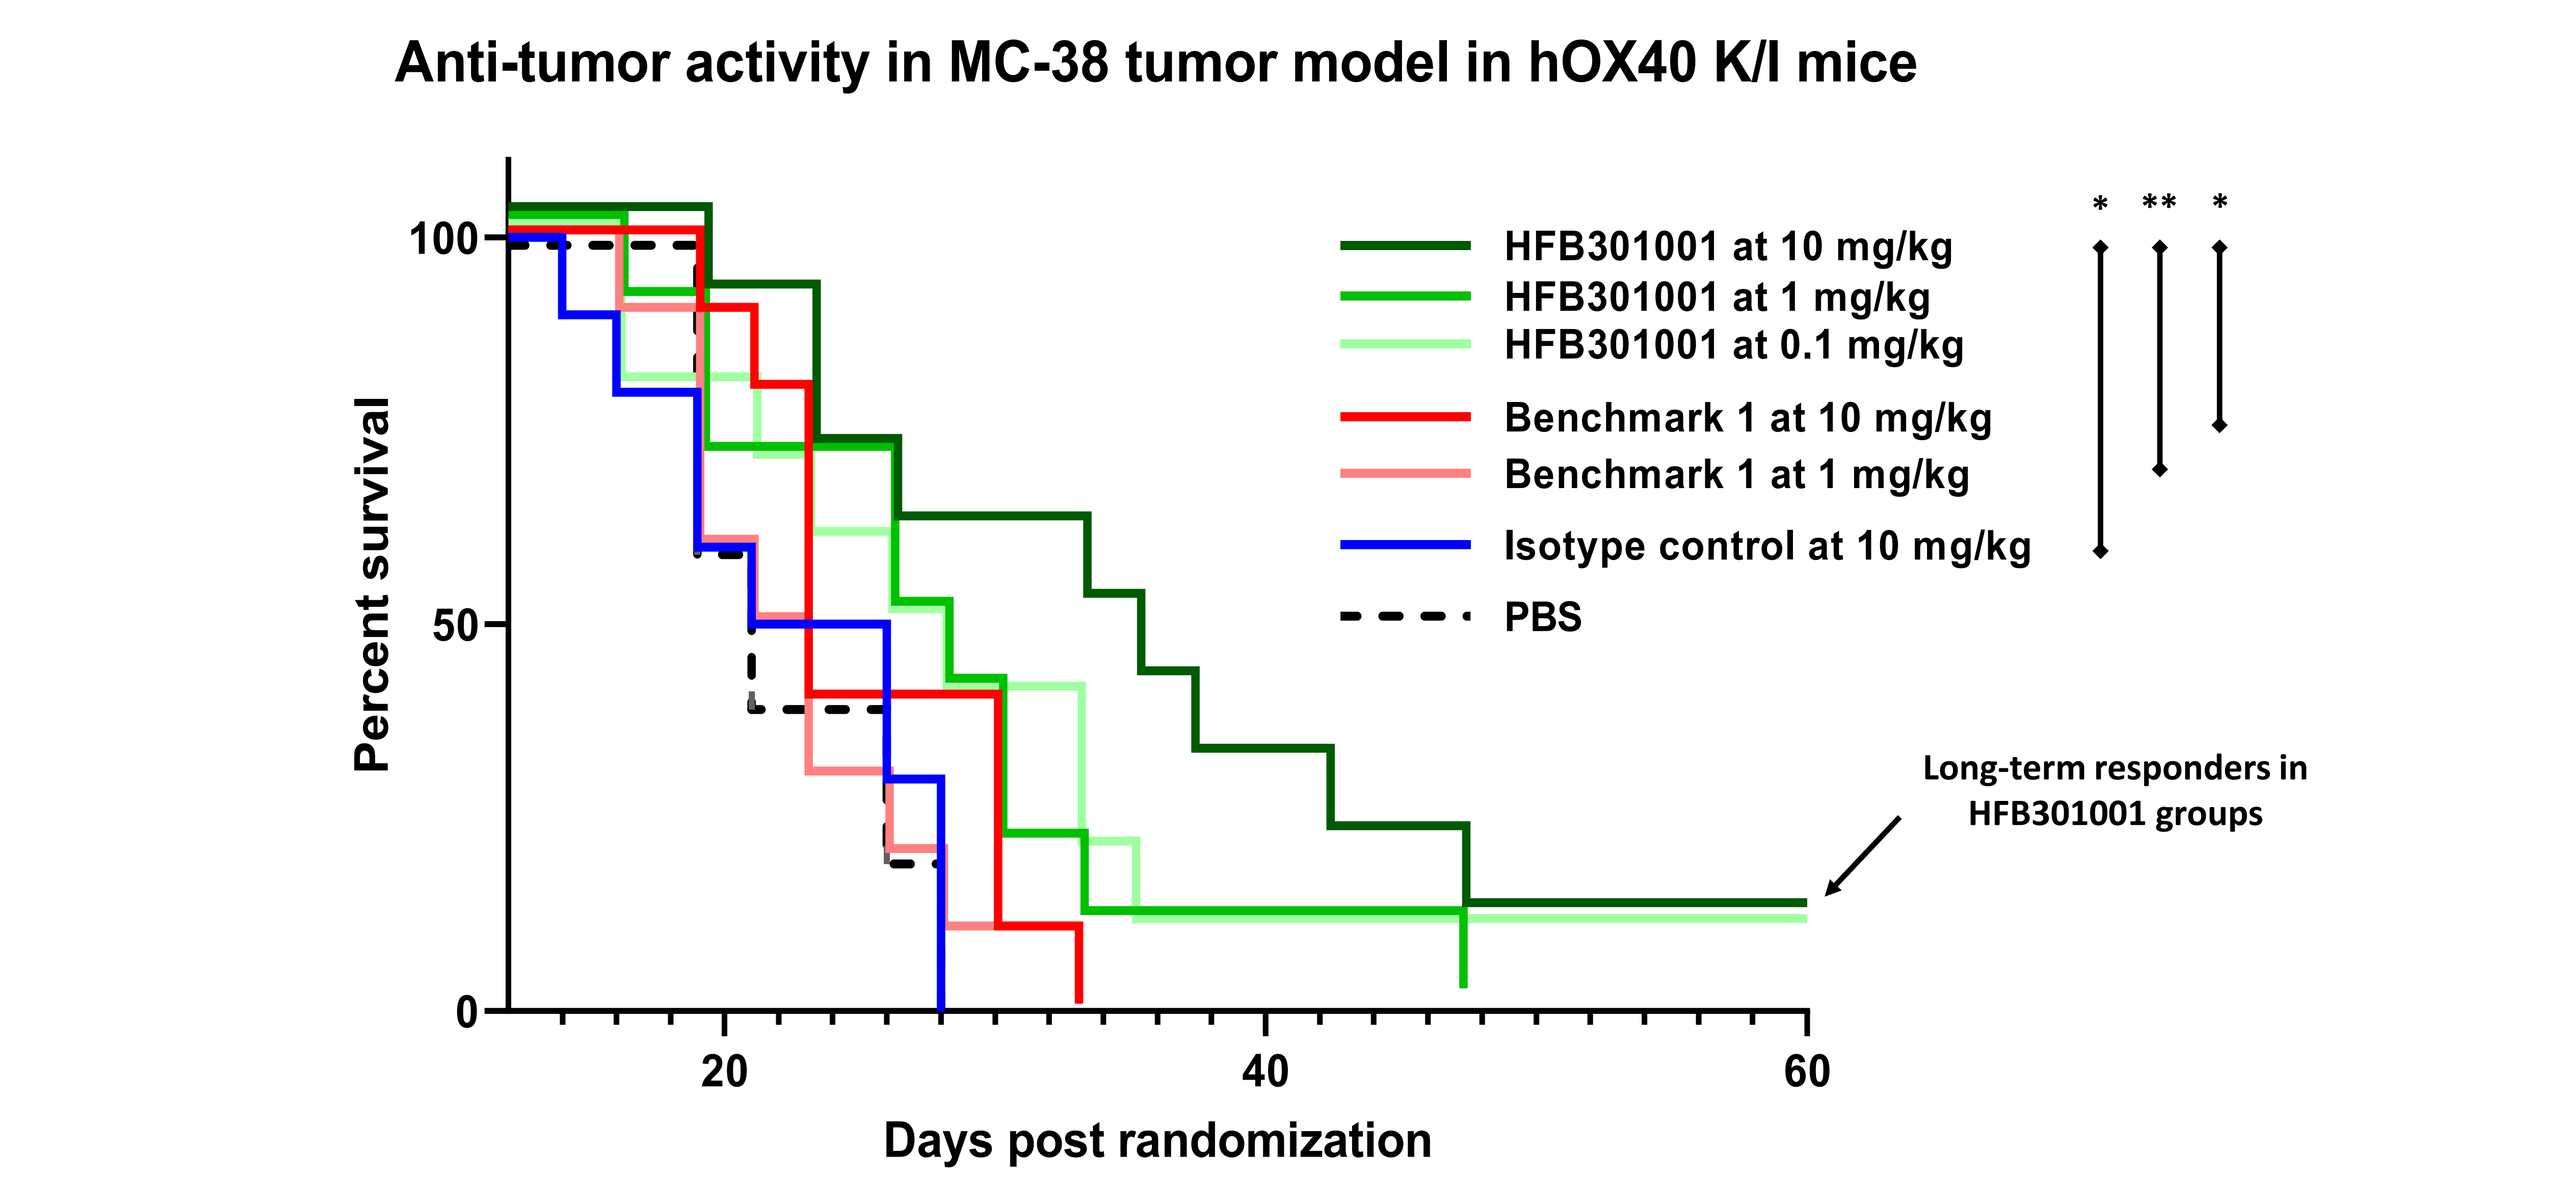
Anti-tumor activity in MC-38 tumor model in hOX40 K/I mice
Percent survival
Days post randomization
HFB301001 at 10 mg/kg
HFB301001 at 1 mg/kg
HFB301001 at 0.1 mg/kg
Benchmark 1 at 10 mg/kg
Benchmark 1 at 1 mg/kg
Isotype control at 10 mg/kg
PBS
Long-term responders in HFB301001 groups

Figure 5. Survival study of anti-OX40 antibodies in MC-38 tumor model in hOX40 K/I mice. All treatments were administered by i.p. injections on D0, D3, D6, D10, and D13. Significance was calculated by Log-rank test (*: $p_{val}<0.05$, **: $p_{val}<0.01$). Under these conditions HFB301001 induced significant survival benefit compared to control and Benchmark 1.

HFB301001 treatment leads to PD modulation consistent with superior activity

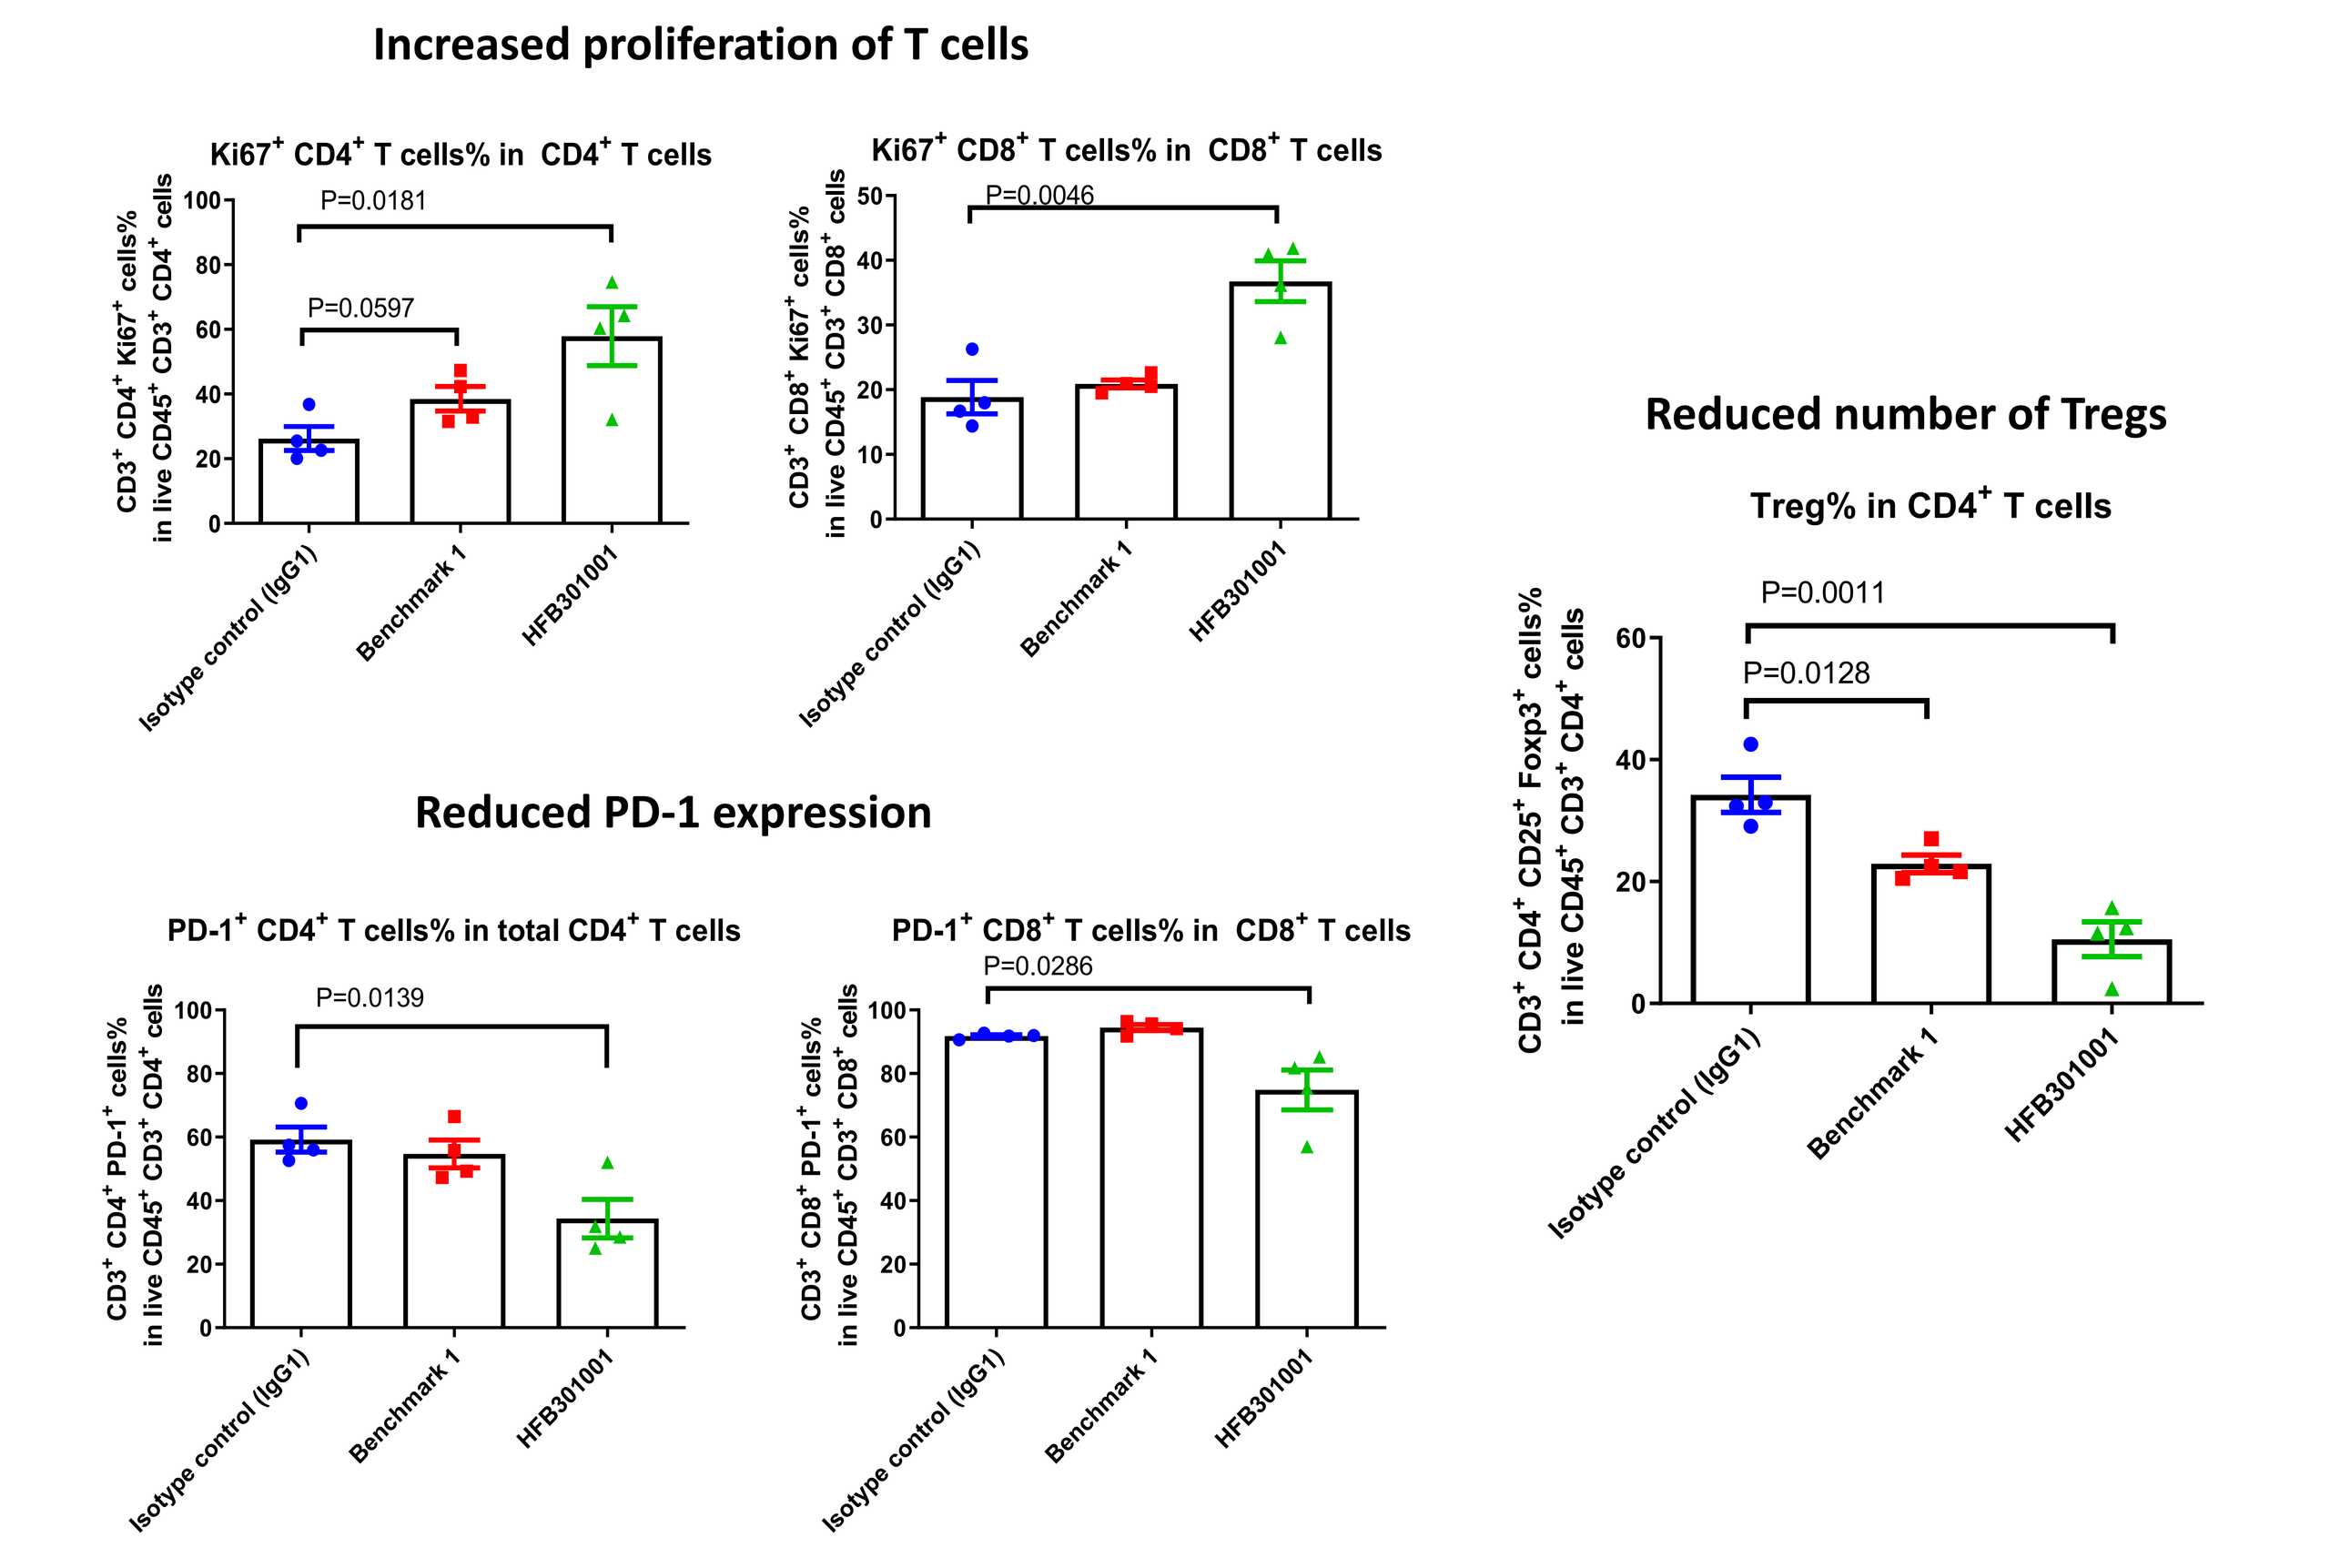
Increased proliferation of T cells
Ki67+ CD4+ T cells% in CD4+ T cells
P=0.0181
P=0.0597
Ki67+ CD8+ T cells% in CD8+ T cells
P=0.0046
Reduced PD-1 expression
PD-1+ CD4+ T cells% in total CD4+ T cells
P=0.0139
PD-1+ CD8+ T cells% in CD8+ T cells
P=0.0286
Reduced number of Tregs
Treg% in CD4+ T cells
P=0.0011
P=0.0128
Isotype control (IgG1)
Benchmark 1
HFB301001

Additional data

- No significant reductions in CD8+ or conventional CD4+ cells observed
- Upregulation of OX40L and CD86 on DCs, HFB301001 works cooperatively with the ligand

Figure 6. Changes induce in tumor T cells after treatment with anti-OX40 antibodies measured by flow cytometry 24 hours after third treatment with 10 mg/Kg of anti-OX40 antibodies in MC-38 murine colorectal cancer model in human OX40 (hOX40) knock-in (KI) mice. A significant increase in KI67+ cells was observed with HFB301001, both in CD4+ and CD8+ T cells. Also, a significant reduction in PD-1+ cells was observed with HFB301001, both in CD4+ and CD8+ T cells. Further, both benchmark and HFB301001 led to significant reduction on tumor Tregs, with a stronger decrease in the HFB301001 group.

POTENTIAL MECHANISM OF DIFFERENTIATION

Differentiation model

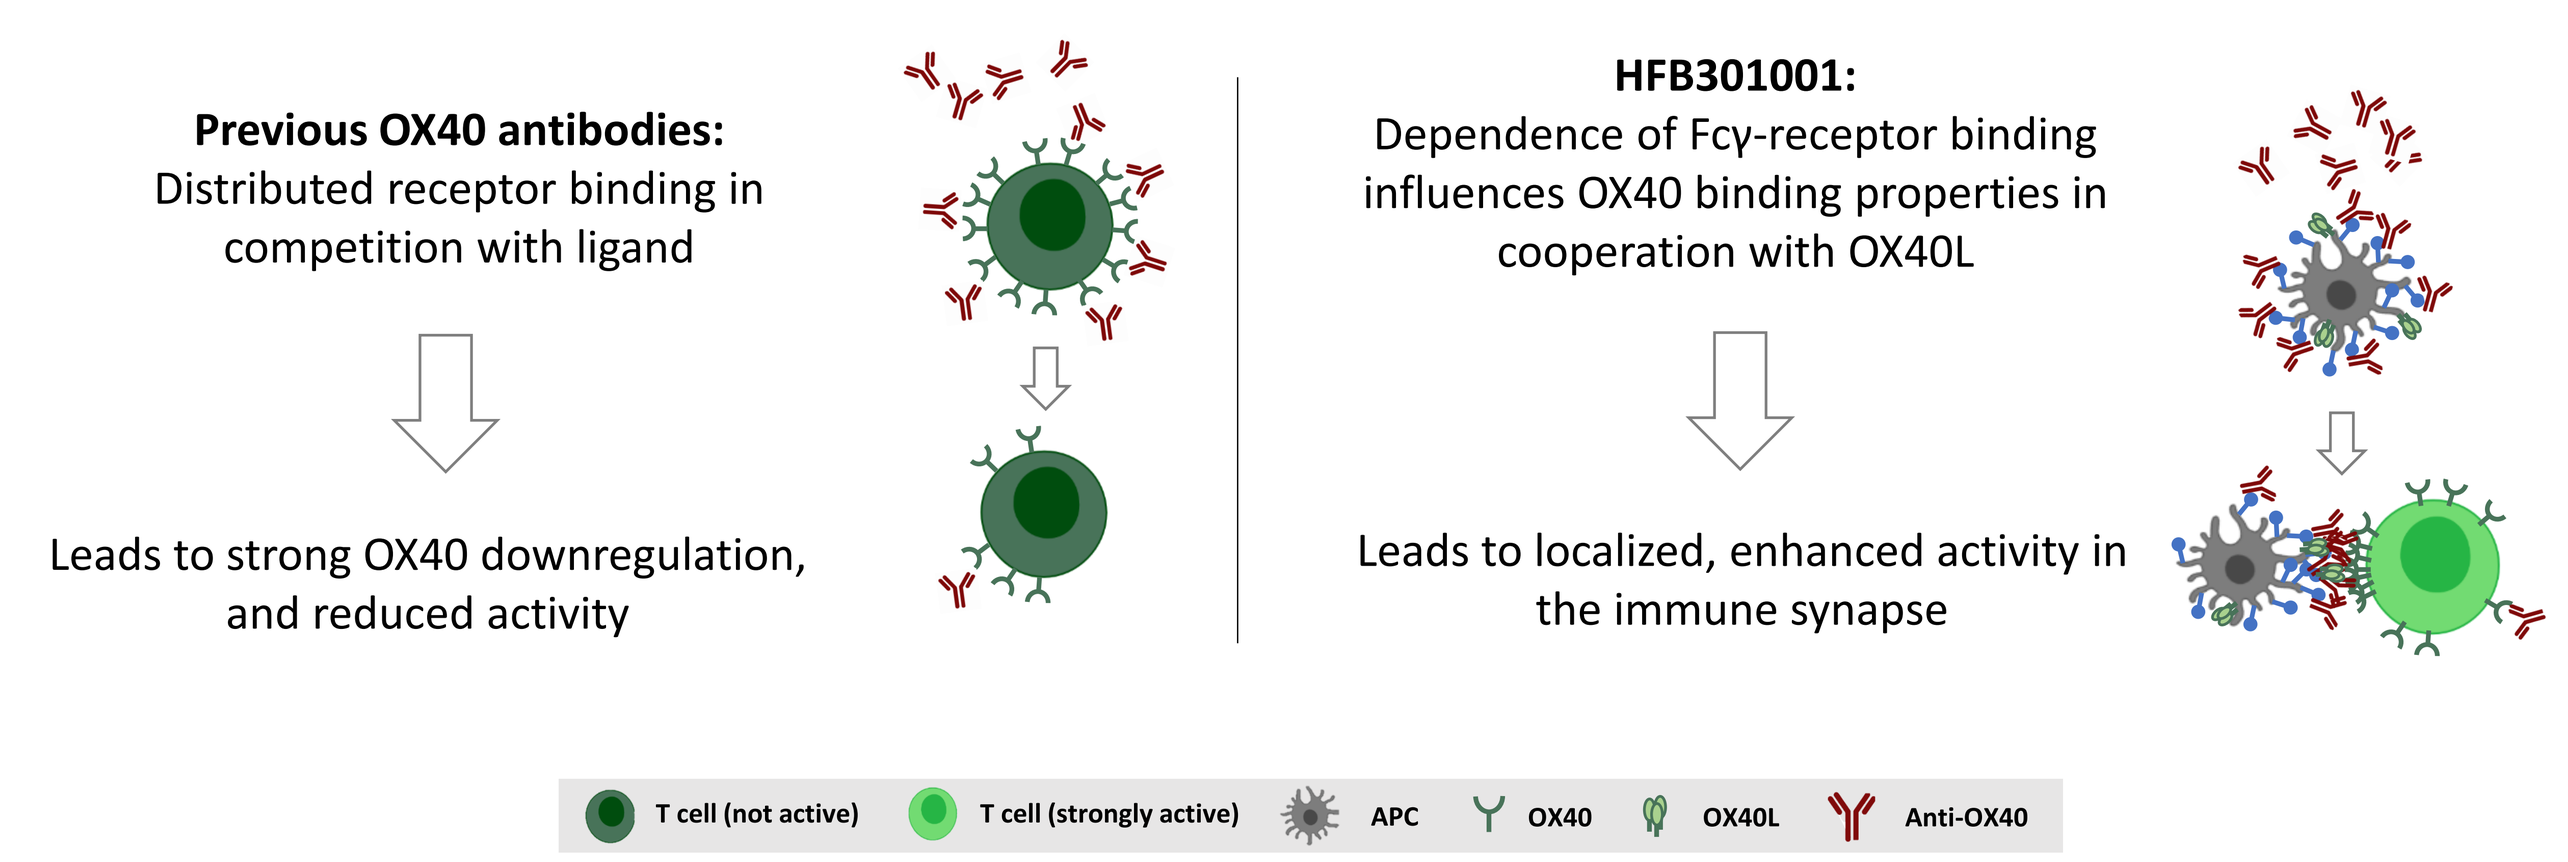
Previous OX40 antibodies:
Distributed receptor binding in competition with ligand
Leads to strong OX40 downregulation, and reduced activity
HFB301001:
Dependence of Fcγ-receptor binding influences OX40 binding properties in cooperation with OX40L
Leads to localized, enhanced activity in the immune synapse
T cell (not active)
T cell (strongly active)
APC
OX40
OX40L
Anti-OX40

CLINICAL STRATEGY

Our strategy for maximizing the probability of success in clinical trials

- ➢ Agonize OX40 in appropriate immunologic context and clinical setting
- ➢ Select for patients that will benefit from treatment using single-cell profiling for biomarker identification
- ➢ Combine with other therapies to increase treatable patient population and deepen responses

DIS single-cell approach for the discovery of predictive response biomarkers

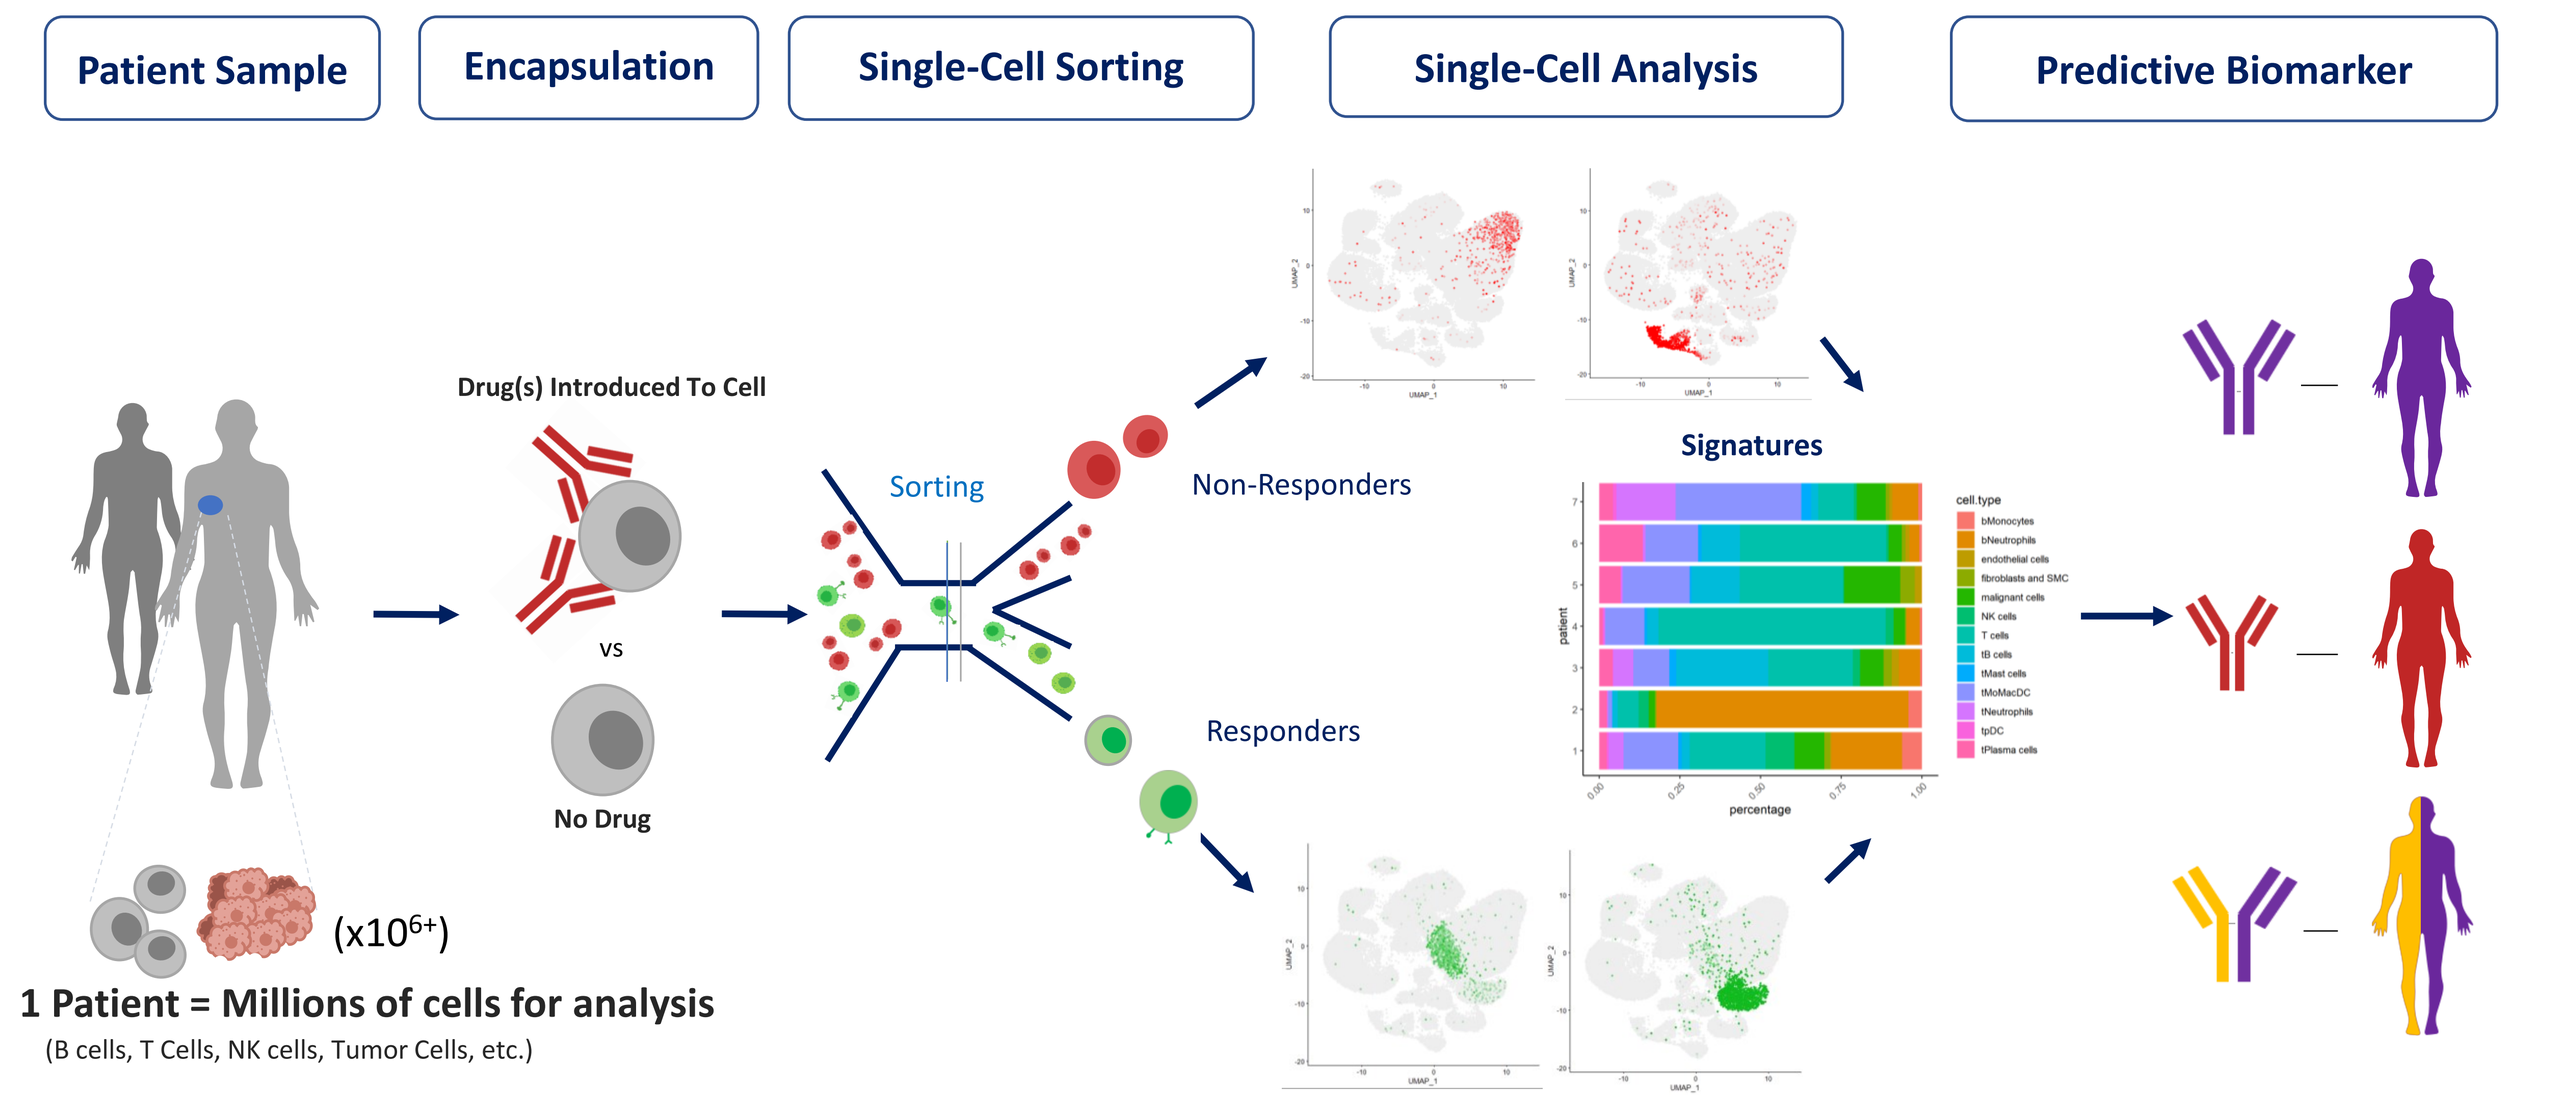
Patient Sample
Encapsulation
Single-Cell Sorting
Single-Cell Analysis
Predictive Biomarker
Drug(s) Introduced To Cell
vs
No Drug
Sorting
Non-Responders
Responders
Signatures
(x10^6+)
1 Patient = Millions of cells for analysis
(B cells, T Cells, NK cells, Tumor Cells, etc.)

- Single-cell resolution increases statistical power by leveraging heterogeneity within a patient's sample
- Reduces number of patients need for predictive biomarker discovery

Biomarker hypothesis: T cell characteristics in the TME determine outcome of HFB301001 treatment

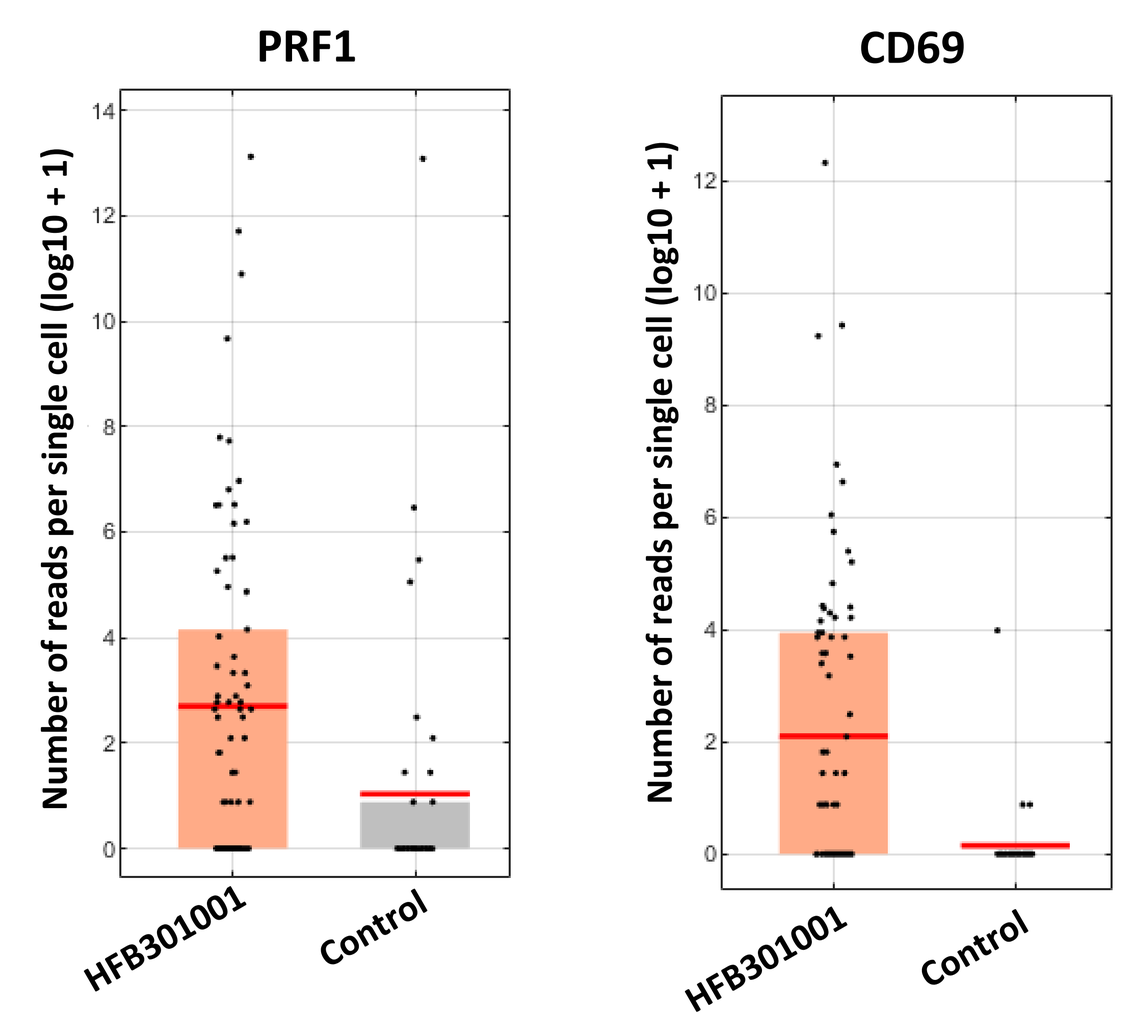
PRF1
CD69
Number of reads per single cell (log10 + 1)
HFB301001
Control

- Gene panel approach represents key T cell characteristics (activation/exhaustion, migration, proliferation, etc.)
- TCR clonotyping has recently proposed as an effective predictor of patient outcome to ICI therapy[3]
- Trafficking between TME and PBMCs enables possible blood-based biomarker screen to enhance clinical success

Figure 7. Increased activity and cytotoxicity of T cell derived from a lung cancer tumor sample was observed after treatment with HFB301001 by single-cell profiling. This data demonstrates the feasibility of profiling of responding T cells at the single-cell level using patient tissue.

3. Wu, T., et al. Nature (2020) Peripheral T cell expansion predicts tumour infiltration and clinical response. doi:10.1038/s41586-020-2056-8

CONCLUSIONS

- HFB301001 binds to a unique epitope on OX40 and does not interfere with OX40L, unlike benchmarks
- HFB301001 does not lead to significant receptor downregulation, unlike benchmarks
- HFB301001 has enhanced anti-tumor activity and prolonged survival in tumor model compared to a benchmark
- ➢ **Differentiated molecule positioned for potentially better clinical activity**

- Single-cell approach to discover predictive response biomarkers for HFB301001
- Clinical biomarker validation strategy planned to treat patients that are most likely to respond
- ➢ **Innovative clinical strategy to maximize patient benefit**

For additional information, please email contact@hifibio.com or visit Hifibio.com